AAV-based evaluation of novel *in silico* promoters to drive expression in rod photoreceptors

P0020

Arifa Naeem[1], Izabela P. Klaska[1], Amelia Lane[1], Toyin Adefila-Ideozu[1], Paul E. Sladen[1], Enrico Mossotto[2], Josefa sullivan[2], Tijmen Vermeule[1], Gemma Estrada Girona[1], Sophia El-Alami[1], Monika Pianka[1], Jasmin Patel[1], Yanai Duran[1], Sophia Kleine Holthaus[1], Ce Feng Liu[2], Anastasios Georgiadis[1]

1. MeiraGTx UK, 34-38 Provost Street, London N1 7NG

2. MeiraGTx US, 450 East 29th Street, 14th Floor New York, NY 10016

Introduction

- Overall, rods outnumber cones by a ratio of 20:1 or greater in the retina. Defects are common in rods and lead to various ocular diseases.
- While multiple cone-specific promoters have been engineered, there is a lack of strong rod-specific promoters that could accelerate the clinical development of gene therapies to treat rod or rod-cone disorders as well as optogenetic applications.
- We sought to design novel promoters to drive expression specifically and at high levels in rods and to test them in both mouse and human photoreceptors.

Methods

- A first generation of rod-specific promoters were rationally designed by intersecting multiple genomic and epigenomic datasets.
- Several types of data were used, including DNA accessibility assays, transcription factor binding affinity, histone markers, validated transcription start sites and regions with cis-regulatory elements predicted to contact the promoter of interest.
- A second generation of promoters were rationally designed by combining elements from those promoters that drove strong and highly specific expression in human retinal organoids and mouse retina from the first generation of rod specific promoters.
- Promoter sequences from second generation were designed, cloned into an AAV backbone carrying eGFP and packaged into AAV5 or AAV7m8, alongside vectors carrying the commonly used rhodopsin kinase (RK or GRK1) promoter driving eGFP expression.
- To assess promoter activity, AAV7m8 vectors were used in human induced pluripotent stem cell (hiPSC)-derived retinal organoids. Three weeks post transduction organoids were dissociated into single cells for flow cytometry analysis or for immunohistochemistry.
- In parallel, promoter activity was assessed in murine retina. Wild type mice received subretinal injections with the AAV5 vectors. Four weeks post vector administration, eyes were harvested for immunohistochemical analysis and qPCR expression analysis to determine specificity and expression levels, respectively.

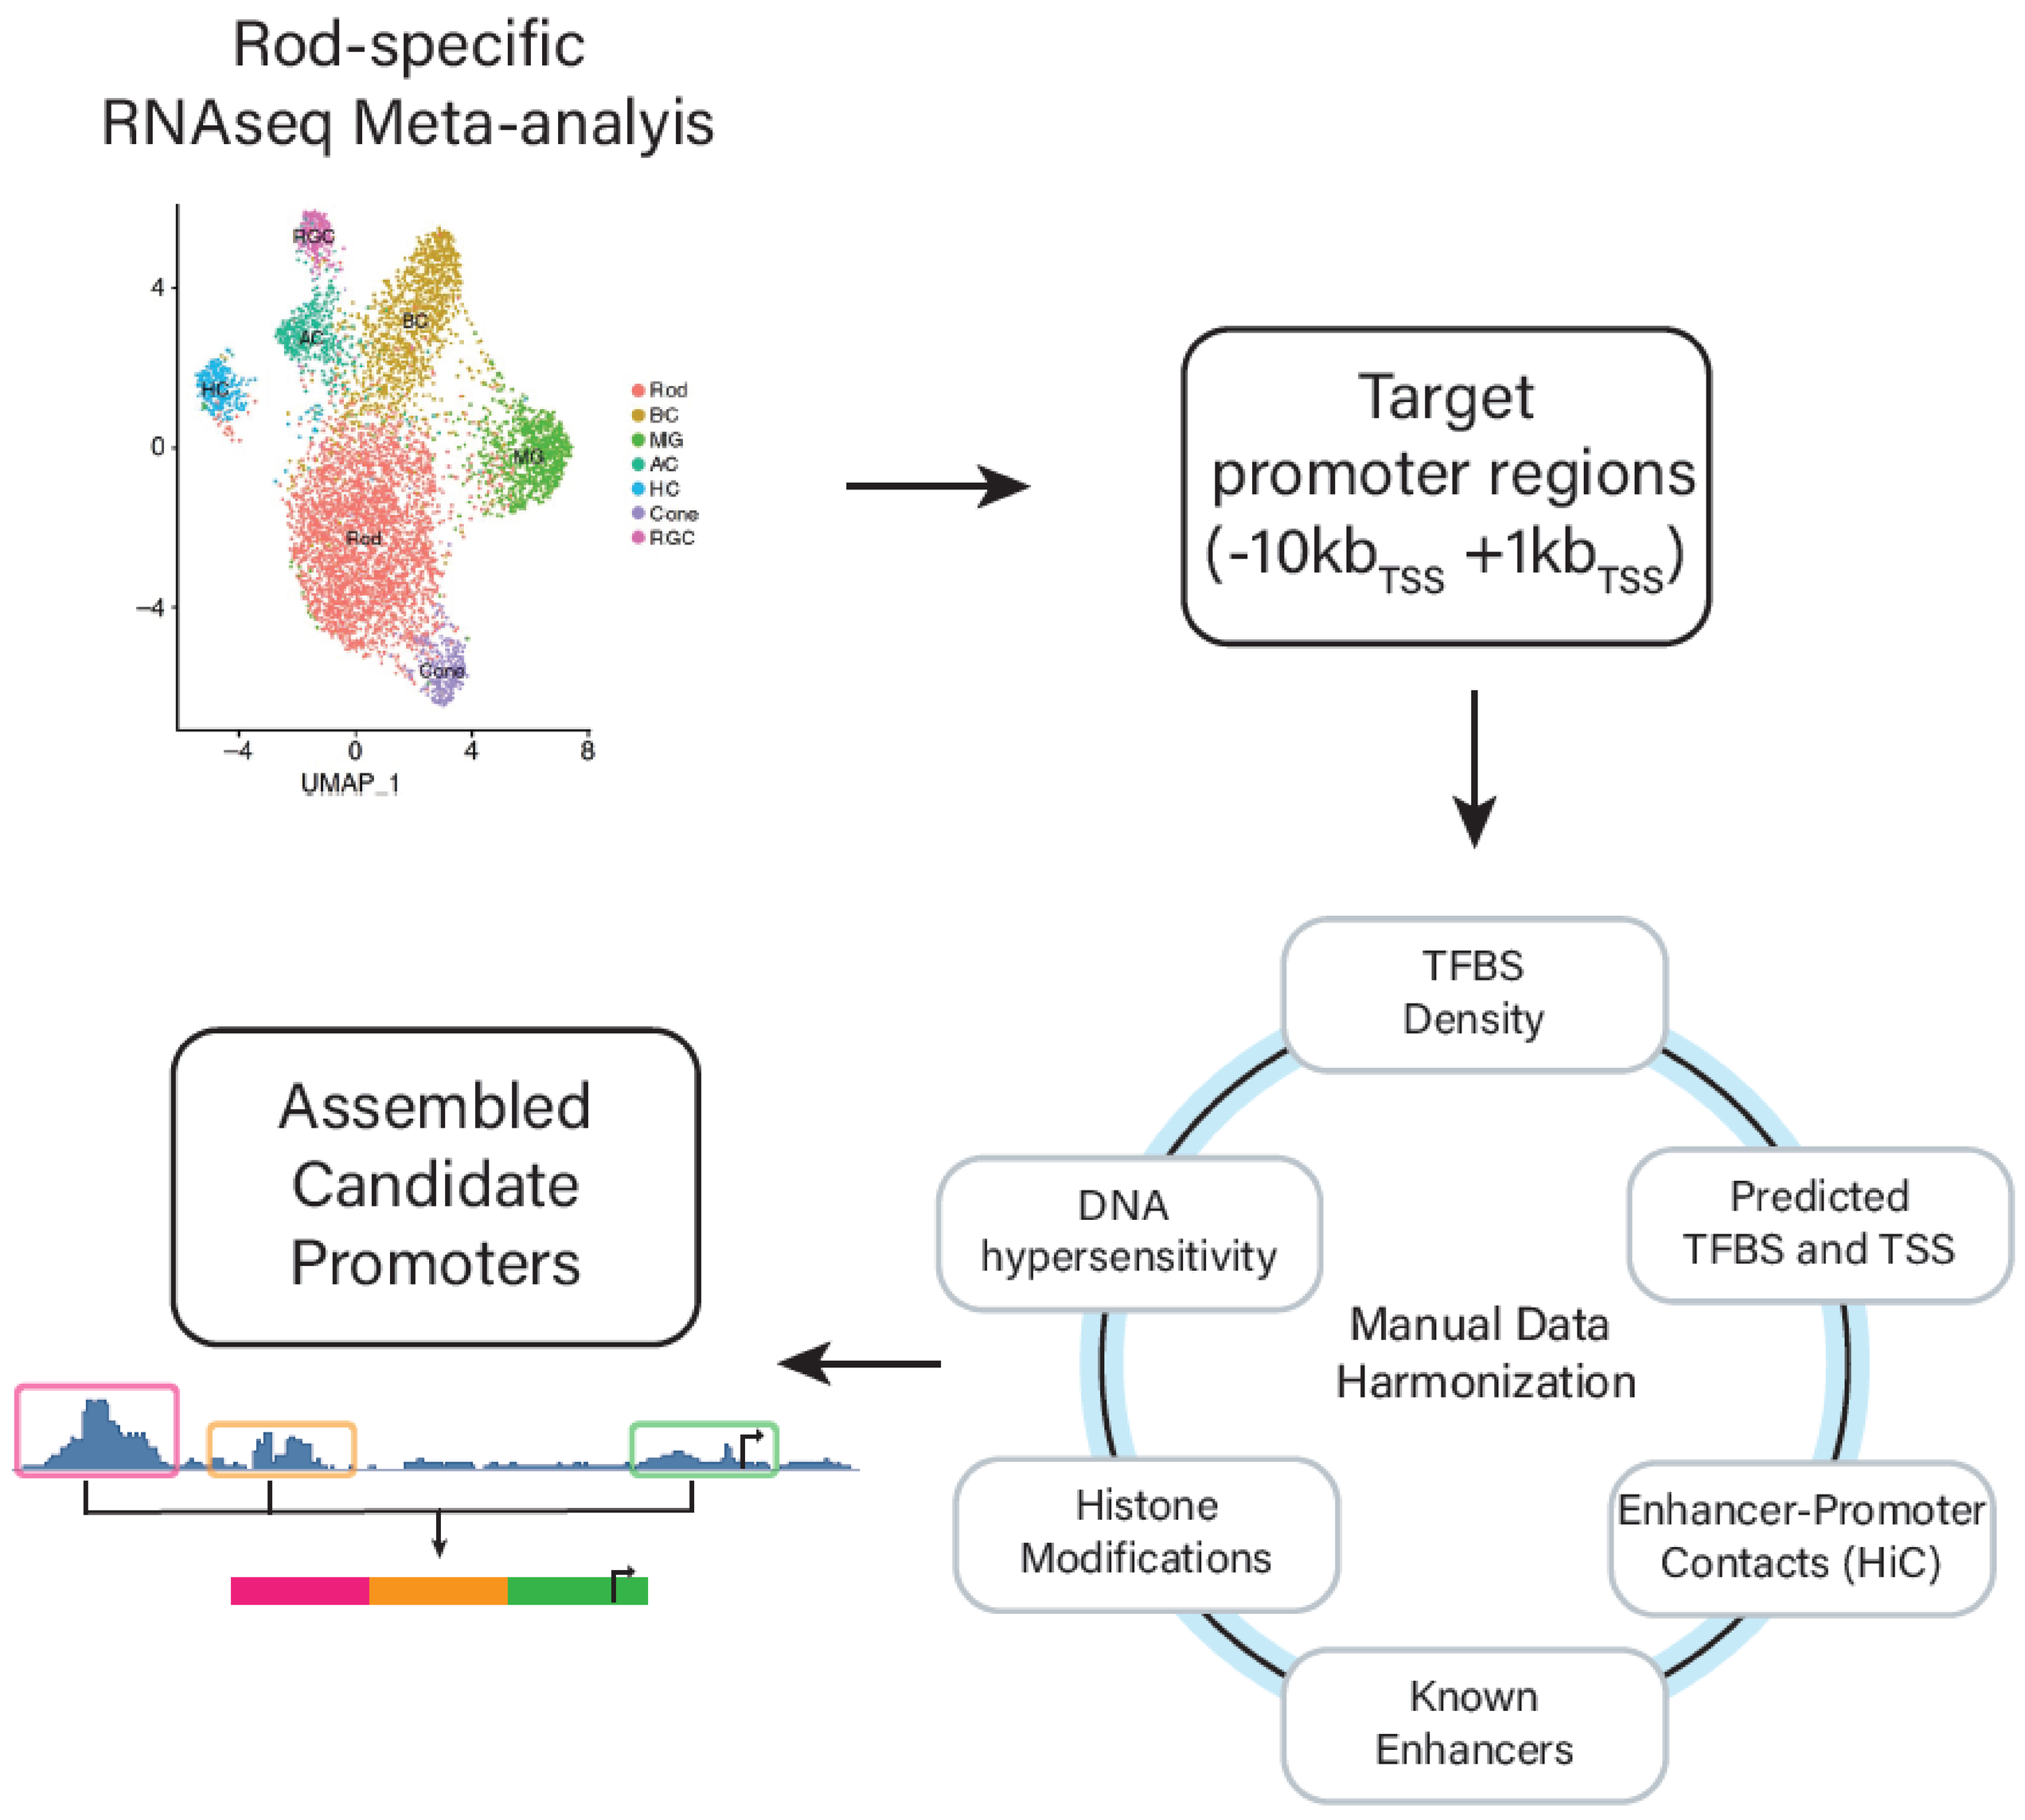
Rod-specific
RNAseq Meta-analyis
Rod
BC
MG
AC
HC
Cone
RGC
UMAP_1
Target
promoter regions
(-10kb$_{TSS}$ +1kb$_{TSS}$)
TFBS
Density
DNA
hypersensitivity
Predicted
TFBS and TSS
Manual Data
Harmonization
Histone
Modifications
Enhancer-Promoter
Contacts (HiC)
Known
Enhancers
Assembled
Candidate
Promoters

Conclusions

- Successful preclinical development of AAV vectors requires the use of promoters which are active in both human and small animal models in order to carry out meaningful assessments of both safety and efficacy.
- Target gene expression levels from the commonly employed RK promoter can be improved by rational design incorporating data from single cell RNA sequencing and other sources.
- In the murine retina, four promoters; 12,14,15 and 16 showed a marked increase (7-17 fold change) compared to the RK baseline in the expression levels and a strong rod-specific tropism (Fig.4,5). In human retinal organoids, five promoters; 12,13,14,15 and 16 showed a marked increase (6-9 fold change) compared to the RK baseline in the intensity of eGFP fluorescence and a strong rod-specific tropism (Fig.1,3).

Results: Promoter activity in hiPSC retinal organoids

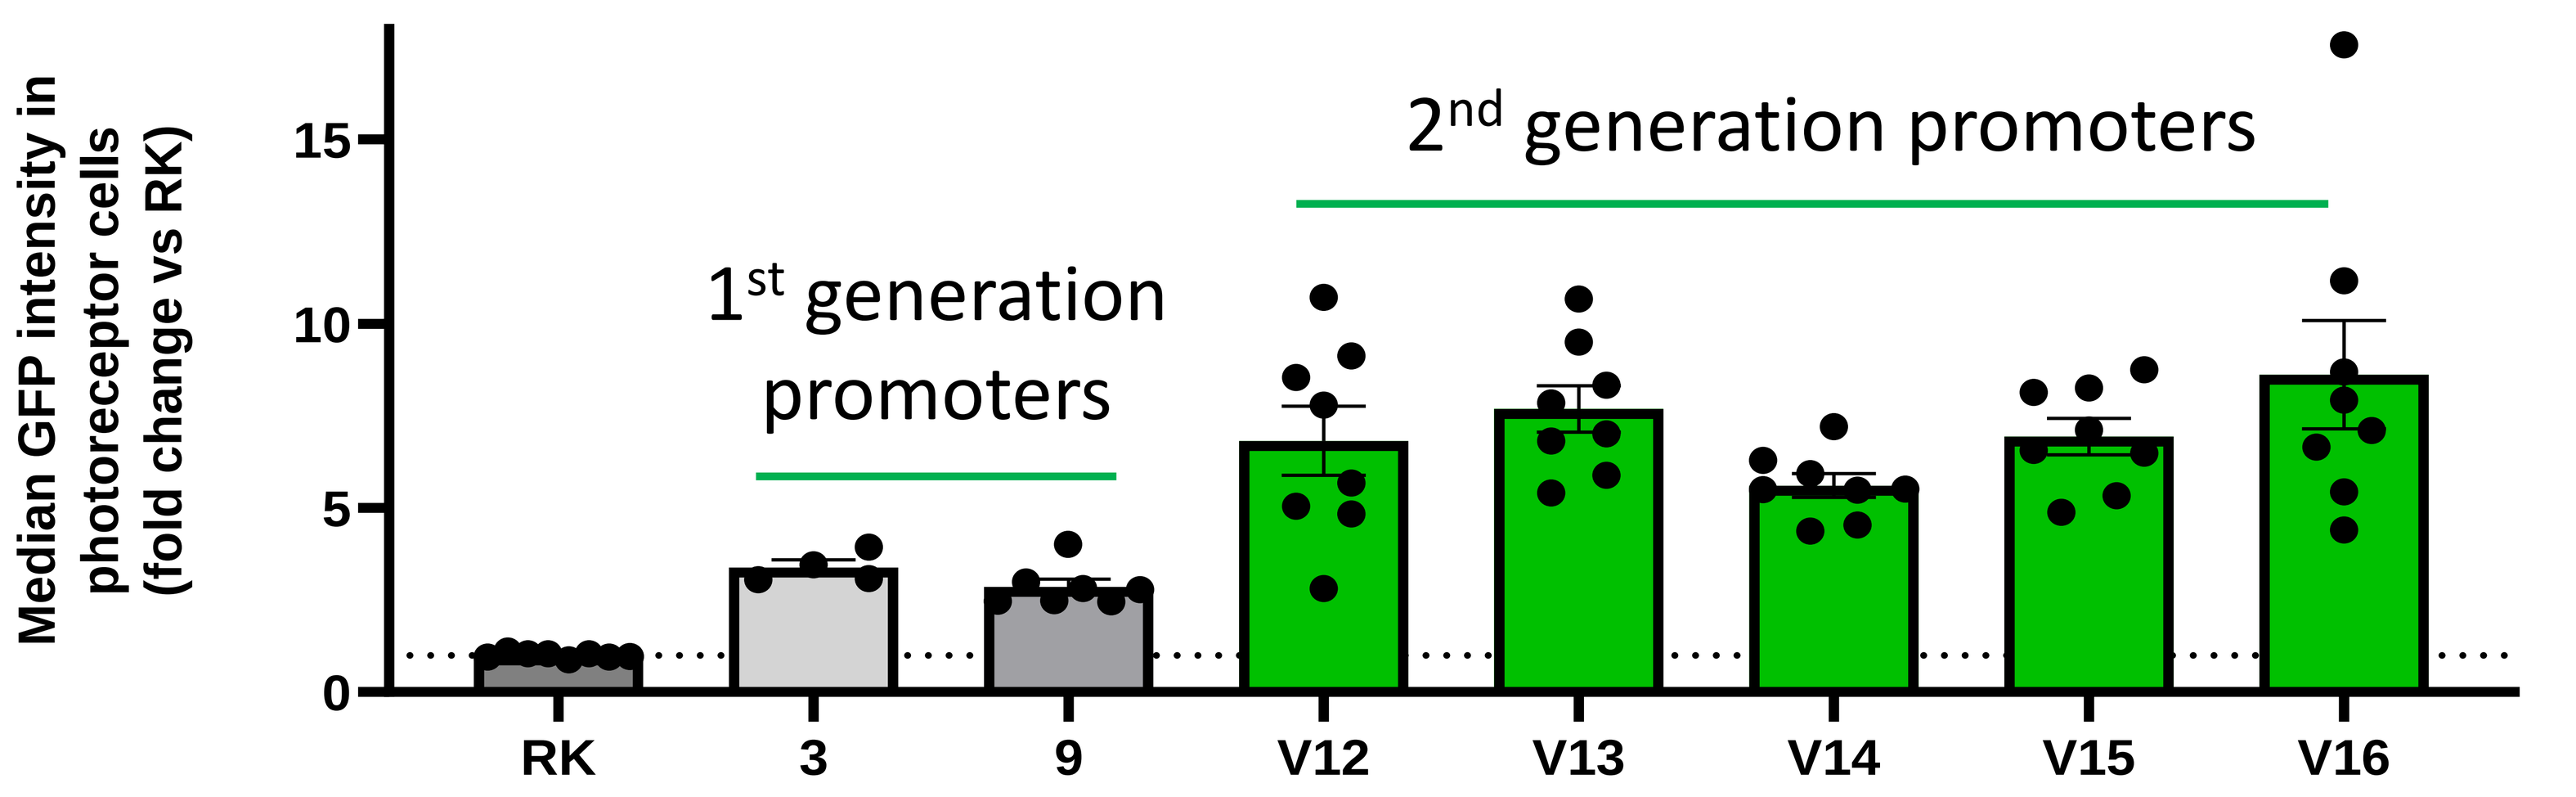
Median GFP intensity in photoreceptor cells (fold change vs RK)
15
10
5
0
1st generation promoters
2nd generation promoters
RK
3
9
V12
V13
V14
V15
V16

Fig 1. GFP expression in retinal organoids. Retinal organoids transduced with AAV7m8 vectors encoding GFP driven by commonly used RK and novel rod promoters. Median GFP intensity in CD73+ve photoreceptors was determined by flow cytometry, Data are expressed as a fold change relative to expression from the RK promoter. Bars show the mean MFI +/- SEM of n= 8 organoids from two independent transduction experiments. 1st generation promoters increase median GFP intensity in photoreceptor cells by 2-3 fold. 2nd generation promoters V12-16 increase expression 5-9 fold.

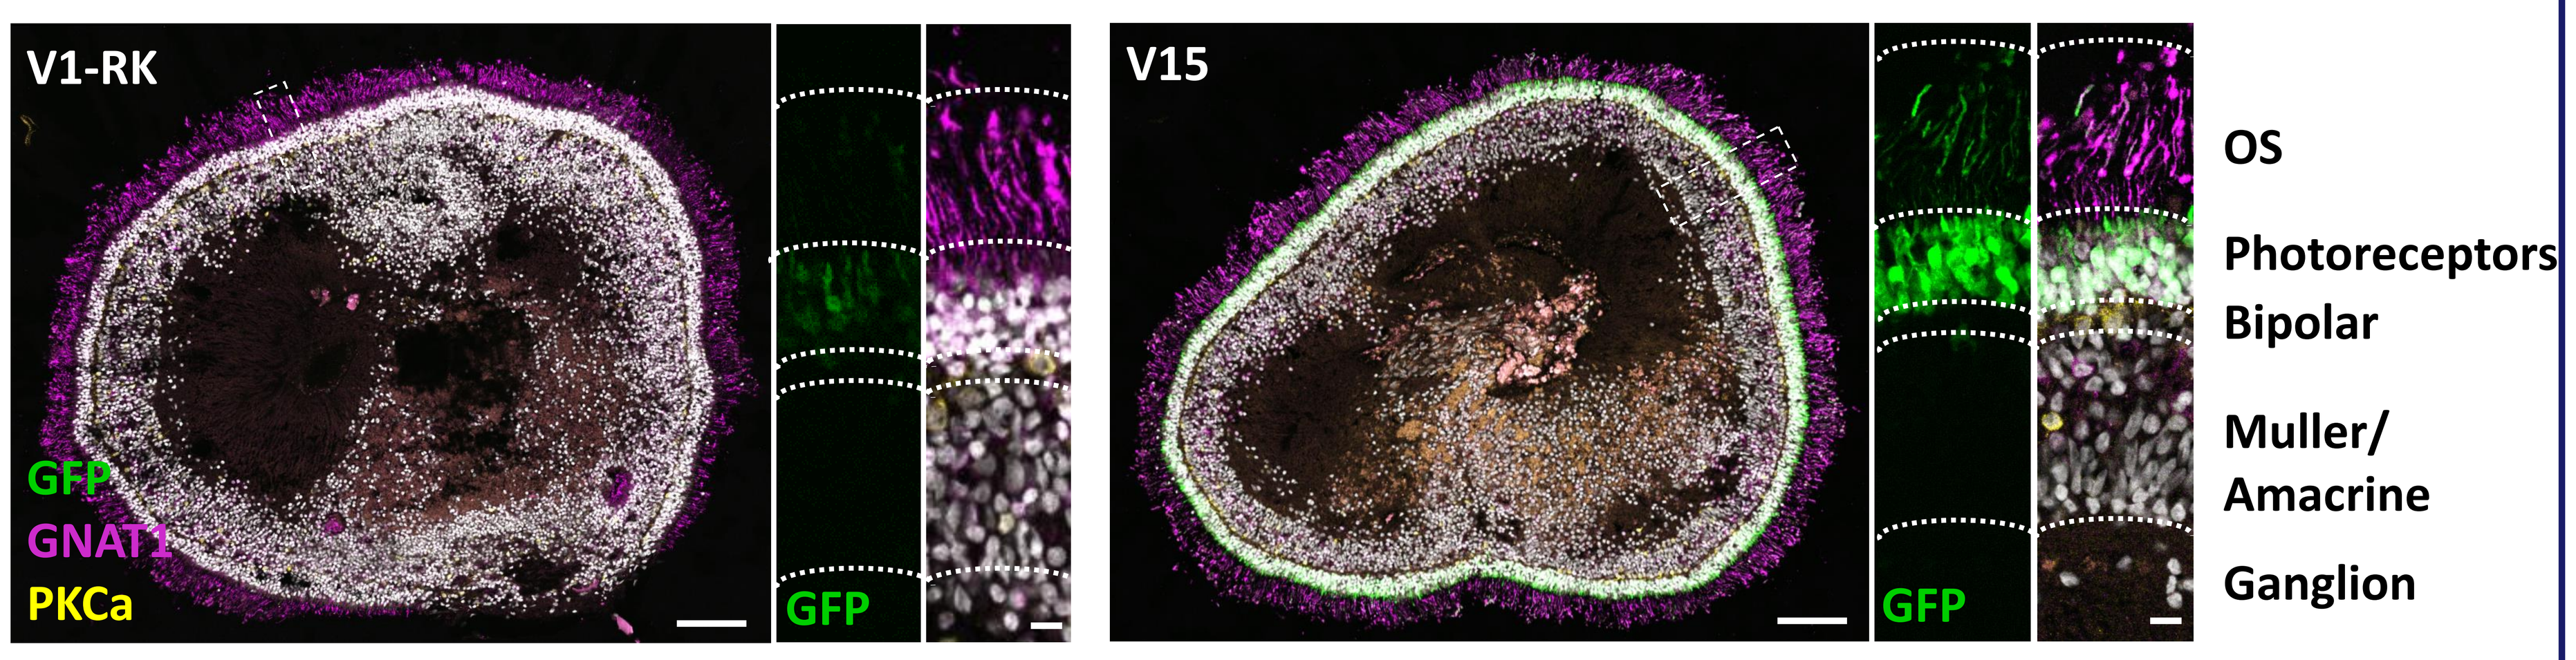
V1-RK
GFP
GNAT1
PKCa
GFP
V15
GFP
OS
Photoreceptors
Bipolar
Muller/
Amacrine
Ganglion

Fig 2. Rod promoters lead to rod photoreceptor specific GFP expression. Cryosections of retinal organoids transduced with AAV7m8 RK-GFP and novel promoter V15 express GFP in photoreceptor layers. 2nd generation rod promoter V15 demonstrates increased GFP expression in photoreceptor cell layer (cell bodies and outer-segments) without compromising specificity. Scale bar = 100um and 10um (insert).

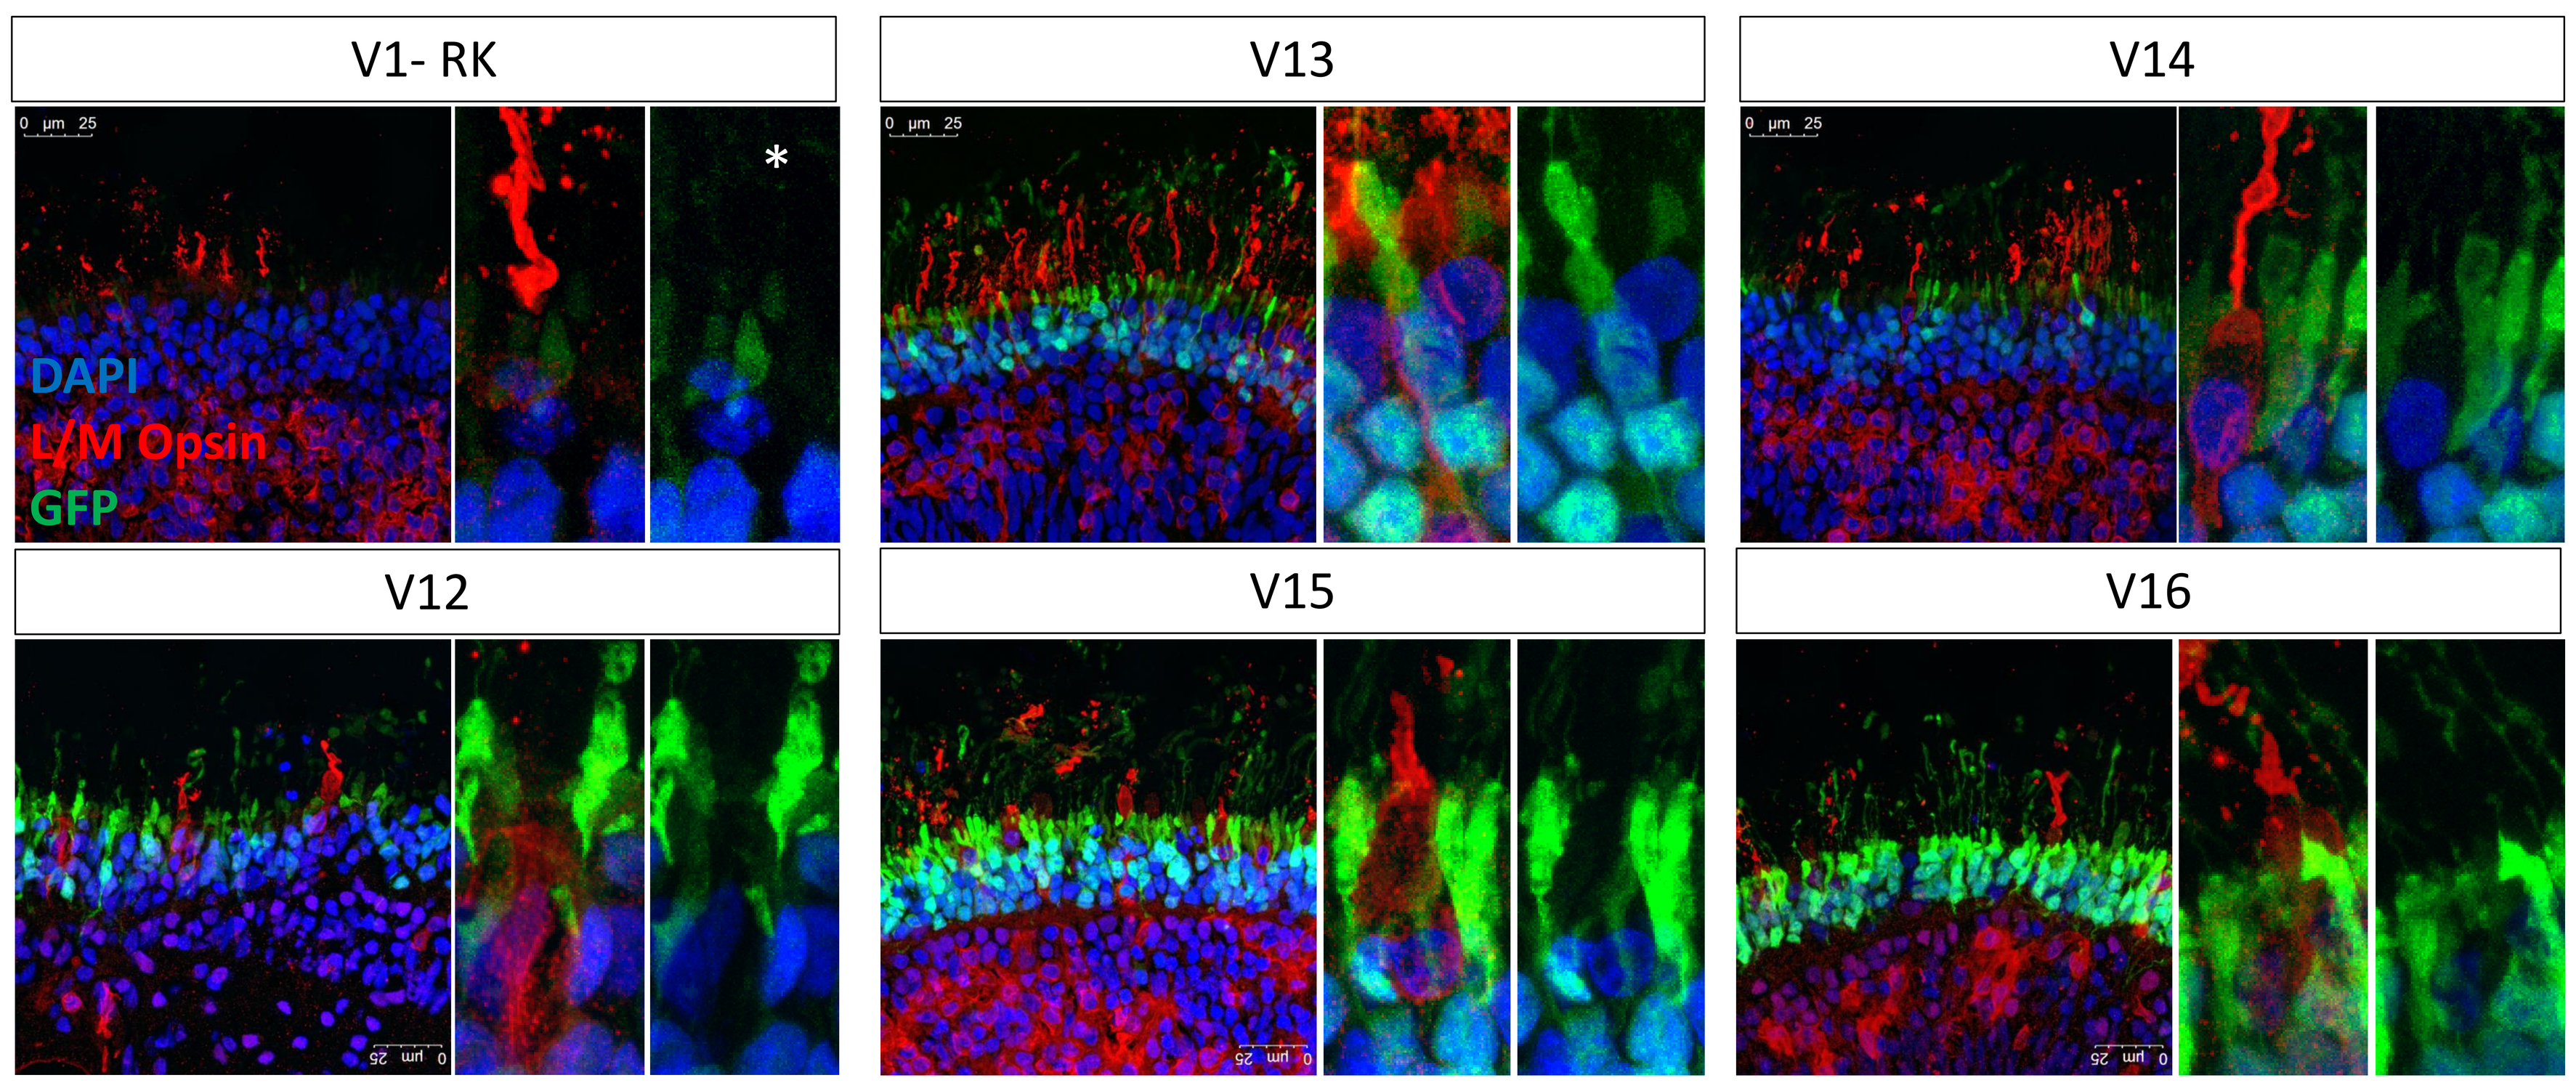
V1- RK
V13
V14
DAPI
L/M Opsin
GFP
*
V12
V15
V16

Fig 3. GFP is not expressed in cone photoreceptor cells. Immunohistochemistry on AAV7m8 transduced organoid frozen sections. L/M Opsin labels cone cells. GFP is not expressed in cone cells showing the specificity of promoter activity to rod cells. *Green channel in V1-RK insert enhanced to show GFP. Scale bar =25µm.

Results: Promoter activity in murine retina

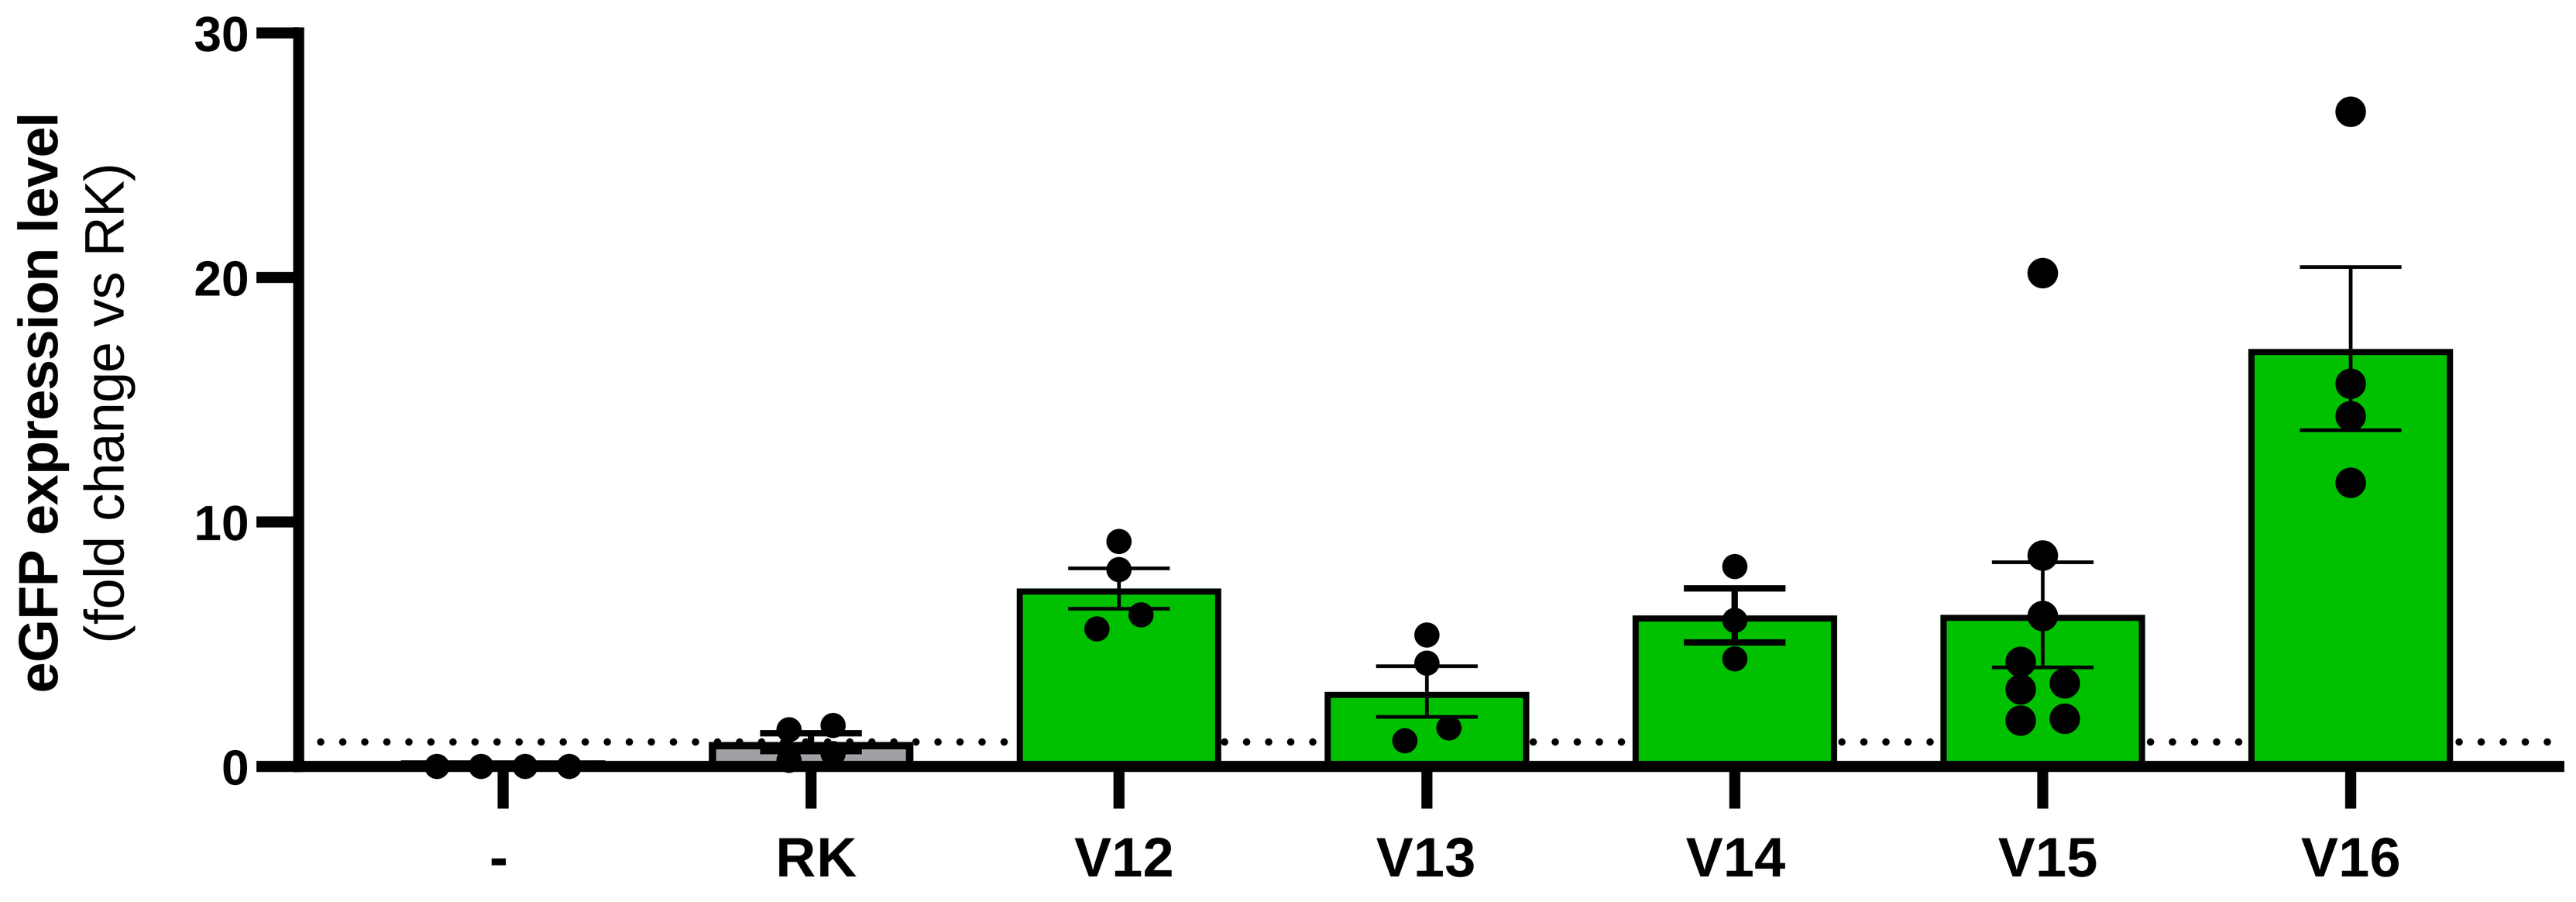
eGFP expression level
(fold change vs RK)
30
20
10
0
-
RK
V12
V13
V14
V15
V16

Fig 4. qPCR analysis of eGFP expression in mouse retina. Adult WT C57BL/6 mice were subretinally injected (4x10^{9} VG/eye) with AAV5 vectors expressing GFP under a range of rod-specific promoters. Retinas were dissected 4 weeks post injection and analyzed using on-target qPCR. 2nd generation promoters V12-16 increase expression by 8 fold. V16 was the best performer of the tested promoters. Data displayed are normalized to eGFP expression levels driven by AAV5 containing an RK promoter (dashed line). Mean +/- SEM (n=4-8 per group).

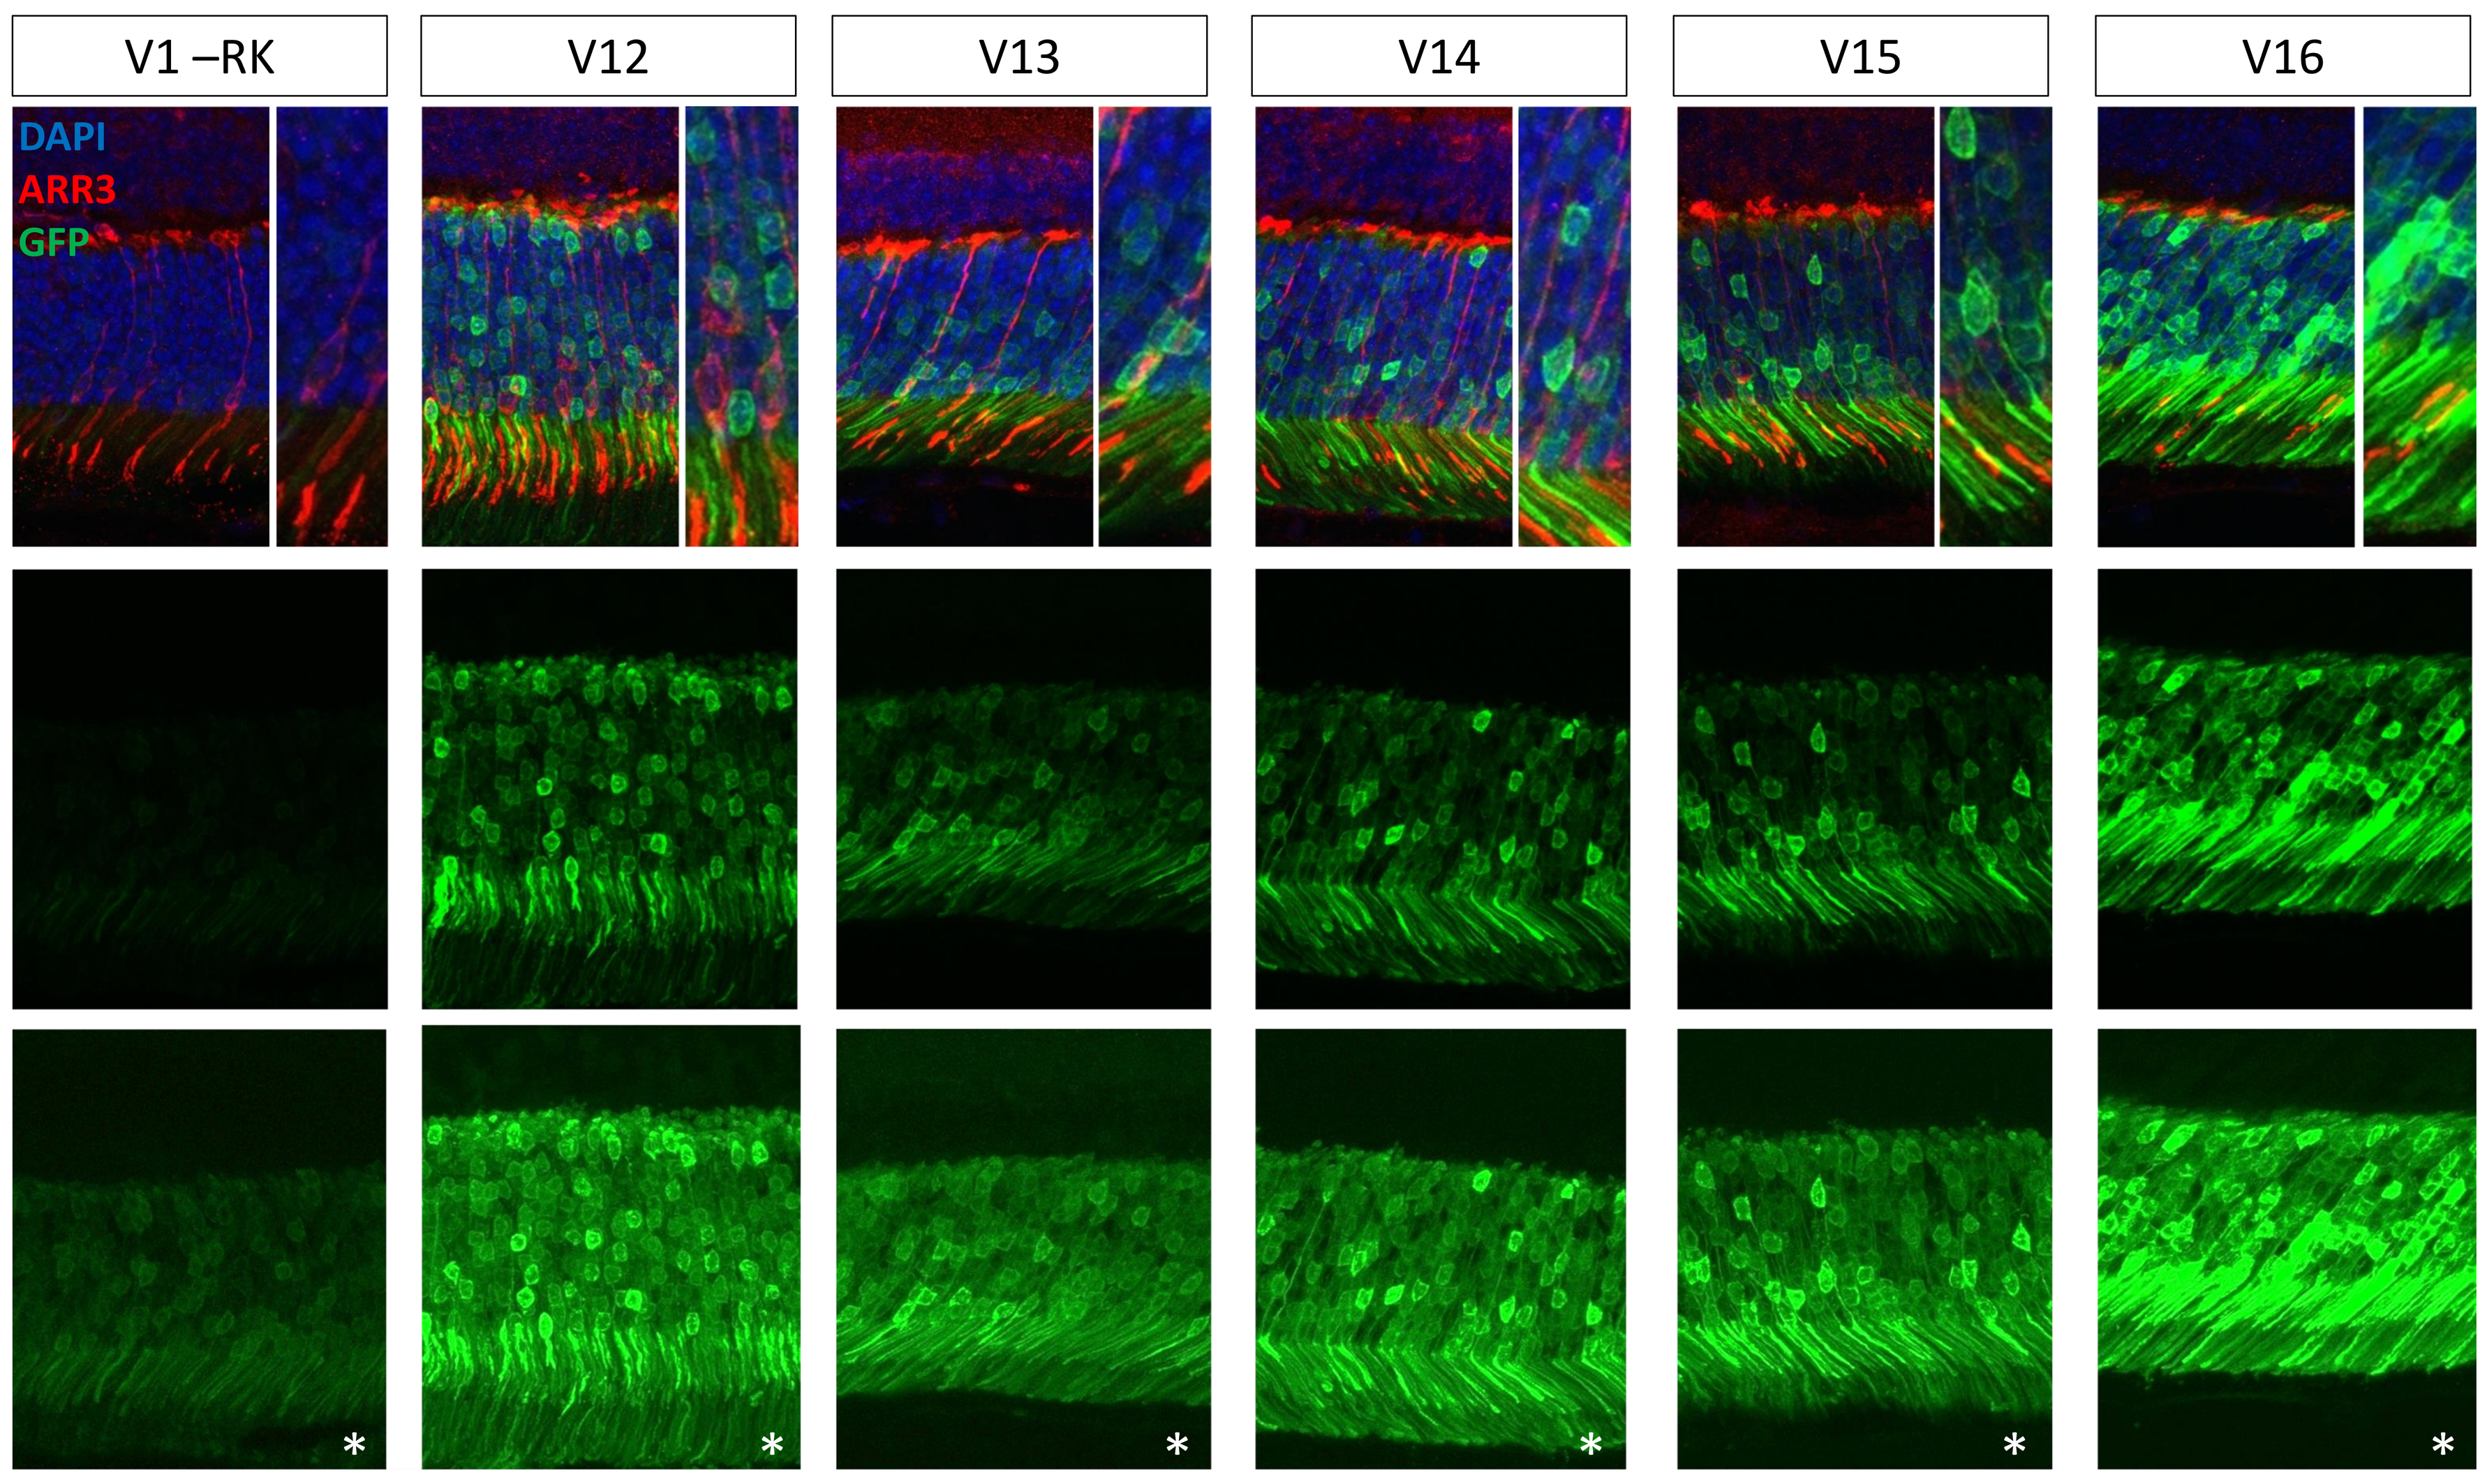
V1 –RK
V12
V13
V14
V15
V16
DAPI
ARR3
GFP
*

Fig 5. GFP is not expressed in cone photoreceptor cells. Immunohistochemistry on AAV5 transduced mouse retina frozen sections. Arrestin 3 (ARR3) labels cone photoreceptor cells. GFP is not expressed in cone cells V12-V16 showing the specificity of promoter activity to rod cells. GFP expression driven by RK promoter is very weak, to visualize "dim" expressing cells contrast/brightness of images within bottom panel has been increased (*). V16 drives the strongest expression of the tested promoters in rods.