Poster #0129

Identification of highly potent and tissue-specific promoters with massively parallel screening

Chenjin Jin[1], Camilla Belamarich[1] , Ann Lettko[1], Dustin Lee[1], Jeroen Bastiaans[1], Edgar Hernandez[1], Ce Feng Liu[1], Enrico Mossotto*[1] and Josefa M. Sullivan*[1]

[1] MeiraGTx New York, *Co-last authors

Optimizing *Cis*-Regulatory Elements to Develop Stronger and Safer Gene Therapies

Benefits of Promoter Engineering for Gene Therapies

- Precise control of therapeutic gene expression[1]
- Cell-specific expression independent of capsid[2]
- Increased potency may decrease immune responses and safety risks[3]
- Promoter kinetics impact the durability of gene therapy
- Reducing size without sacrificing strength or specificity allows efficient packaging or larger cargo[4]

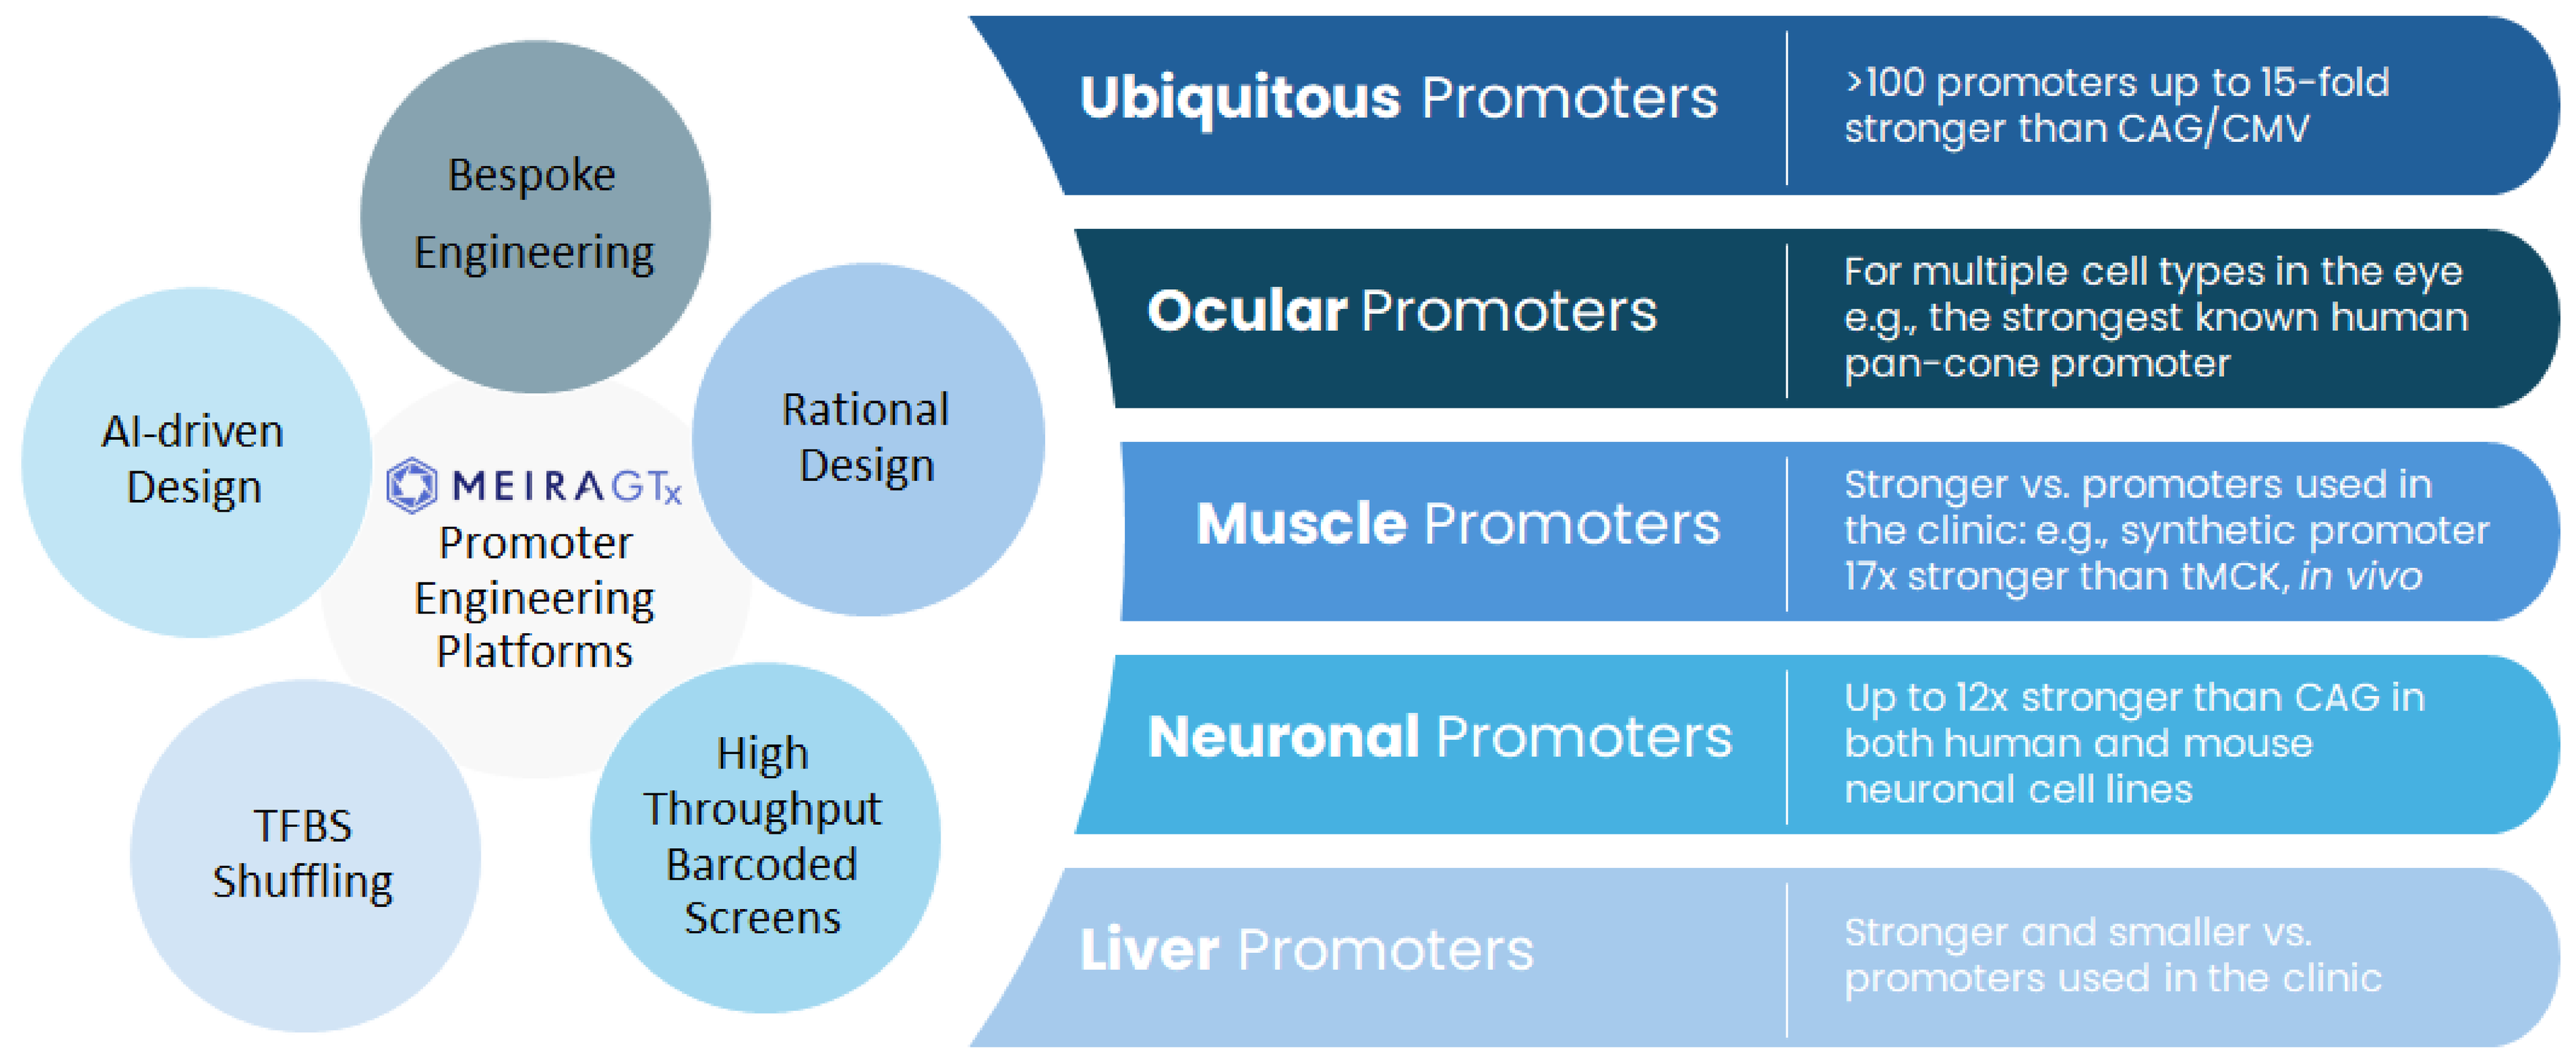
Bespoke Engineering
Rational Design
AI-driven Design
MEIRAGTx Promoter Engineering Platforms
TFBS Shuffling
High Throughput Barcoded Screens
Ubiquitous Promoters
>100 promoters up to 15-fold stronger than CAG/CMV
Ocular Promoters
For multiple cell types in the eye e.g., the strongest known human pan-cone promoter
Muscle Promoters
Stronger vs. promoters used in the clinic: e.g., synthetic promoter 17x stronger than tMCK, in vivo
Neuronal Promoters
Up to 12x stronger than CAG in both human and mouse neuronal cell lines
Liver Promoters
Stronger and smaller vs. promoters used in the clinic

We have built libraries of synthetic promoters and *cis*-regulatory elements available for viral vector optimization. These libraries have a broad range of size, cellular specificity, and potency which can be used in a diverse range of therapeutic applications. Here, we identify potent and small promoters by screening libraries of >200,000 sequences in various tissues.

Identification of Potent and Specific Mini Promoters using MeiraGTx Promoter Design Platform

MPRA screens identify small, but potent promoters in various tissues. (A) Bespoke libraries of >200,000 sequences can be screened using our MPRA platform. **(B)** Representative analysis of the NGS-based expression of a synthetic promoter library, here showing screening results from N2A cells, a mouse neuroblastoma line. Higher ratios indicate stronger promoter activity. **(C)** Specificity and expression level of high-performing candidates selected from screens in various tissues.

A.

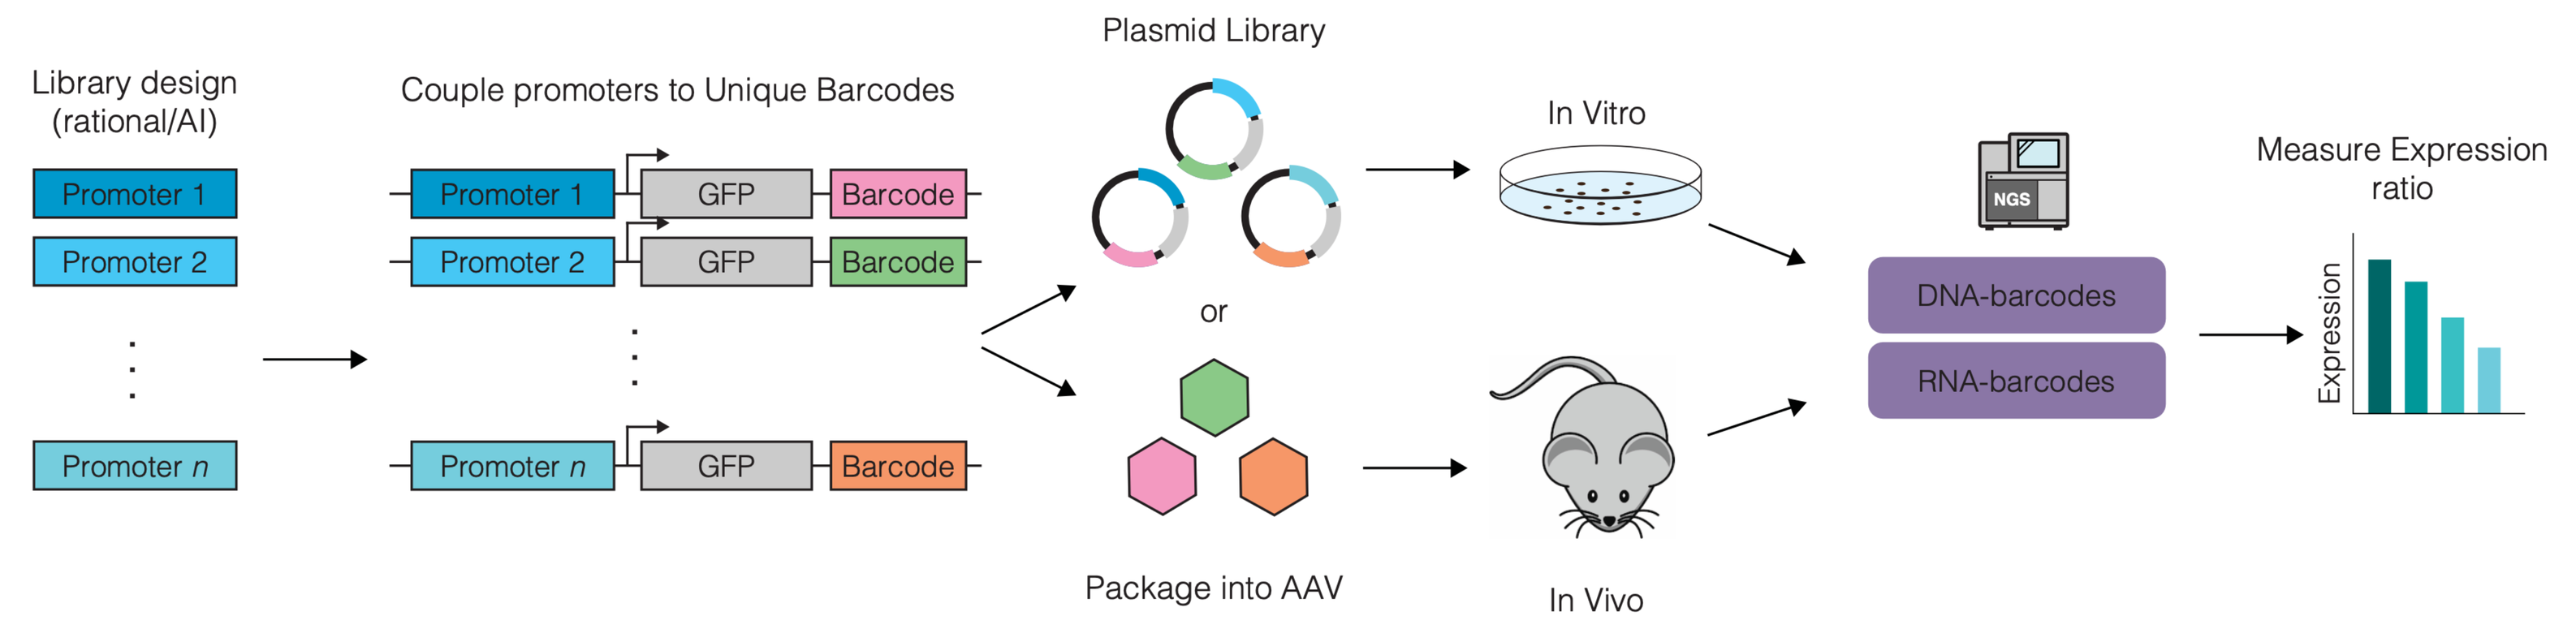
Library design (rational/AI)
Promoter 1
Promoter 2
Promoter n
Couple promoters to Unique Barcodes
GFP
Barcode
Plasmid Library
or
Package into AAV
In Vitro
In Vivo
NGS
DNA-barcodes
RNA-barcodes
Measure Expression ratio
Expression

B.

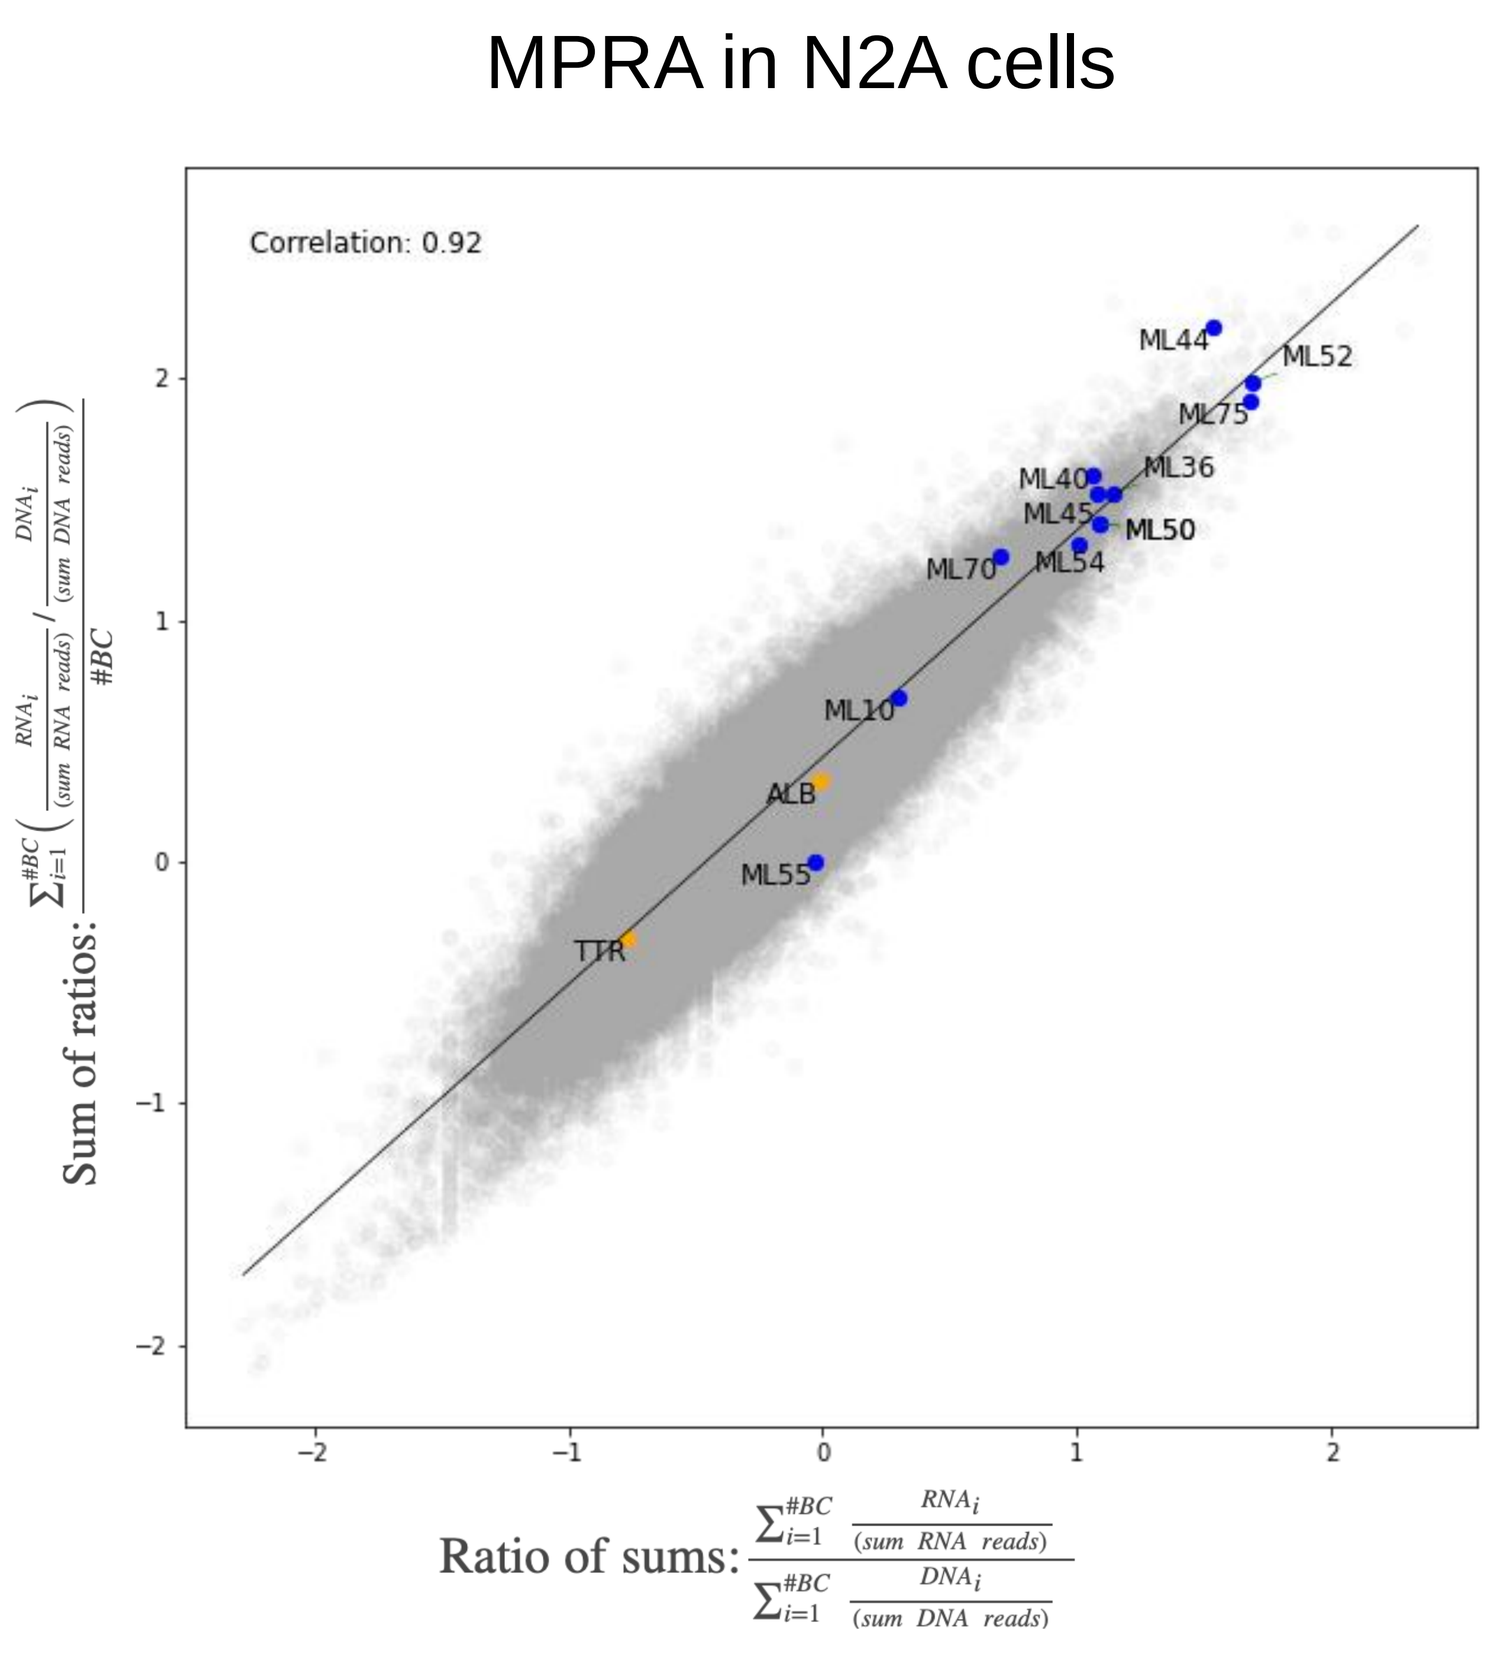
MPRA in N2A cells
Correlation: 0.92
ML44
ML52
ML75
ML40
ML36
ML45
ML50
ML70
ML54
ML10
ALB
ML55
TTR
Sum of ratios
Ratio of sums

C.

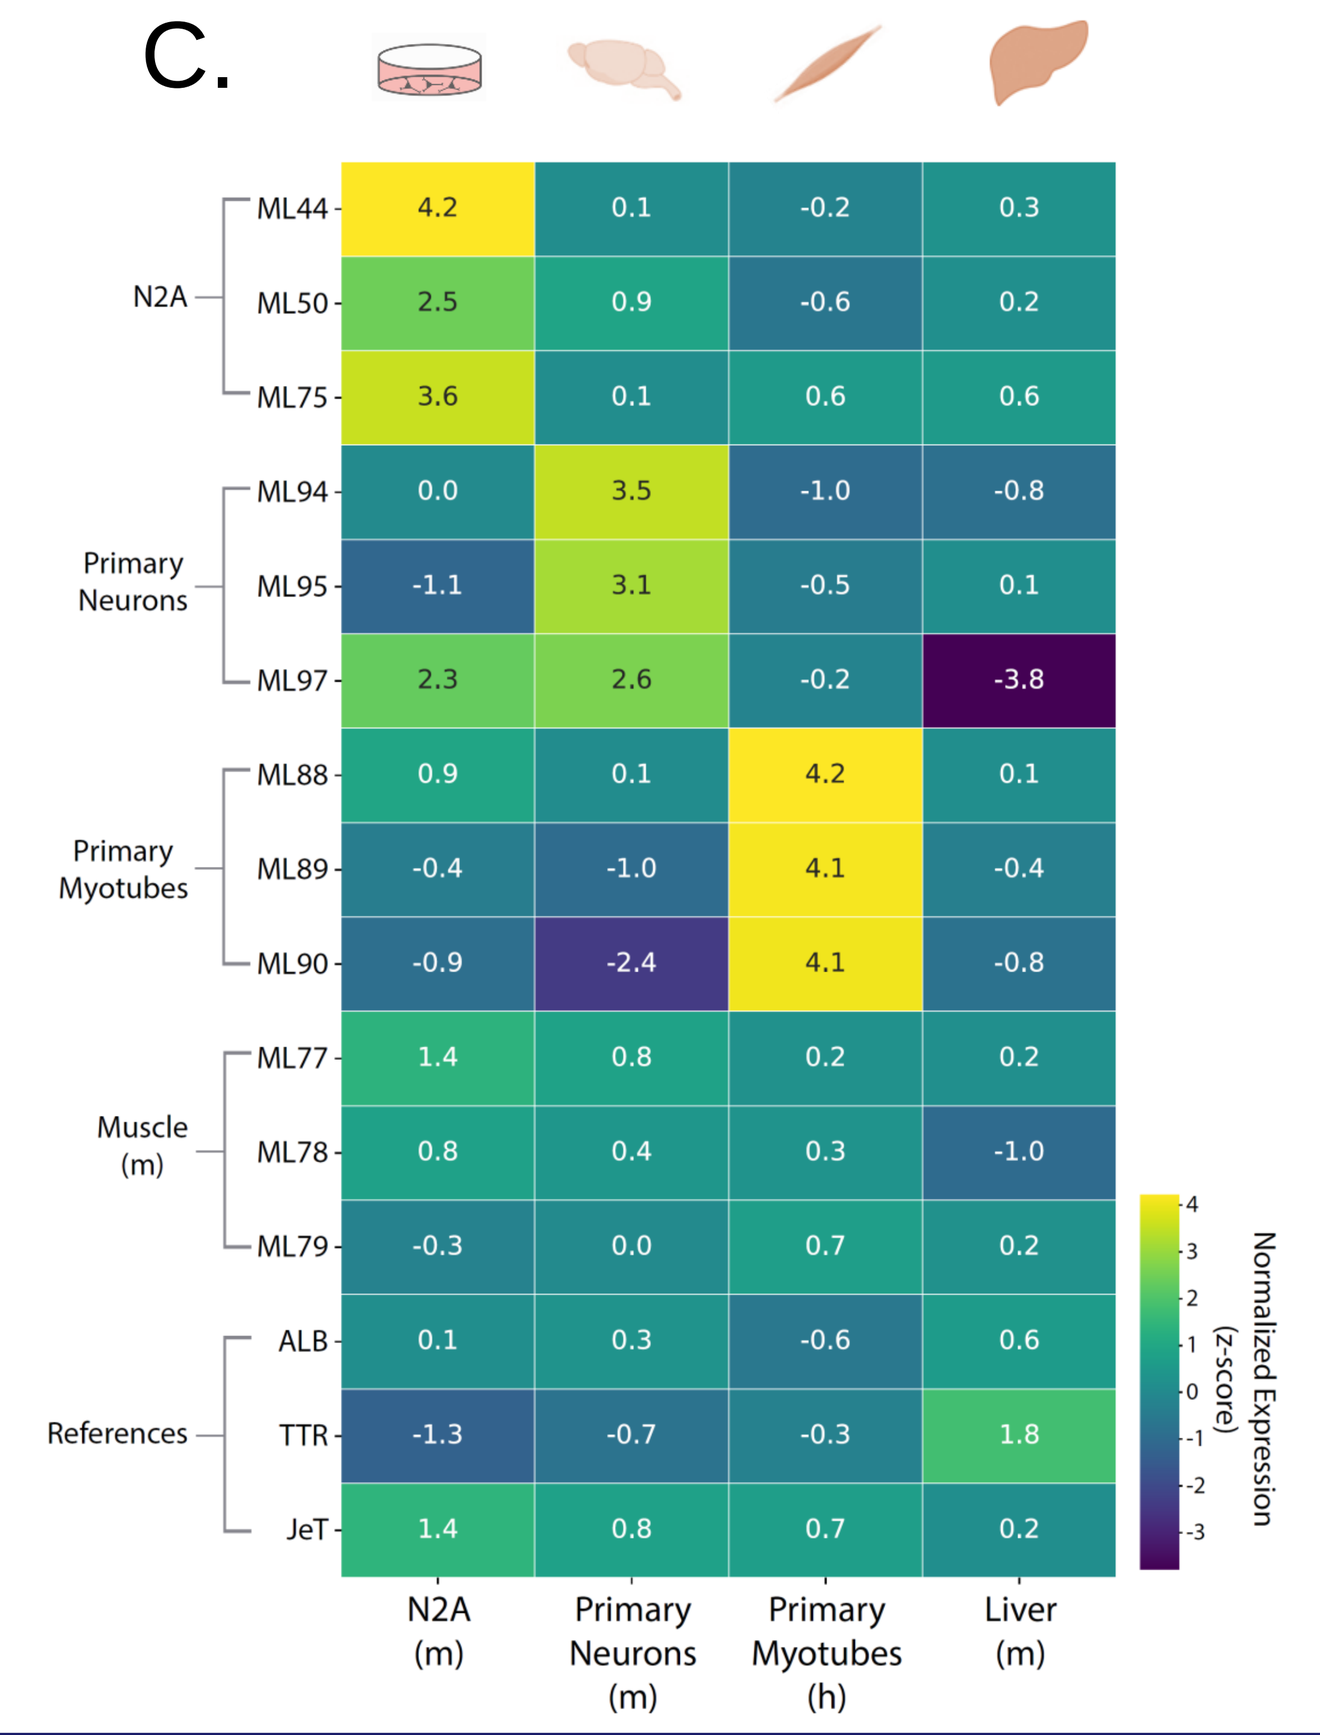
N2A
ML44 4.2 0.1 -0.2 0.3
ML50 2.5 0.9 -0.6 0.2
ML75 3.6 0.1 0.6 0.6
Primary Neurons
ML94 0.0 3.5 -1.0 -0.8
ML95 -1.1 3.1 -0.5 0.1
ML97 2.3 2.6 -0.2 -3.8
Primary Myotubes
ML88 0.9 0.1 4.2 0.1
ML89 -0.4 -1.0 4.1 -0.4
ML90 -0.9 -2.4 4.1 -0.8
Muscle (m)
ML77 1.4 0.8 0.2 0.2
ML78 0.8 0.4 0.3 -1.0
ML79 -0.3 0.0 0.7 0.2
References
ALB 0.1 0.3 -0.6 0.6
TTR -1.3 -0.7 -0.3 1.8
JeT 1.4 0.8 0.7 0.2
N2A (m)
Primary Neurons (m)
Primary Myotubes (h)
Liver (m)
Normalized Expression (z-score)

Mini Promoters are up to 12x Stronger than CAG

Mini promoters identified by screening in N2A cells are stronger than CAG in vitro. (A) The activity of promising mini-promoter candidates was validated using a flow cytometry-based *in vitro* assay. Promoter candidates were cloned upstream of mClover3 in a dual-reporter plasmid, which also contains a tdTomato (RFP) expression cassette that is used as an internal transfection control. Promoter activity is quantified as the ratio of the median fluorescence intensity of mClover3 and tdTomato in single, live tdTomato+ cells. **(B)** Relative expression of promoters in mouse N2A and human HuH7 cells using the dual reporter assay. **(C)** Strong correlation of NGS expression (barcodes) and independent assay expression (protein fluorescence) shows high confidence in the predictive power of high-throughput screening and bioinformatic hit selection.

A.

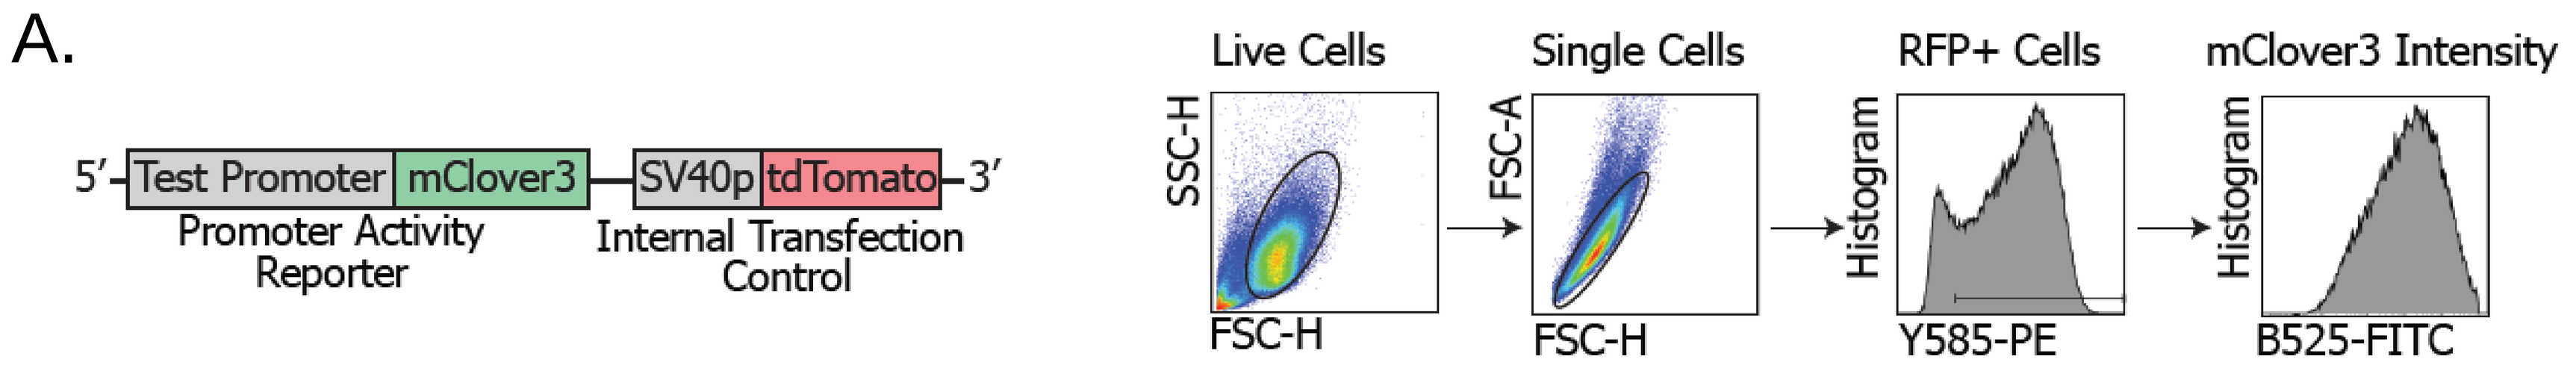
5′ Test Promoter mClover3 SV40p tdTomato 3′
Promoter Activity Reporter
Internal Transfection Control
Live Cells
Single Cells
RFP+ Cells
mClover3 Intensity
SSC-H
FSC-H
FSC-A
Histogram
Y585-PE
B525-FITC

B.

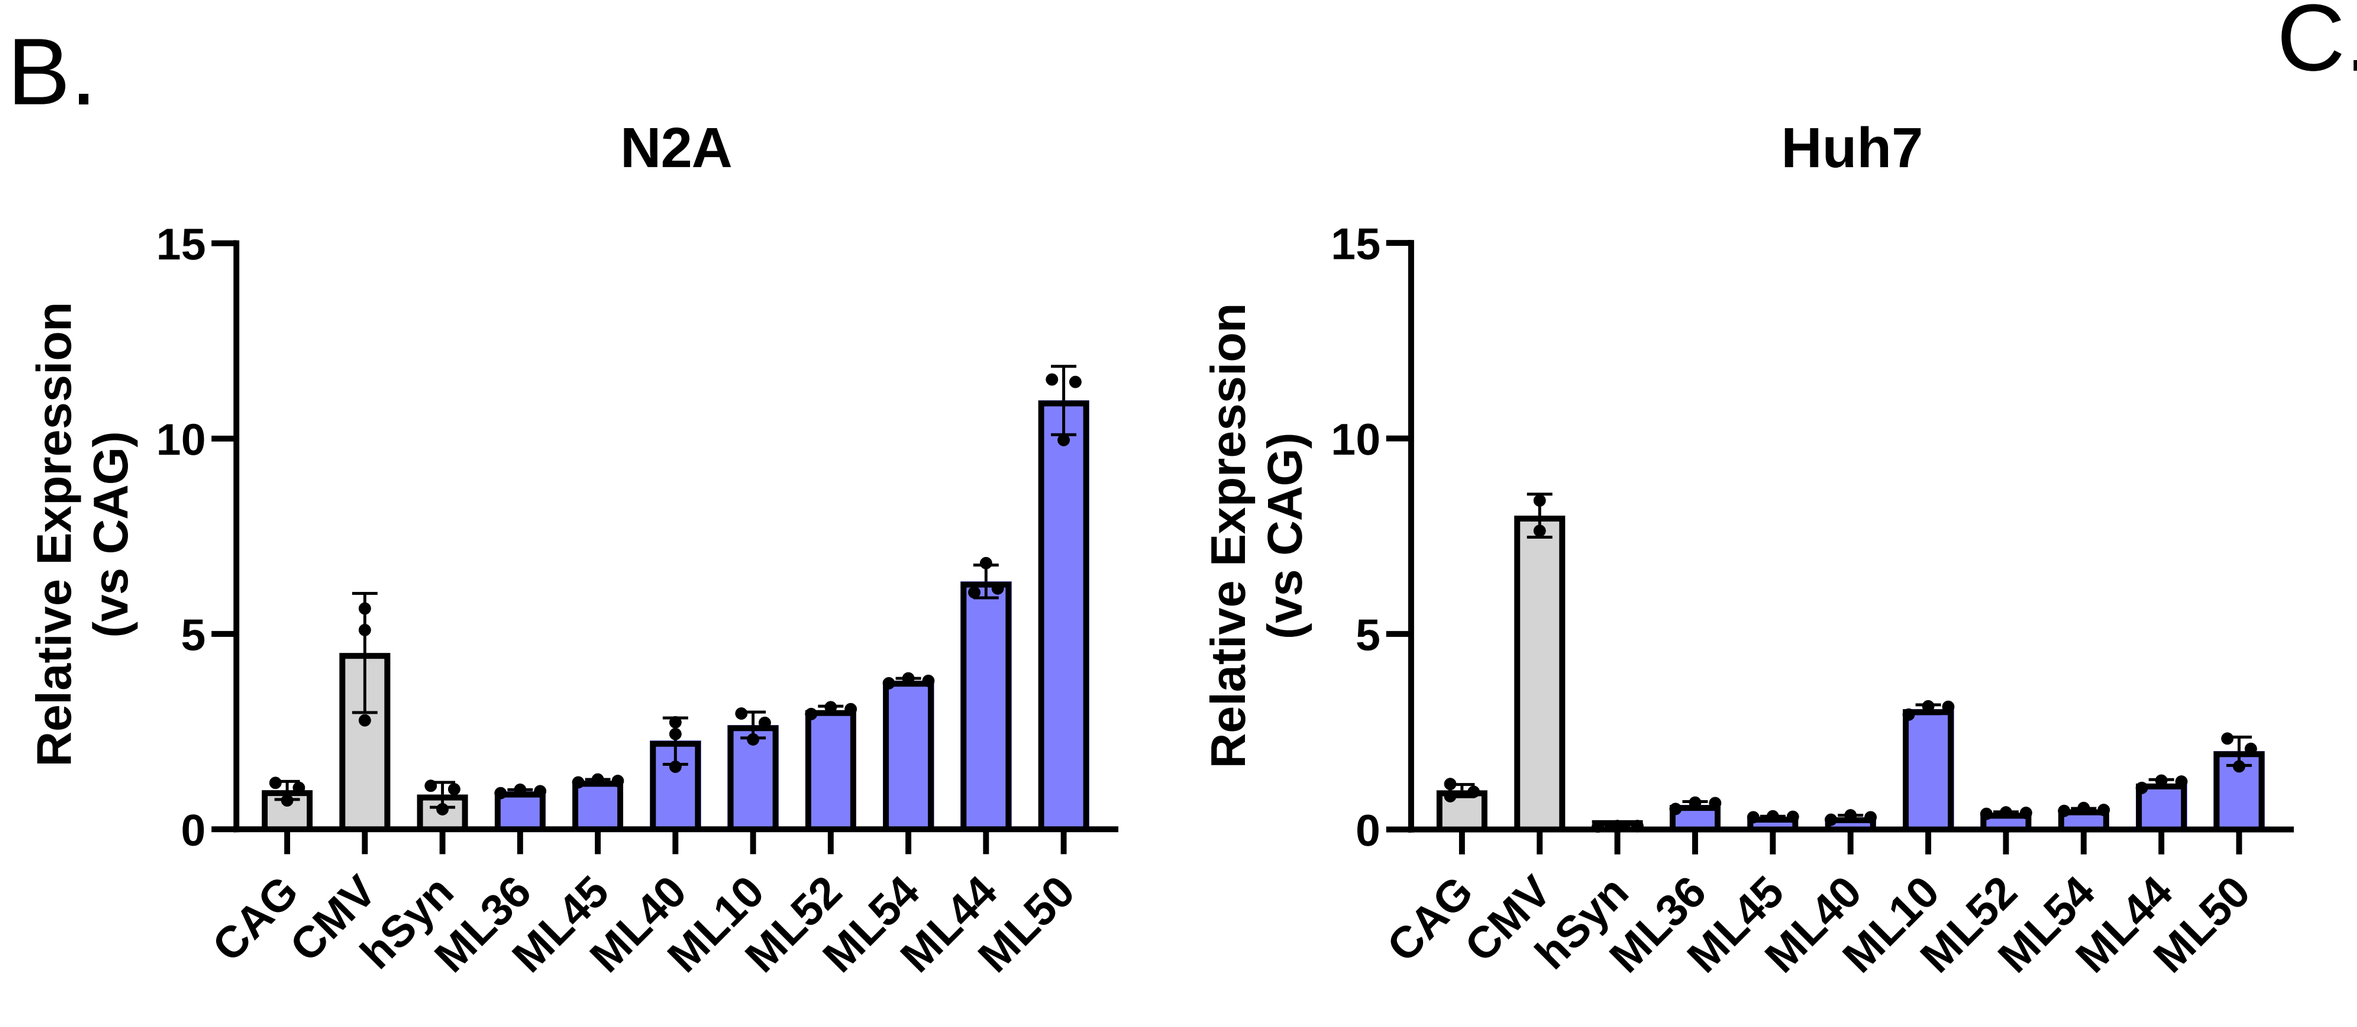
N2A
Huh7
Relative Expression (vs CAG)
CAG
CMV
hSyn
ML36
ML45
ML40
ML10
ML52
ML54
ML44
ML50

C.

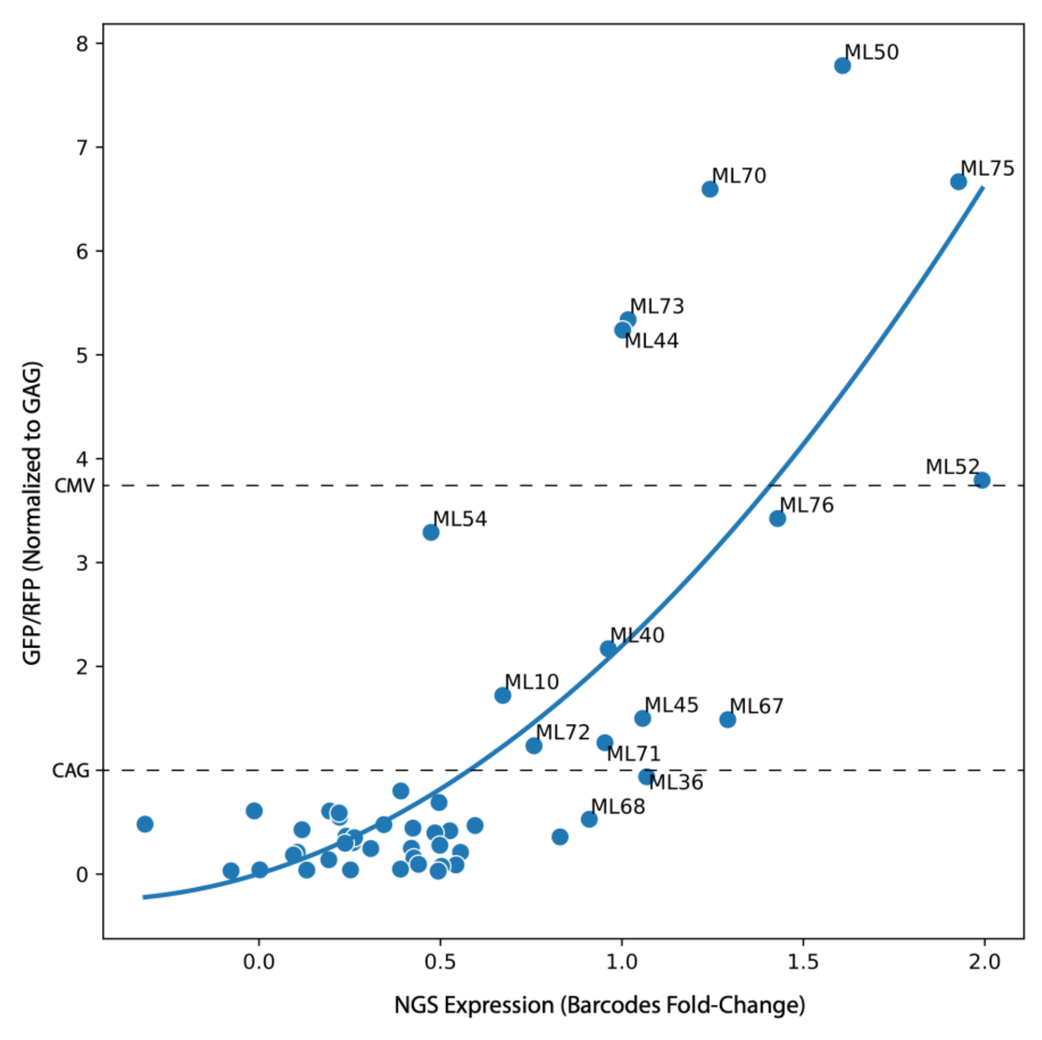
GFP/RFP (Normalized to CAG)
NGS Expression (Barcodes Fold-Change)
CMV
CAG
ML50
ML70
ML75
ML73
ML44
ML52
ML54
ML76
ML40
ML10
ML45
ML67
ML72
ML71
ML36
ML68

Mini Promoters Drive High Expression in the CNS

Expression and specificity of MeiraGTx CNS promoter candidates. (A) On day 4 *in vitro*, primary mouse cortical neurons were transduced at 50,000 multiplicity of infection with AAV9-miRFP713 driven by candidate promoters. After 7 days, neurons were imaged to assess miRFP713 expression. Scale bar: 300 µm. **(B)** Image quantification at 7 days post infection shows that ML44 and ML50 express equal to or stronger than JeT and hSyn despite their small size. **(C)** Promoter activity was assessed *in vivo* by bioimaging the mIRFP713 reporter gene 4 weeks after systemic or direct injection. **(D)** Quantification of promoter expression in the mouse brain (left), liver (middle) and muscle (right).

A.

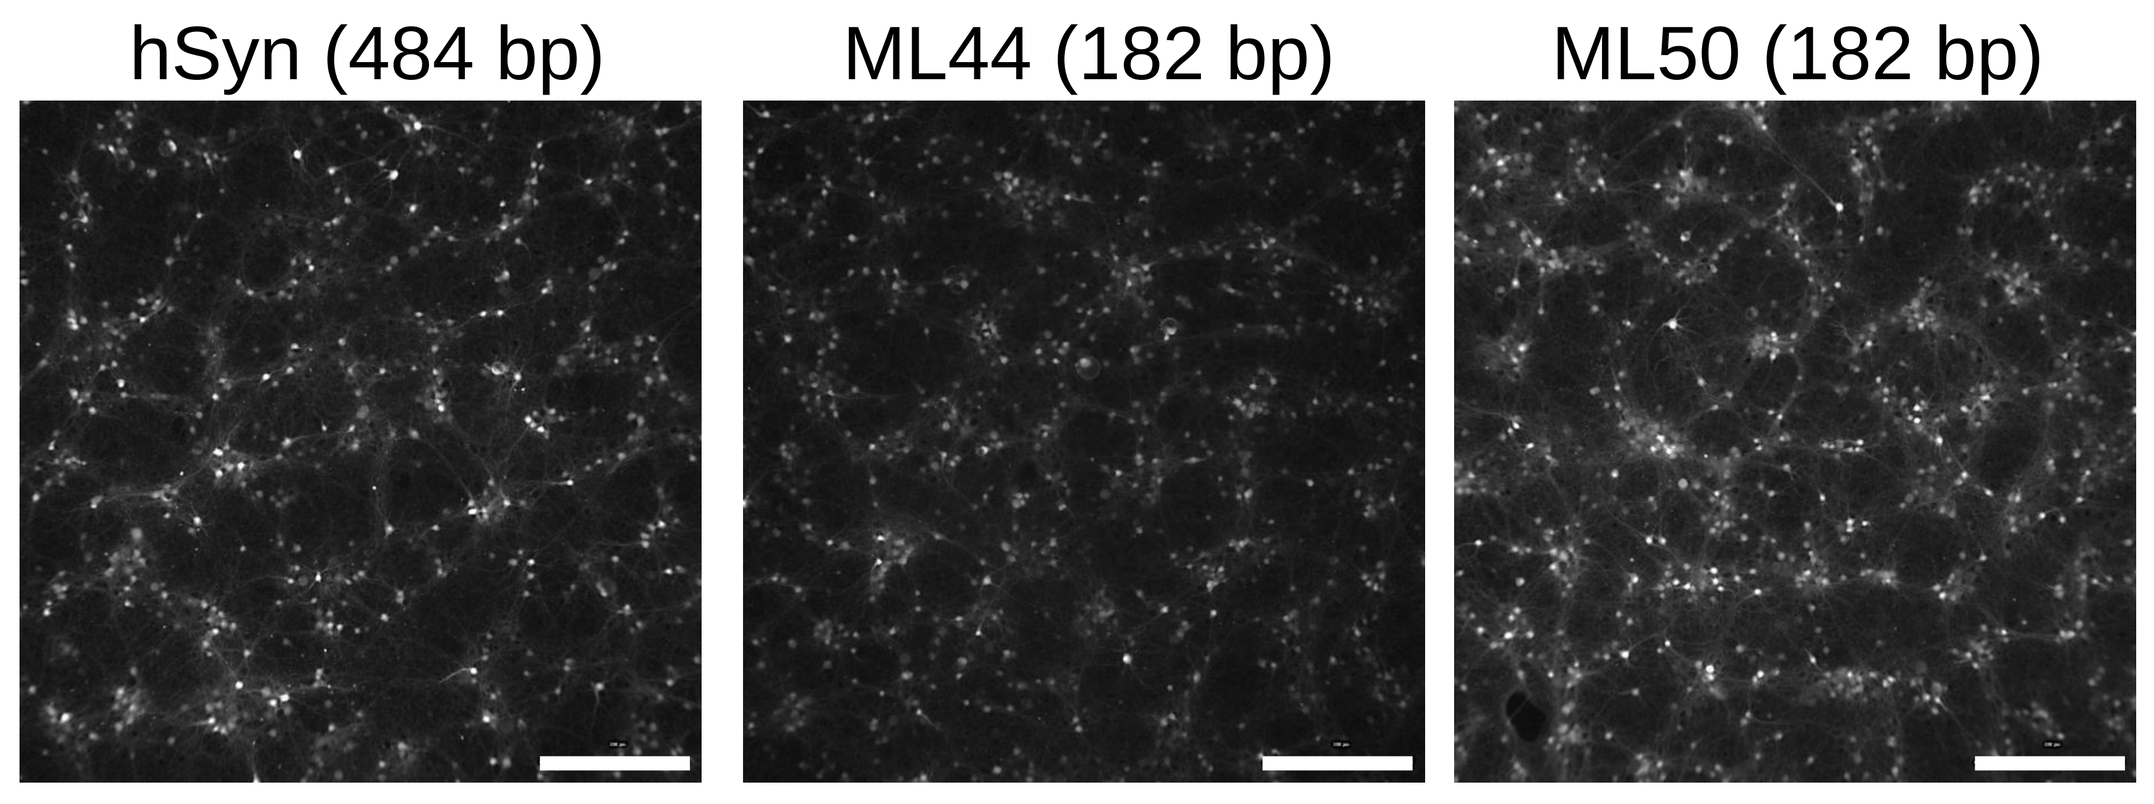
hSyn (484 bp)
ML44 (182 bp)
ML50 (182 bp)

B.

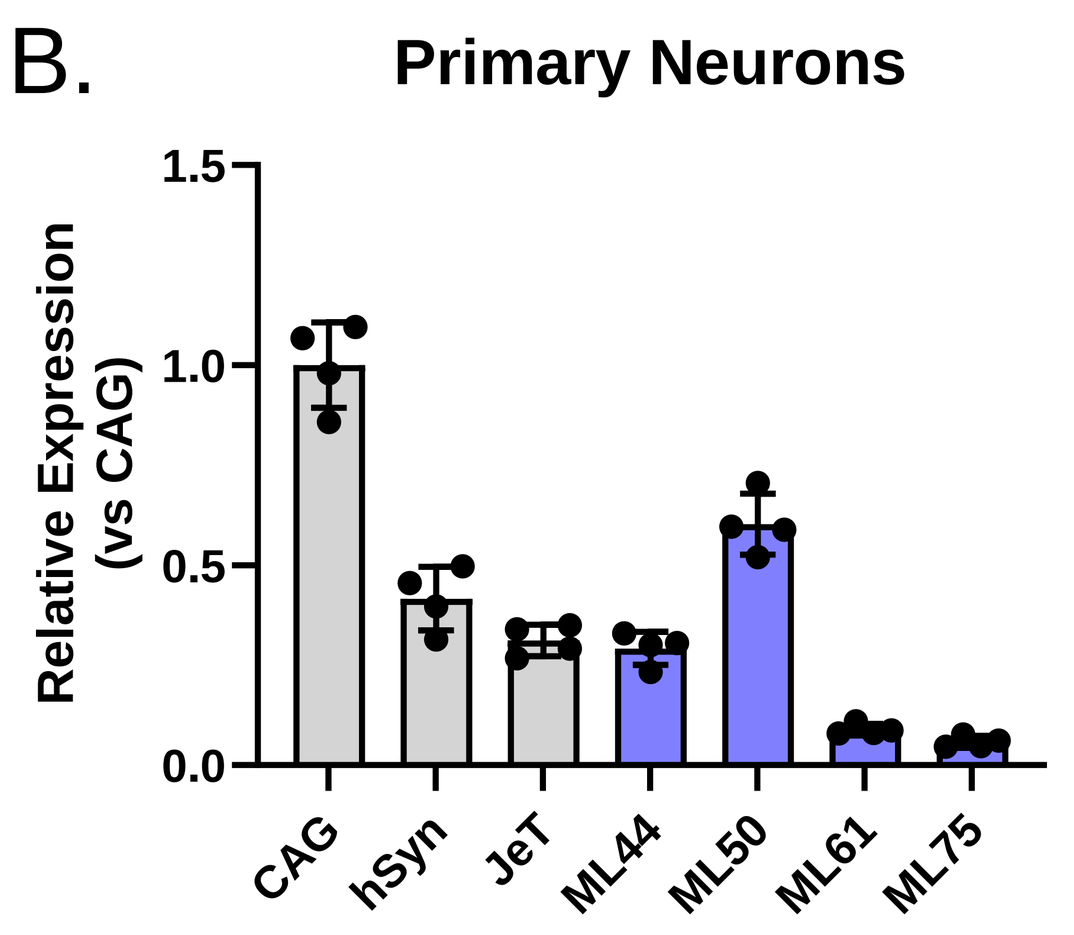
Primary Neurons
Relative Expression (vs CAG)
CAG
hSyn
JeT
ML44
ML50
ML61
ML75

C.

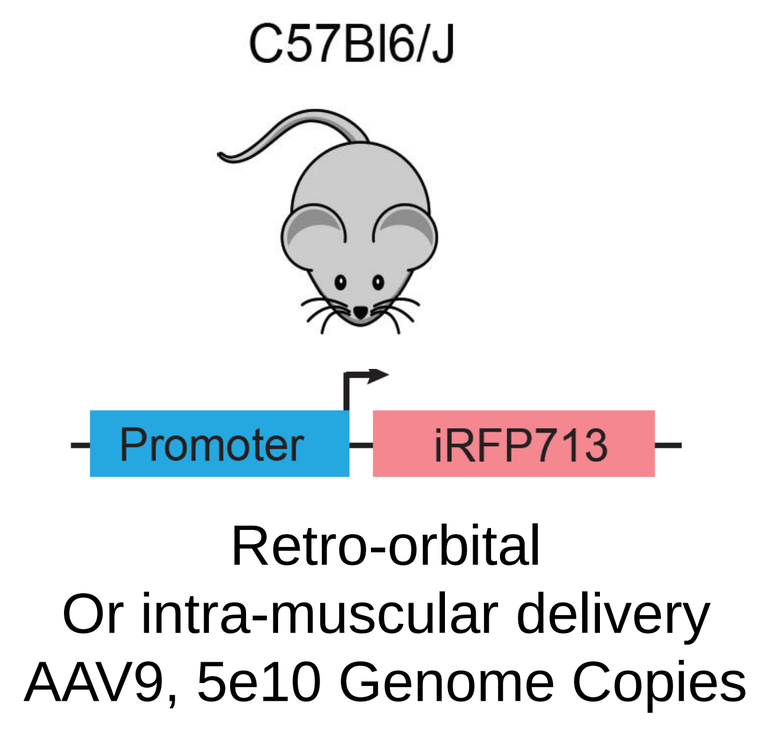
C57Bl6/J
Promoter
iRFP713
Retro-orbital
Or intra-muscular delivery
AAV9, 5e10 Genome Copies

D.

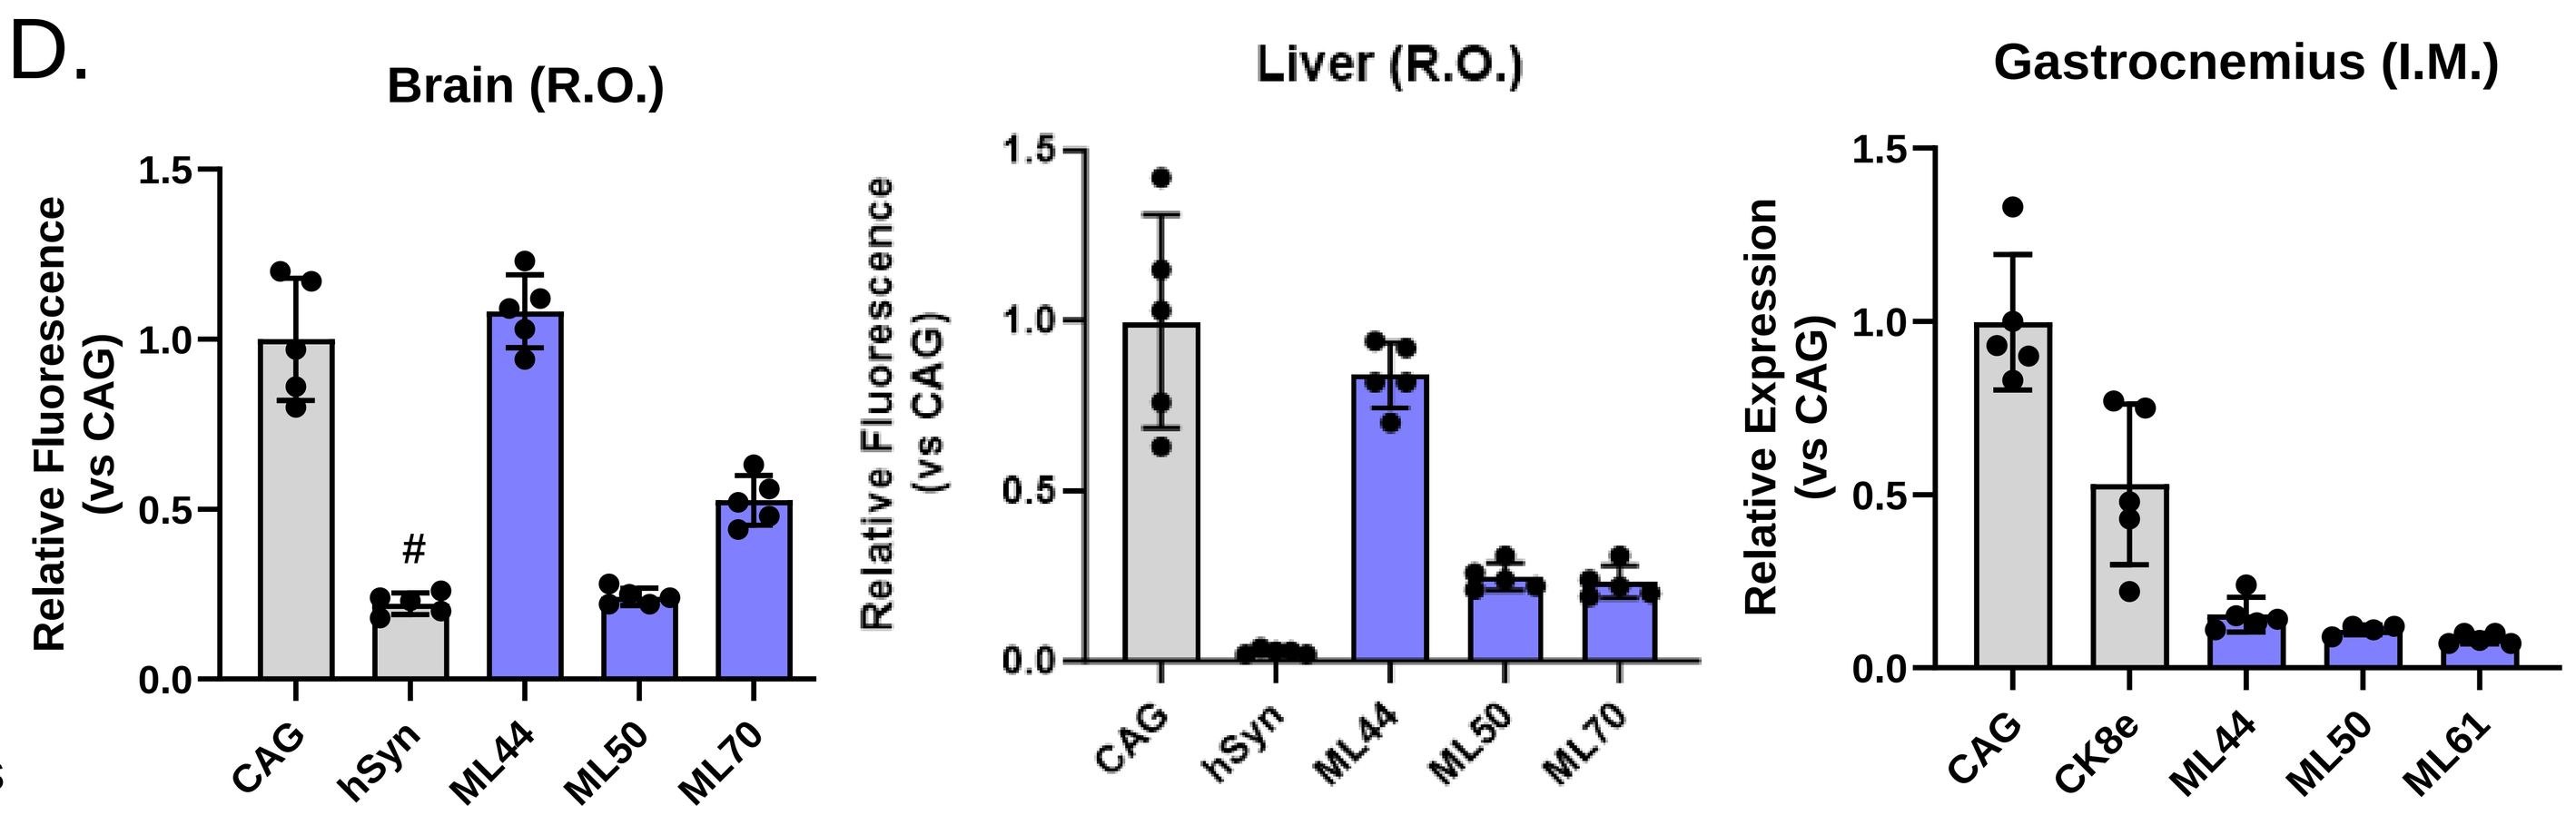
Brain (R.O.)
Liver (R.O.)
Gastrocnemius (I.M.)
Relative Fluorescence (vs CAG)
Relative Expression (vs CAG)
#
CAG
hSyn
ML44
ML50
ML70
CK8e
ML61

MeiraGTx Promoters are Stronger than Ck8e in Human Myotubes

Expression of MeiraGTx muscle promoter candidates in human iPSC-derived myotubes. (A) Experimental paradigm to test promoter expression in human induced pluripotent stem cell (hiPSC) derived myotubes. hiPSC-derived myoblasts are differentiated for 8 days. Cultures are then transduced with 200,000 multiplicity of infection with AAV9-mIRFP713 driven by candidate promoters. Cells are imaged and quantified 11 days after infection. (**B**) Representative images of transduced myotubes showing higher expression driven by MeiraGTx promoters compared to Ck8e. **(C)** Mini promoters identified from the mouse muscle barcode screen are equal to or stronger than Ck8e while 60% smaller.

A.

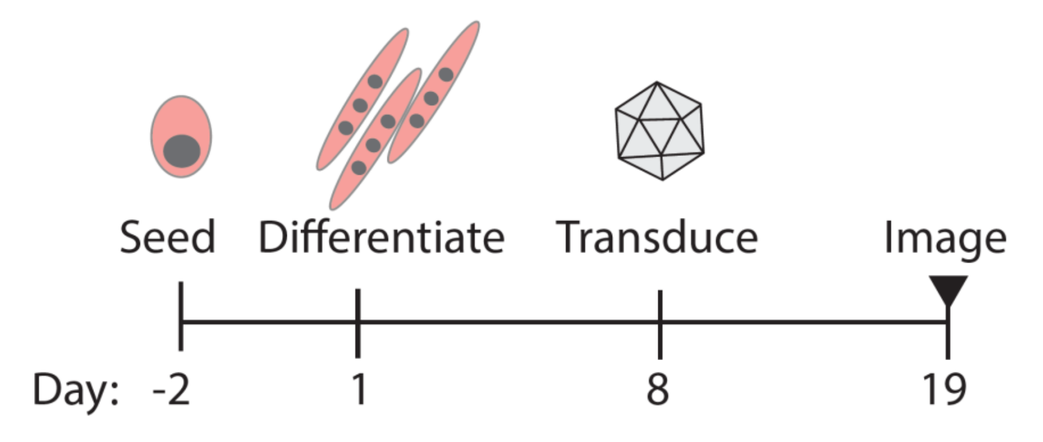
Seed
Differentiate
Transduce
Image
Day: -2
1
8
19

B.

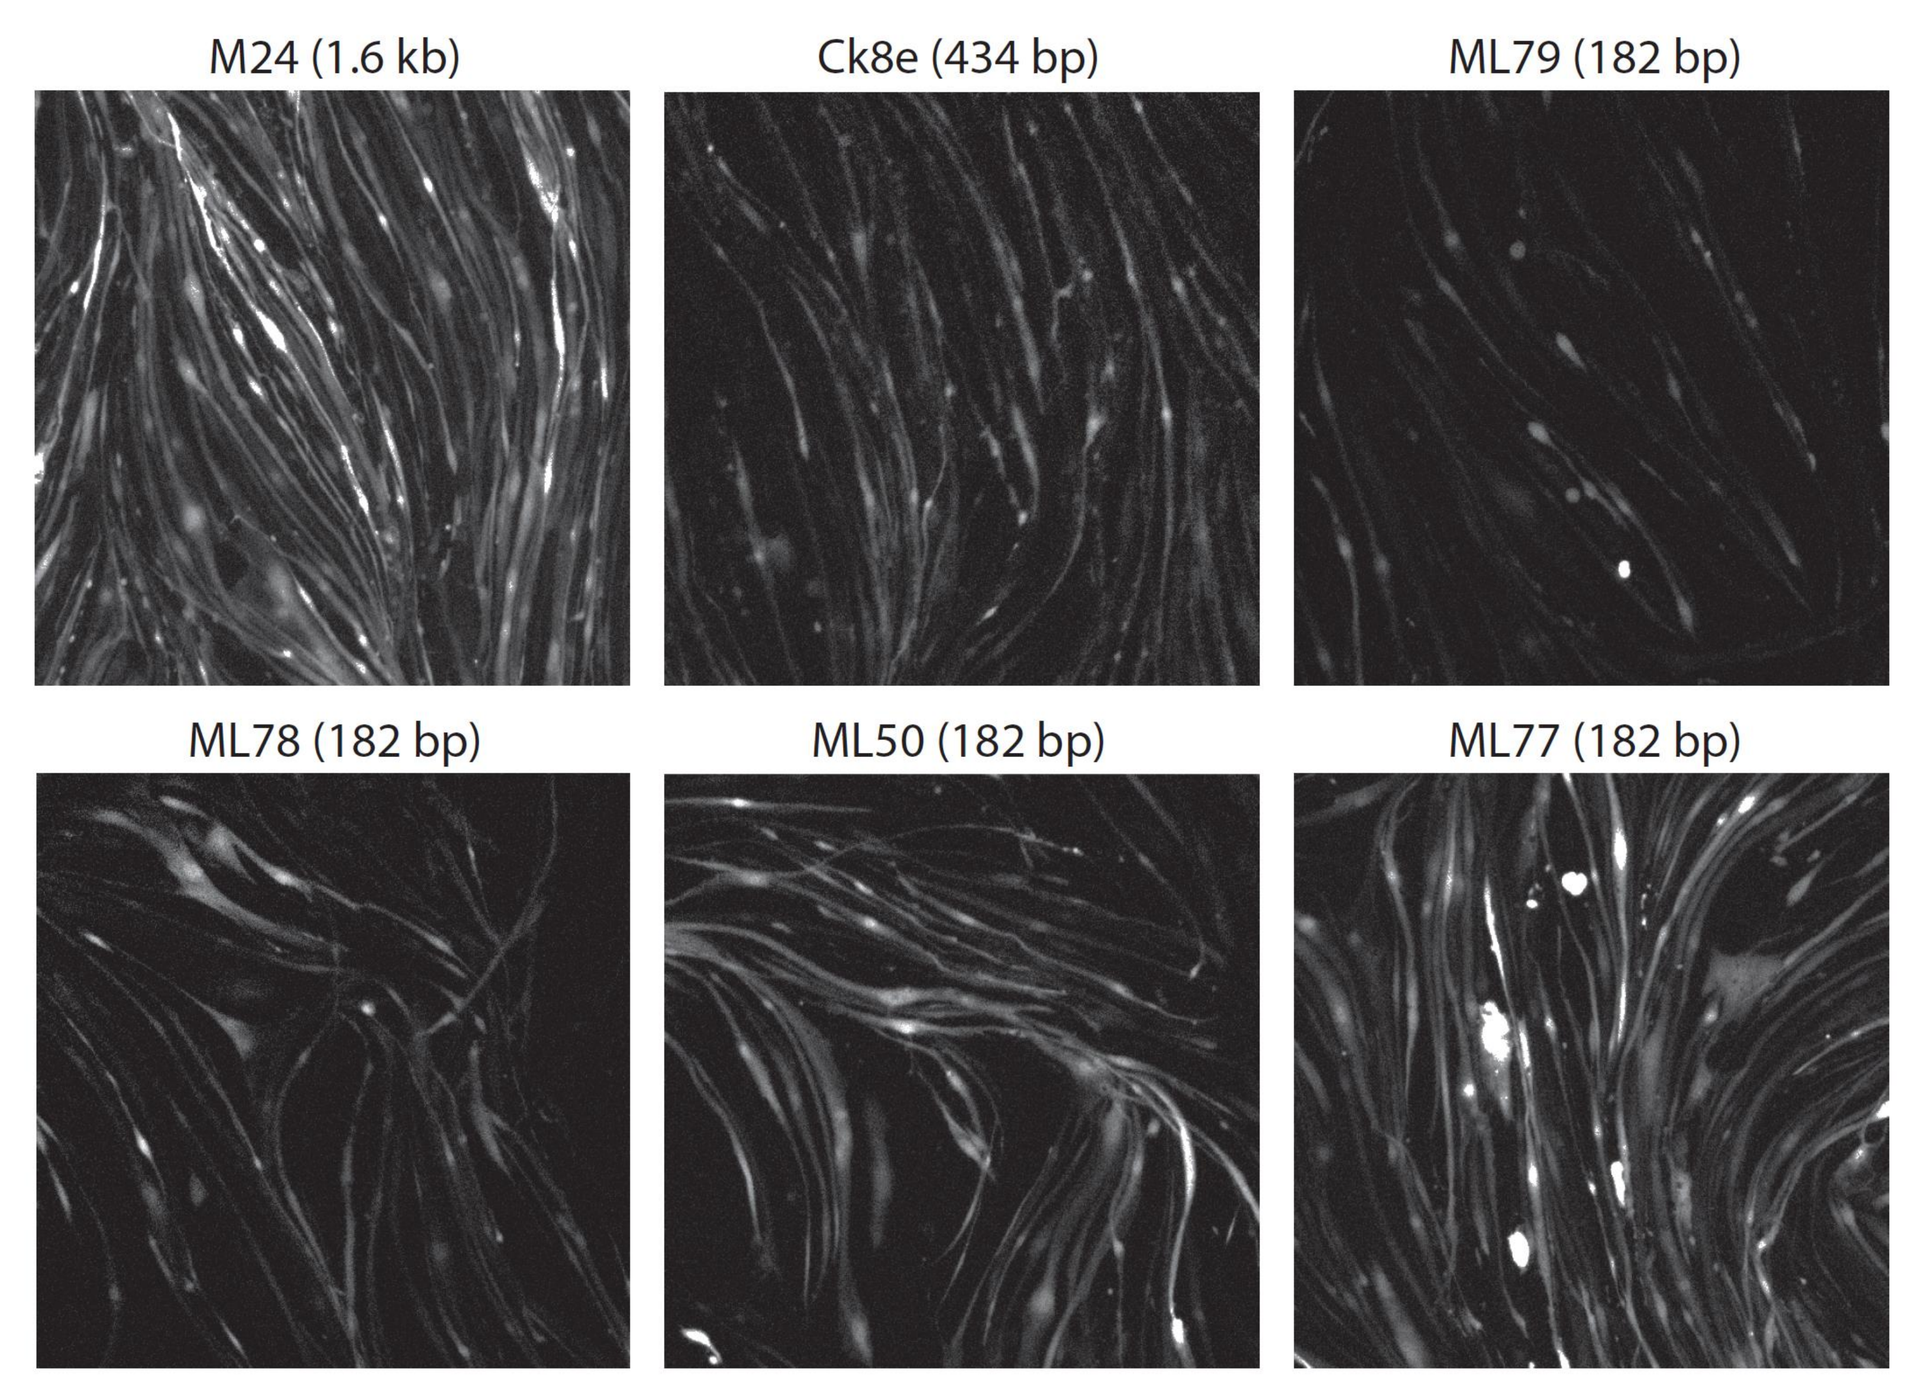
M24 (1.6 kb)
Ck8e (434 bp)
ML79 (182 bp)
ML78 (182 bp)
ML50 (182 bp)
ML77 (182 bp)

C.

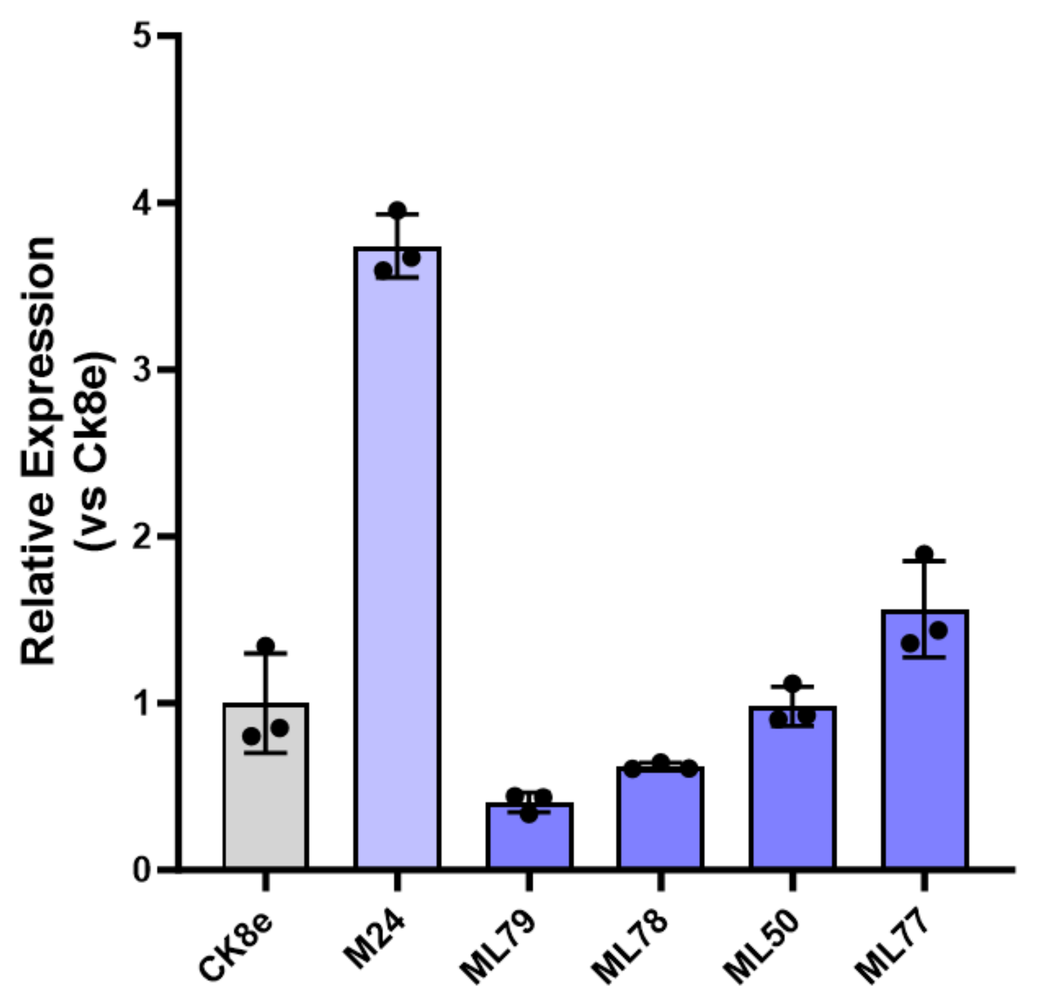
Relative Expression (vs Ck8e)
CK8e
M24
ML79
ML78
ML50
ML77

Conclusions

- MeiraGTx has a robust and predictive platform to screen hundreds of thousands of sequences in complex models *in vitro* and *in vivo,* independent of species.
- Here we show high-performing 182 bp promoters selected by our platform in brain, muscle and liver models.
- Independent validation at the protein expression level confirms the predictive power of our promoter engineering platform.
- ML44 and ML50 show strong expression in neuronal models despite small size.
- Muscle candidate ML77, is 56% stronger than Ck8e in hiPSC-derived myotubes and is ~60% smaller (182 bp vs 443 bp).
- <u>MeiraGTx synthetic mini promoters exhibit greatly improved potency for driving therapeutic gene expression in the CNS and muscle.</u>
- Future work:
 - Quantify expression and cellular specificity in the mouse CNS *in vivo*
 - Evaluate potency of neuronal and muscle candidates in human iPSC-derived models and *in vivo*
 - Examine promoter kinetics using long-term *in vivo* bioimaging

References

1. Wang et al. Adeno-associated virus vector as a platform for gene therapy delivery. *Nat Rev Drug Discov*. 2019 May;18(5):358-378.
2. Au et al*.* Gene Therapy Advances: A Meta-Analysis of AAV Usage in Clinical Settings. *Front Med* (Lausanne). 2022 Feb 9;8:809118.
3. Cordier et al. Muscle-specific promoters may be necessary for adeno-associated virus-mediated gene transfer in the treatment of muscular dystrophies. *Hum Gene Ther*. 2001 Jan 20;12(2):205-15.
4. Wu et al. Effect of genome size on AAV vector packaging. *Mol Ther*. 2010 Jan; 18(1): 80–86.