Poster #167

Potency assay cell line development for ocular gene therapy vectors

Gemma Estrada Girona, Tassos Georgiadis *(MeiraGTx London)*

When producing good manufacturing practice (GMP) adeno-associated virus (AAV) drug products for clinical or commercial use, the ability to assess their potency is an essential part of release and stability. Cell-based potency assays are a regulatory requirement for commercialisation of AAV gene therapies but poor AAV *in vitro* transducibility as well as inability to test expression due to the use of tissue-specific promoters in the viral vector hinders the development of efficient and robust assays. Similarly, basic pre-clinical research, where easy-to-implement tests for faster paced experiments and iterations are desirable, suffers from the same limitations. We therefore set out to examine ways with which to increase vector transducibility as well as establishing ocular promoter trans-activation in the most commonly used cell lines (HEK293 and HeLa). We assessed different methods to increase the transducibility of AAV5 and AAV8 vectors as well as trans-activate photoreceptor-specific (Rhodopsin kinase) and RPE-specific (RPE65) promoters. In dose-ranging experiments, we identified potent dCas9-mediated trans-activators for both promoters, and established exogenous factor supplementation that increased the transducibility of both capsids multiple folds over baseline. Further studies will enable us to combine these in an *in vitro* cell-based potency assay platform for GMP batch release and stability testing.

Increasing vector transducibility

Over-expression of AAVR

KIAA0319L (encoding for AAVR protein) is one of the most important genes involved in AAV infection[1]. Its knock-out hinders infectivity[1,2]. Thus we hypothesized whether its over-expression could lead to enhanced transduction and tested it via transient transfection and CAG-EGFP vectors.

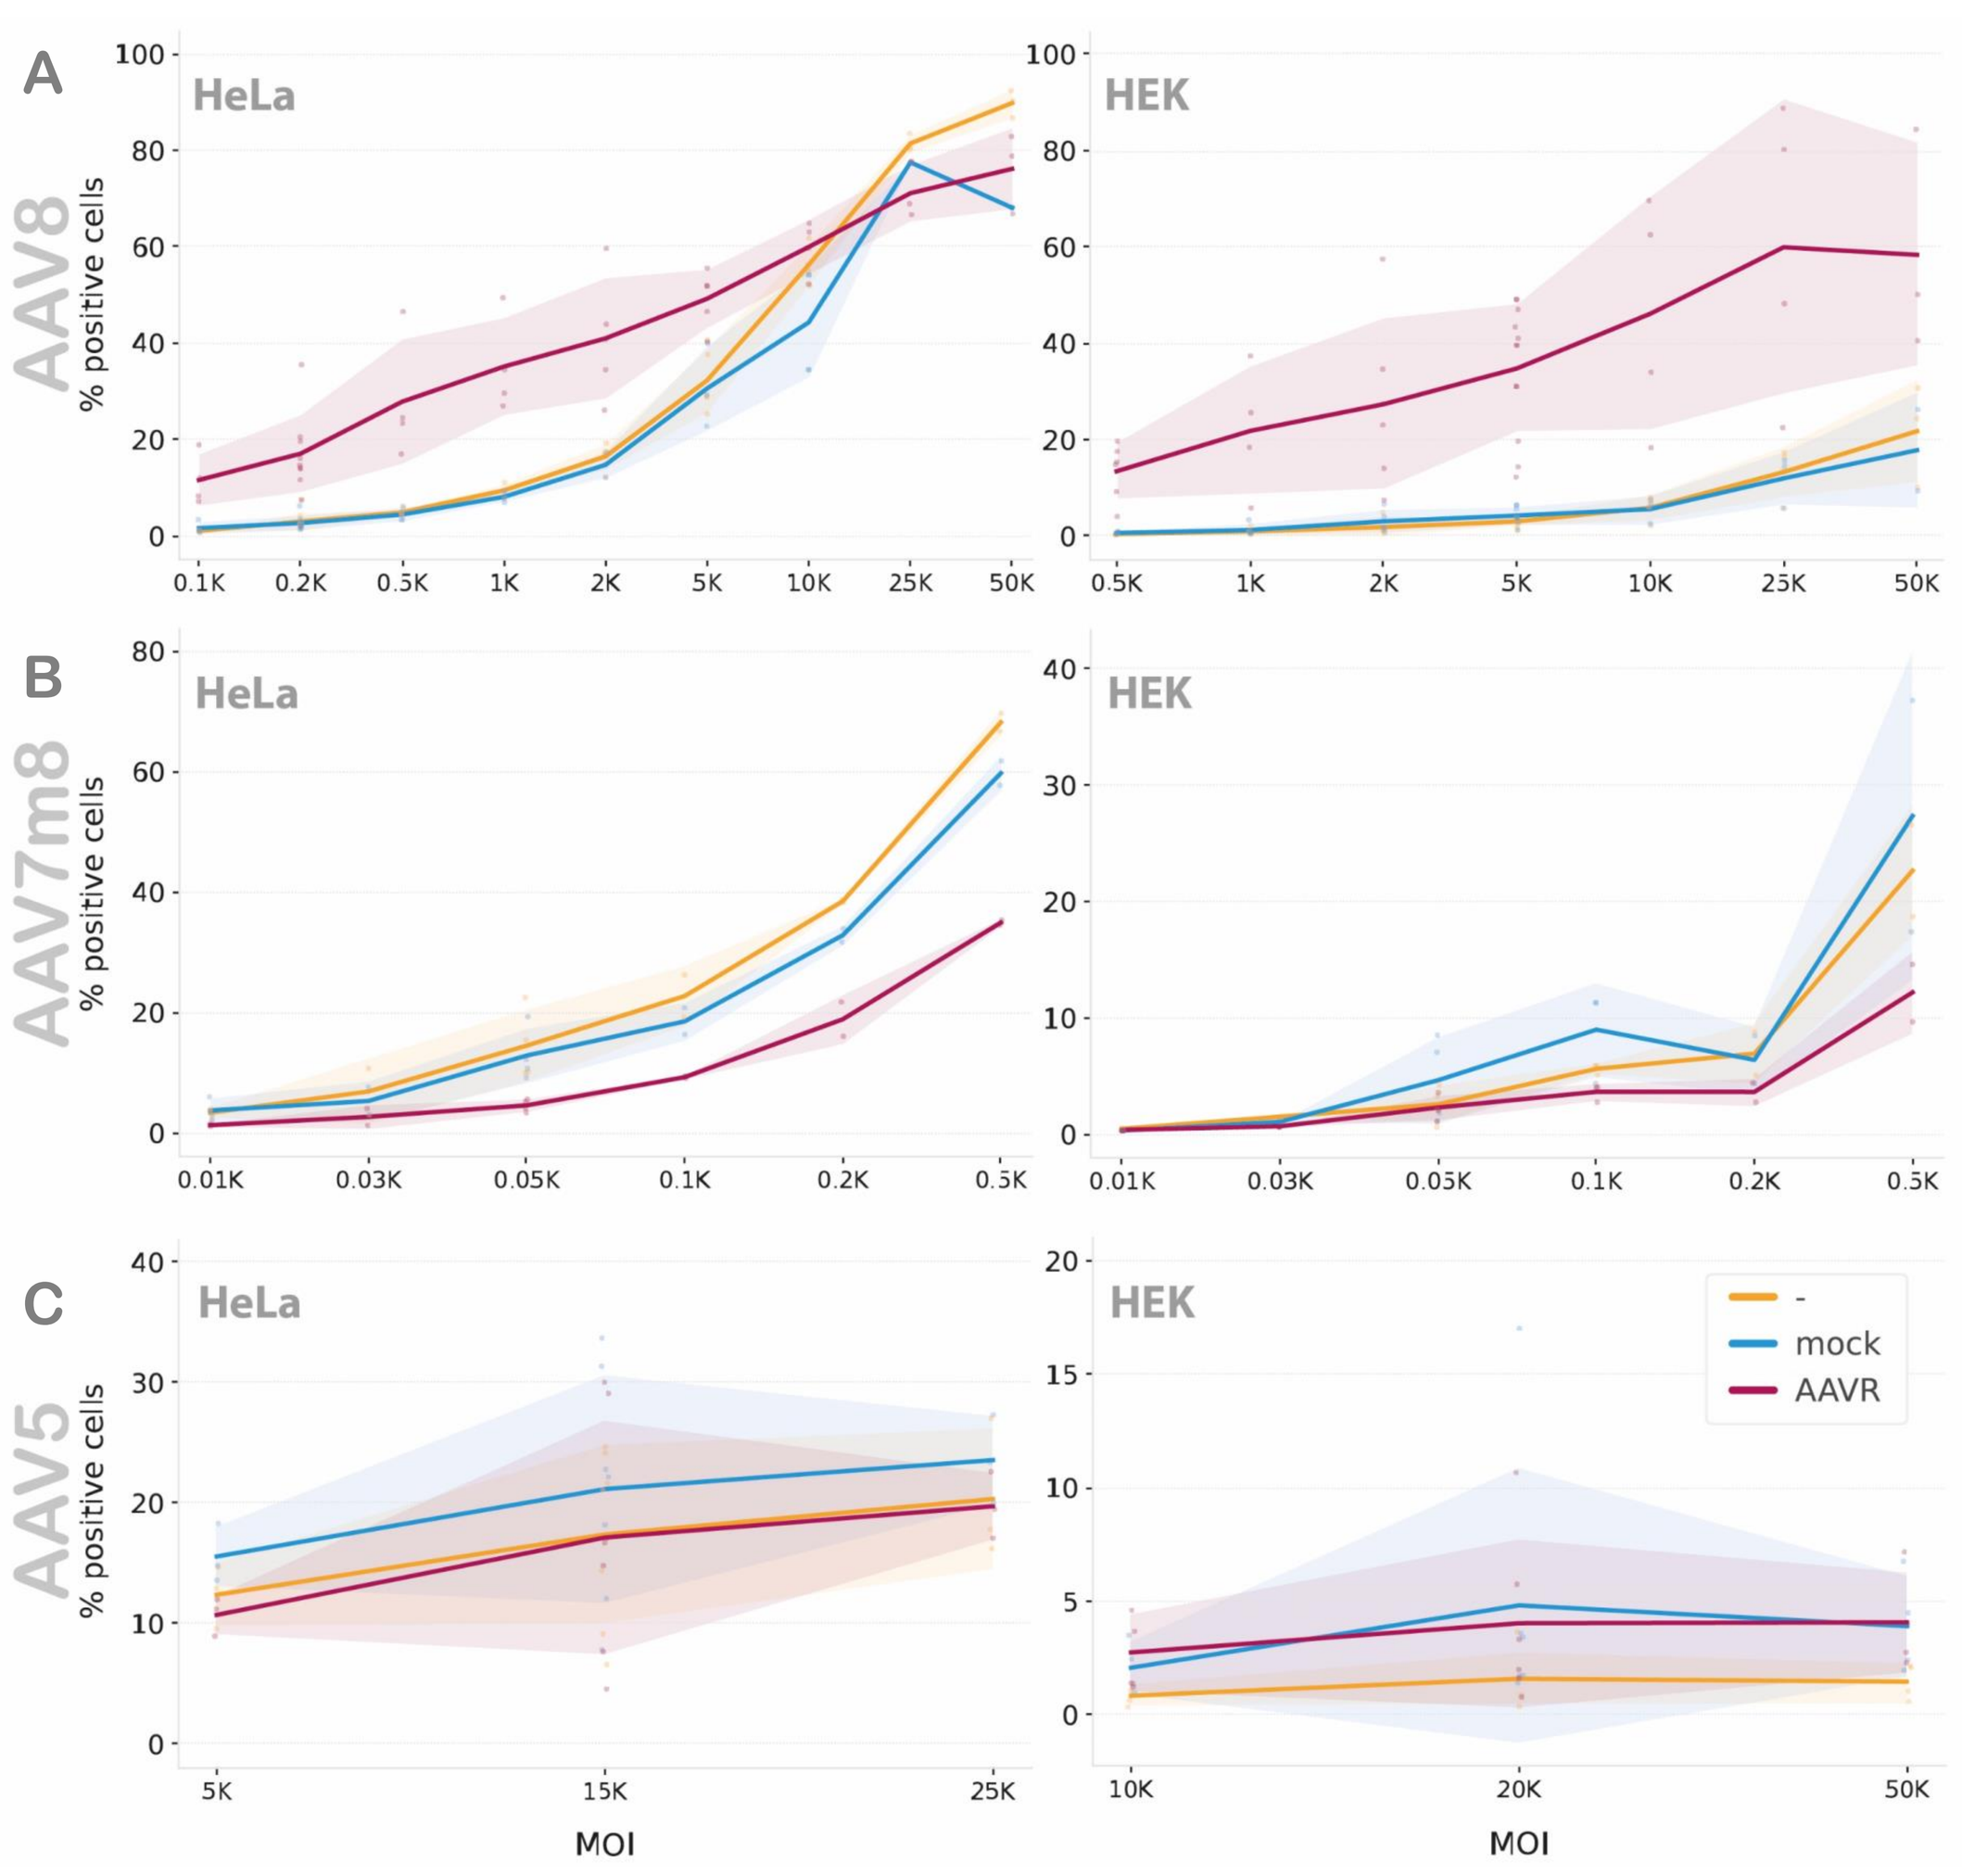
A
AAV8
HeLa
HEK
B
AAV7m8
C
AAV5
% positive cells
-
mock
AAVR
MOI

Over-expression of *AAVR enhances AAV8 transducibility in both HEK293 (HEK) and HeLa* cell lines (Fig. 1A), with fold increases ranging from 2.7 to 34 (high to low MOI) in HEK and 1.5 to 11 in HeLa. In HeLa however, additional receptor does not produce beneficial effects for the highest tested MOIs. A similar effect can also be observed for AAV7m8, a much better transducer, not benefitting from additional AAVR (Fig. 1B). We speculate that this might be due to a very efficient baseline transduction (note difference in MOI needed to achieve similar levels of positive cells). Surprisingly though, over-expression of AAVR in either cell line did *not result in higher levels when using AAV5*, the worst transducer tested here (Fig. 1C).

Attempts at modifying the AAVR construct and over-expression of other factors important in the transduction process did not yield any enhancements in transduction *(data not shown).*

Figure 1. Effect of AAVR over-expression in HeLa and HEK cells upon AAV5, 7m8 and 8 transduction. Plotted as mean (independent experiments with n= 3-12 for AAV8 1-4 for 7m8 and 4-7 for AAV5 [- and AAVR]) with shaded SD. Cells are: seeded (day0), transfected (day1), transduced (day2) and analysed by flow cytometry (day4). "mock" (pUC19) is included to control for transfection effect with same amount of DNA as AAVR transfection.

Chemical supplementation

Different types of molecules have been explored to enhance transgene expression upon AAV transduction: 1- Topoisomerase inhibitors (e.g. etoposide[3,4,5] and doxorubicin[6,7]) 2- DNA synthesis inhibitors (e.g. hydroxyurea[3,8]) 3- Proteasome inhibitors (e.g. LLnL[6]) 4- Metal ions[9].

In our hands, metal ions had no beneficial effects on transduction with AAV8 or AAV7m8 in HeLa, COS-7 and HEK cells *(data not shown)*. In addition, due to high toxicity, reproducibility was very challenging, making them unsuitable for a robust potency assay in these cell lines.

We then tested doxorubicin, a known chemotherapeutic which has been shown to enhance transduction of AAV2 and AAV5 in human airway epithelia and mouse lungs[6], in mice, cat and monkey cortex with different serotypes[7], and in rat striatum[10] and retinal cells[11] with AAV2.

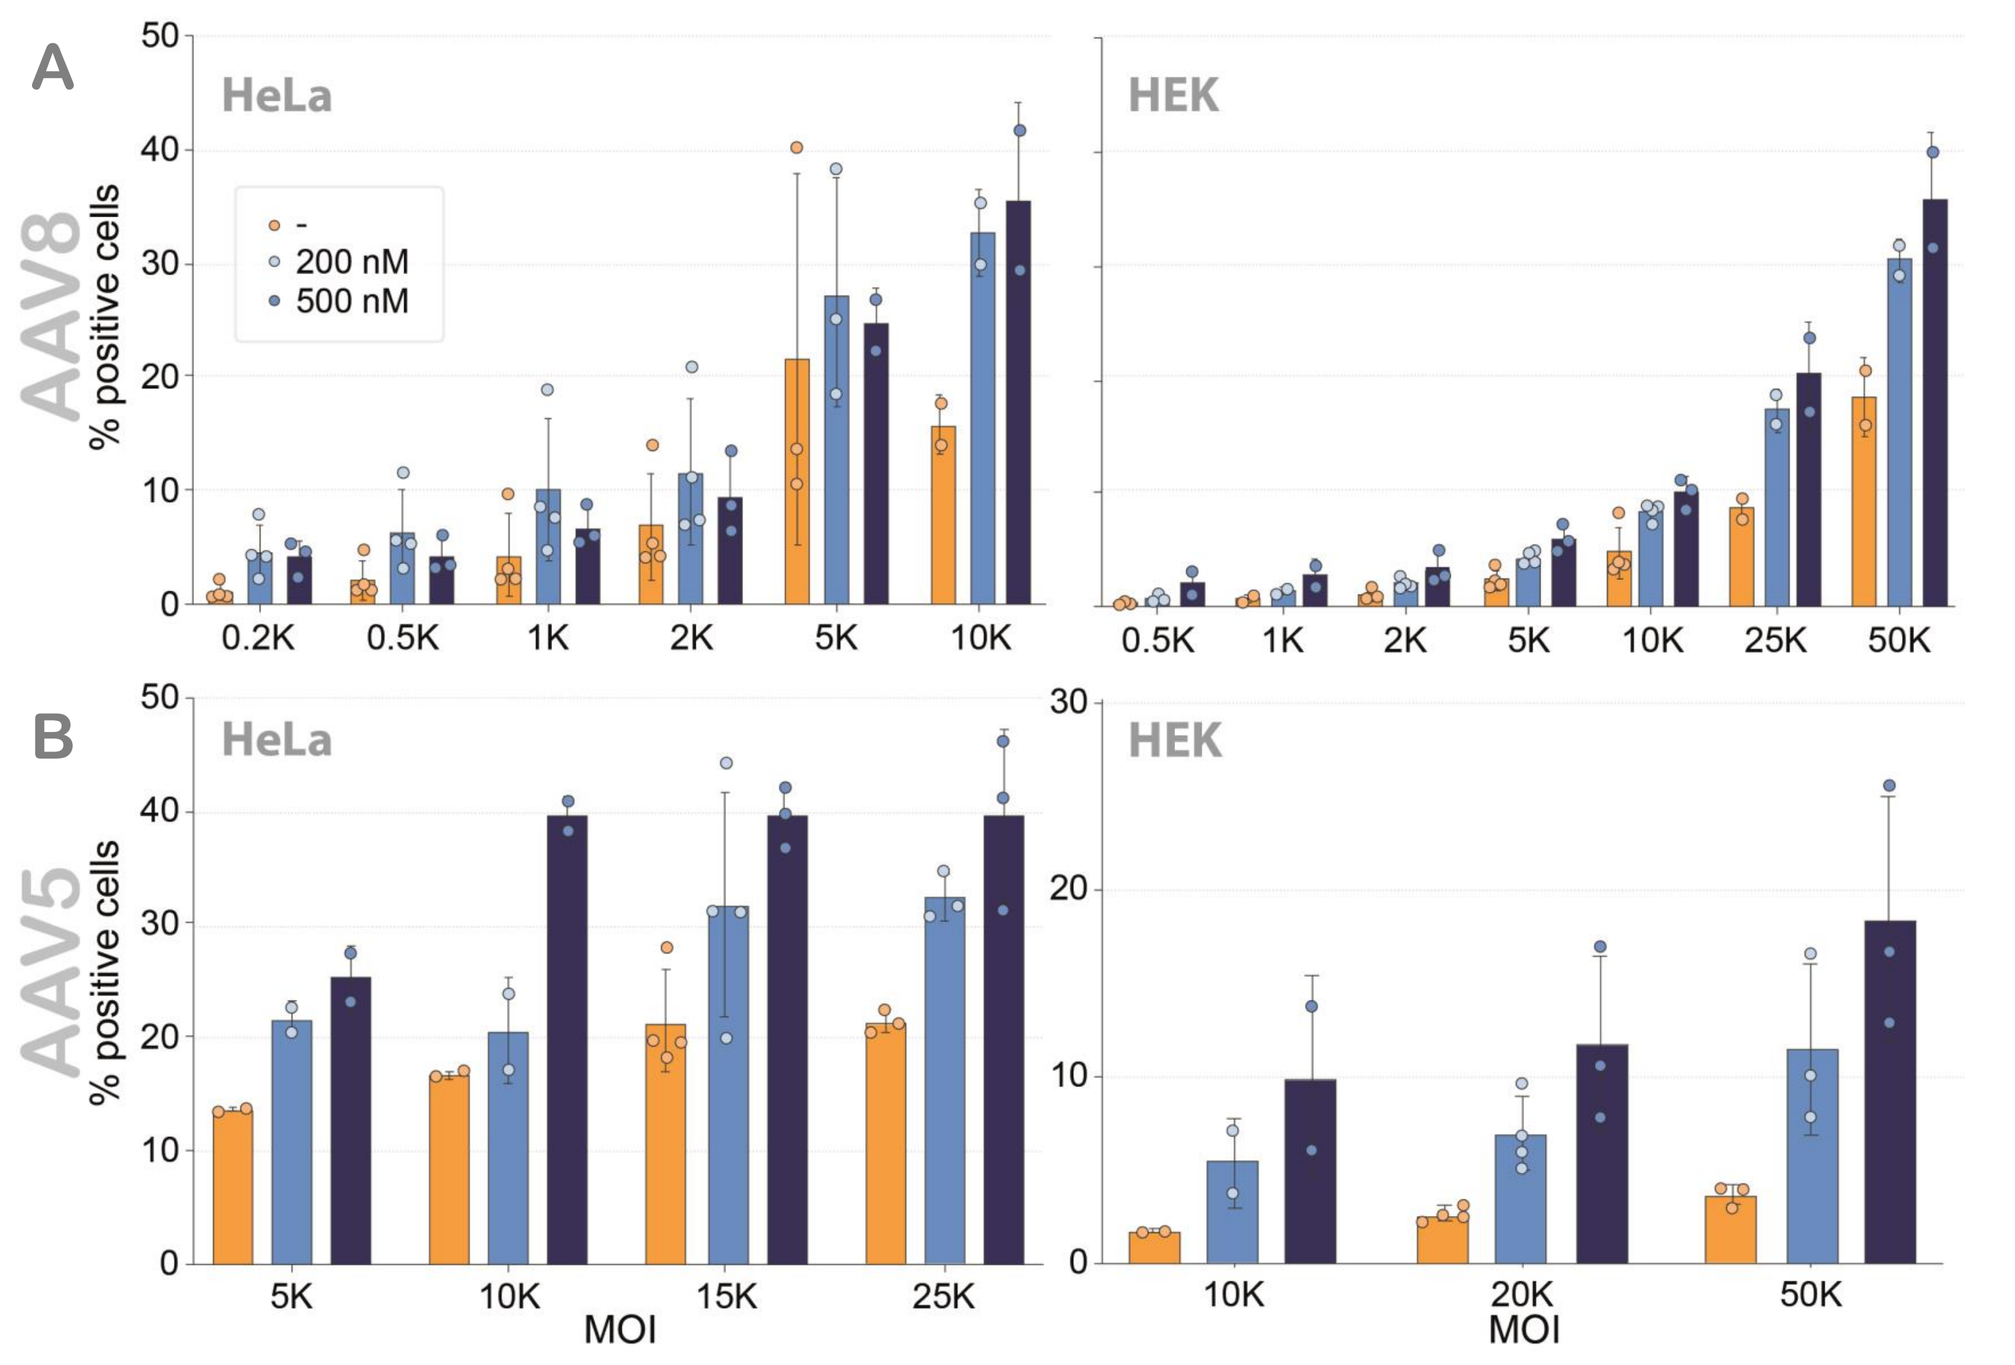
A
AAV8
HeLa
HEK
-
200 nM
500 nM
B
AAV5
% positive cells
MOI

Addition of doxorubicin at two different concentrations in both HeLa and HEK cells resulted in a *consistent increase in transgene expression for both AAV8* (Fig. 2A) *and AAV5 transductions* (Fig. 2B). Fold changes averaged at ~2 for HeLa and at ~2-5 for HEK. It should be noted that cell morphology changed dramatically in all cases, but it did not lead to cell death within the timeframe tested here.

Figure 2. Effect of doxorubicin in HeLa and HEK cells upon AAV5 and 8 transduction. Plotted as mean with SD. Cells are: seeded (day0), transduced (day1), treated (day2) and analysed by flow cytometry (day4).

Ocular promoter trans-activation

Therapeutic vectors often carry tissue or cell type specific promoters which are challenging to be tested *in vitro*. Approaches based on CRISPR/dCas9 transcriptional activation, where a catalytically inactive variant of Cas9 is fused to effector domains, have arisen in the last few years. In this study, we use a transactivation complex (by Chavez and colleagues[12]) termed dCas9-VPR (dCas9 hereafter). We test the system using AAVs harboring an EGFP under control of the promoter of choice and assess the level of trans-activation based on percent of positive cells.

Human rhodopsin kinase promoter (hRK)

hRK is active only in photoreceptor cells. To target dCas9 we used previously published gRNAs[13], (introduced together via transient transfection). Trans-activation was achieved efficiently with AAV7m8 in both cell lines with low MOIs (Fig. 3A). The system also *worked with AAV8*, although with very low efficiency in HEK cells (Fig. 3B). Transduction levels with *AAV5 in HeLa cells were low* and had poor dose effects (Fig. 3C). hRK could not be successfully activated in HEK with AAV5 transduction (Fig. 3C).

Human retinal pigment epithelium RPE65 promoter (NA65)

NA65 is active only in RPE cells. We designed and tested 8 gRNAs to target the dCas9-VPR complex to the promoter *(data not shown)*, combining the two best in the final construct. We also noticed, that the basal expression level of NA65, although very low, was 2-3 times higher than that of hRK in both cell lines. When the system was tested with AAV8, we achieved *similar levels of trans-activation as those for hRK* (Fig. 4A). For AAV5, expression levels were again much lower, although trans-activation was slightly more efficient than in the hRK case (Fig. 4B).

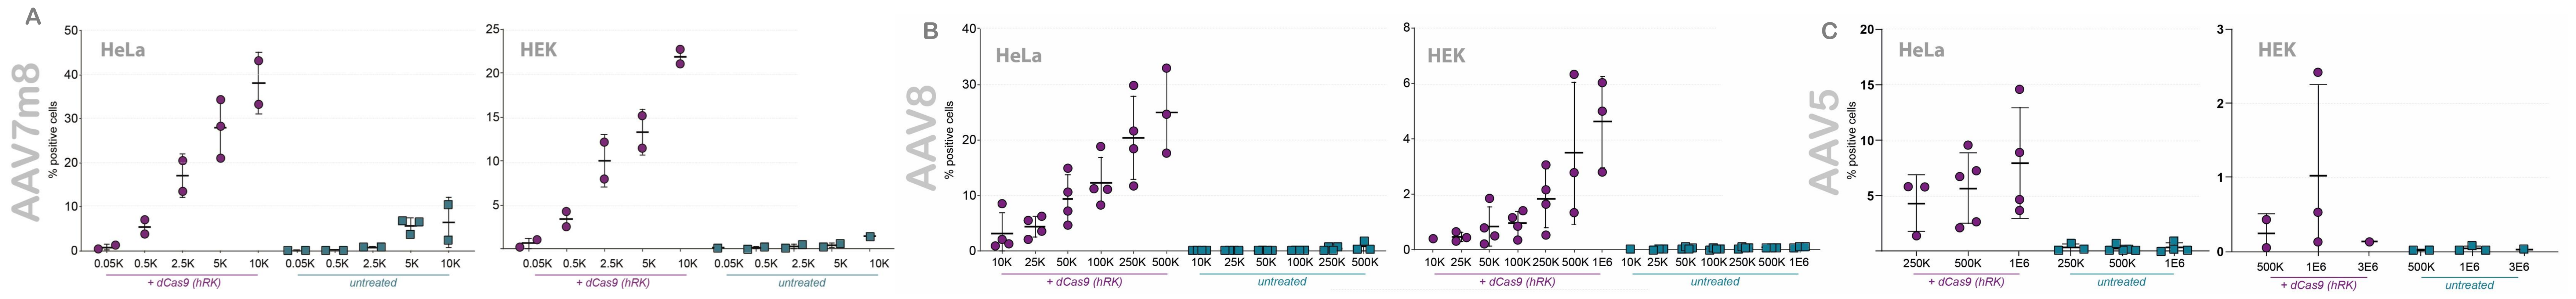
A
AAV7m8
B
AAV8
C
AAV5
HeLa
HEK
% positive cells
+ dCas9 (hRK)
untreated

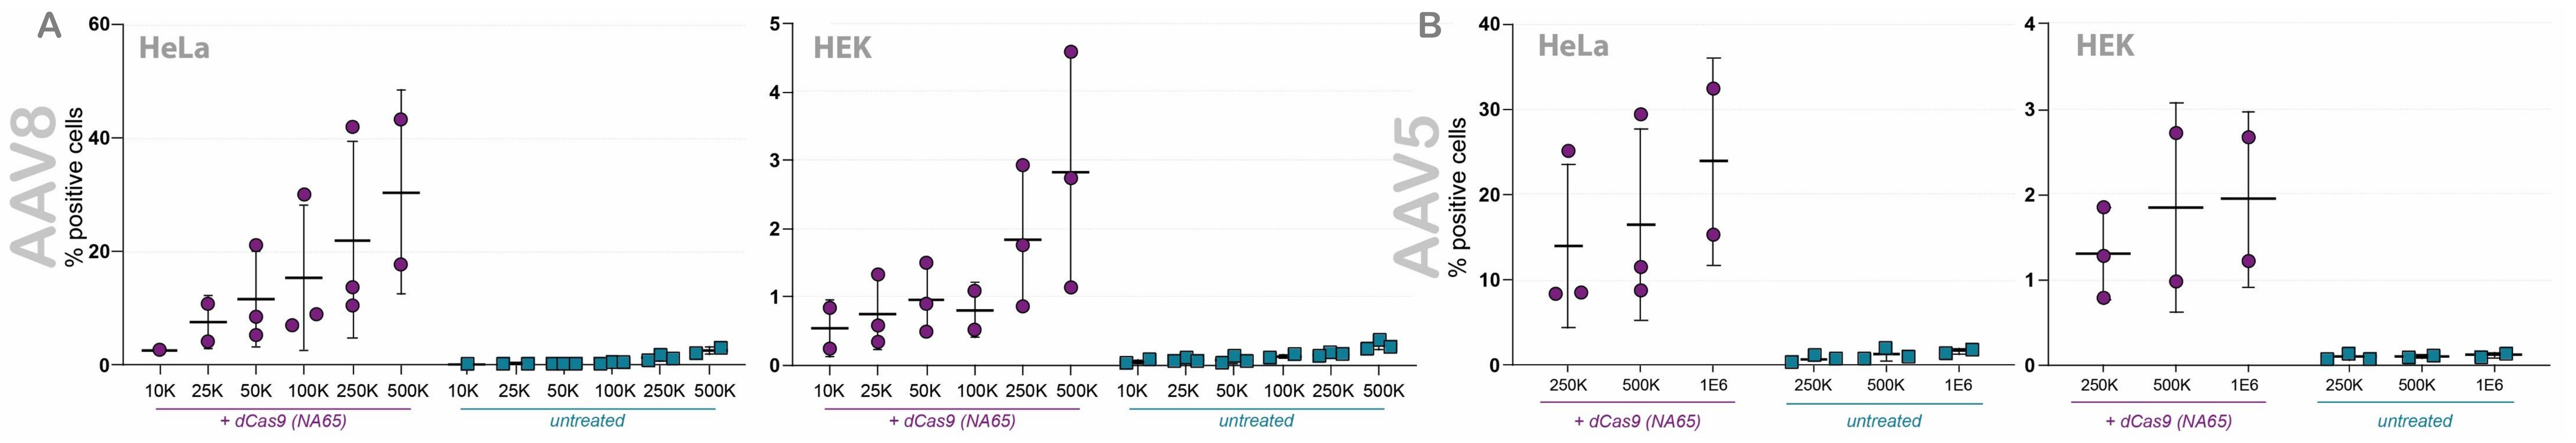
A
AAV8
B
AAV5
HeLa
HEK
% positive cells
+ dCas9 (NA65)
untreated

(TOP) Figure 3. Trans-activation of hRK promoter in HeLa and HEK cells. Plotted as mean (individual points represent independent experiments) with SD. Cells are: seeded (day0), transfected (day1), transduced (day2) and analysed by flow cytometry (day4).

(RIGHT) Figure 4. Trans-activation of NA65 promoter in HeLa and HEK cells. Plotted as mean (individual points represent independent experiments) with SD. Cells are: seeded (day0), transfected (day1), transduced (day2) and analysed by flow cytometry (day4).

Building up: combining different elements

Enhanced transduction: AAVR and doxorubicin

Over-expression of AAVR results in higher transduction levels for AAV8 but has little effect for AAV5. Separately, doxorubicin supplementation increases expression for both. Because they act through different pathways, could their effects act synergistically?

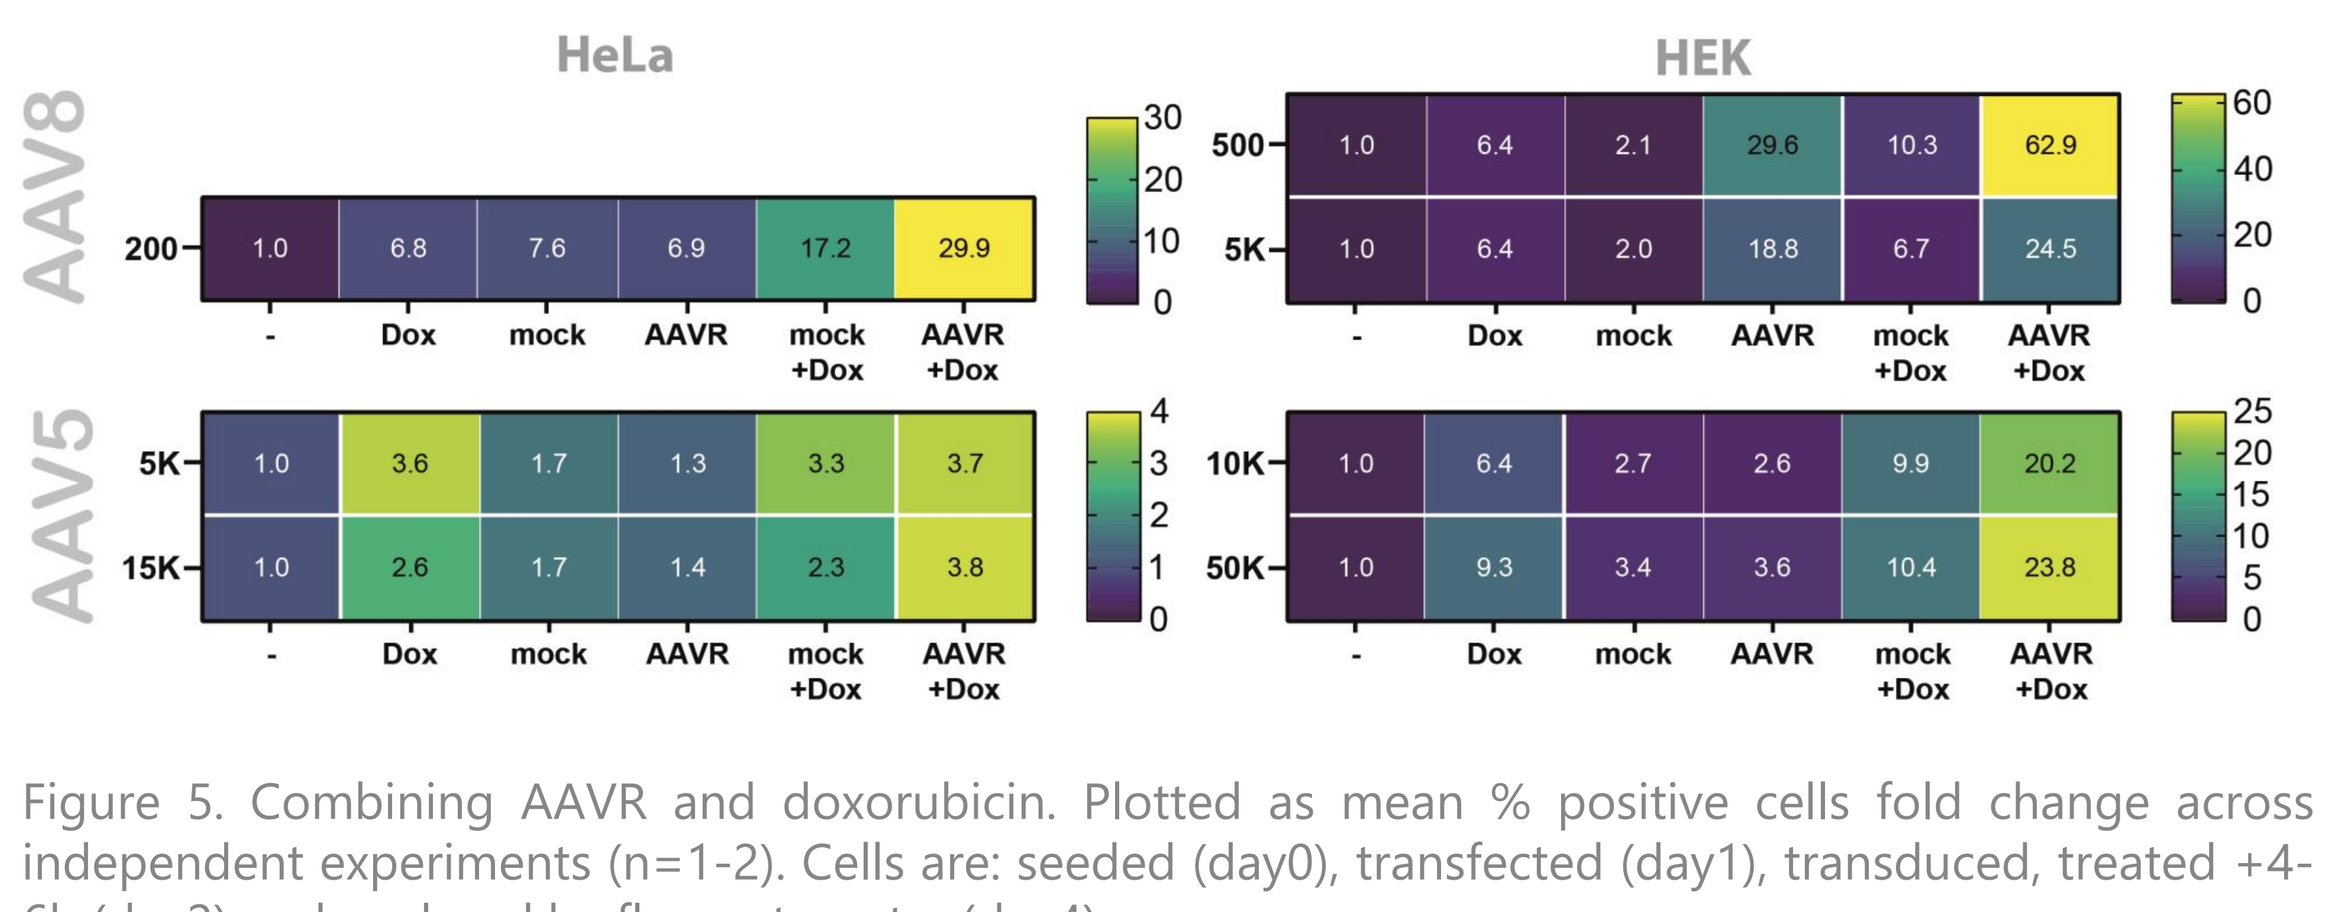
HeLa
HEK
AAV8
AAV5
200
1.0 6.8 7.6 6.9 17.2 29.9
500
1.0 6.4 2.1 29.6 10.3 62.9
5K
1.0 6.4 2.0 18.8 6.7 24.5
5K
1.0 3.6 1.7 1.3 3.3 3.7
15K
1.0 2.6 1.7 1.4 2.3 3.8
10K
1.0 6.4 2.7 2.6 9.9 20.2
50K
1.0 9.3 3.4 3.6 10.4 23.8
- Dox mock AAVR mock +Dox AAVR +Dox

Figure 5. Combining AAVR and doxorubicin. Plotted as mean % positive cells fold change across independent experiments (n=1-2). Cells are: seeded (day0), transfected (day1), transduced, treated +4-6h (day2) and analysed by flow cytometry (day4).

Our preliminary results show that the combination of AAVR over-expression and doxorubicin addition, has additive effects that result in a larger increase in transgene expression. Although the fold difference is both serotype and cell line dependent, higher levels were achieved in all cases tested.

(RIGHT) Figure 6. Combining AAVR and dCas9 for hRK (A,C) and NA65 (B,D). Plotted as mean (indep. expts n= 2-4 for AAV8, 2 for AAV5 (hRK) n= 3-4 for AAV8 and 2-3 for AAV5 (NA65)) with shaded SD. Cells are: seeded (day0), transfected (day1), transduced (day2) and analysed by flow cytometry (day4).

Enhanced trans-activation: AAVR and dCas9

Trans-activation of both hRK and NA65 promoters was achieved efficiently in HeLa and HEK cells for AAV8, but only to low levels with AAV5. Given that the systems are efficient per se *(transfection data not shown)*, could we increase further the expression levels if AAVR is added when the Cas9 system is present? With AAV8, for which both systems independently yielded good results, we observed additive effects which resulted in high levels of transgene expression even at low MOIs for both hRK (Fig. 6A) and NA65 (Fig.6B). For AAV5, however, addition of AAVR does not enhance trans-activation for hRK vectors (Fig. 6C) although it seems to have a mild enhancement for NA65 (Fig. 6D).

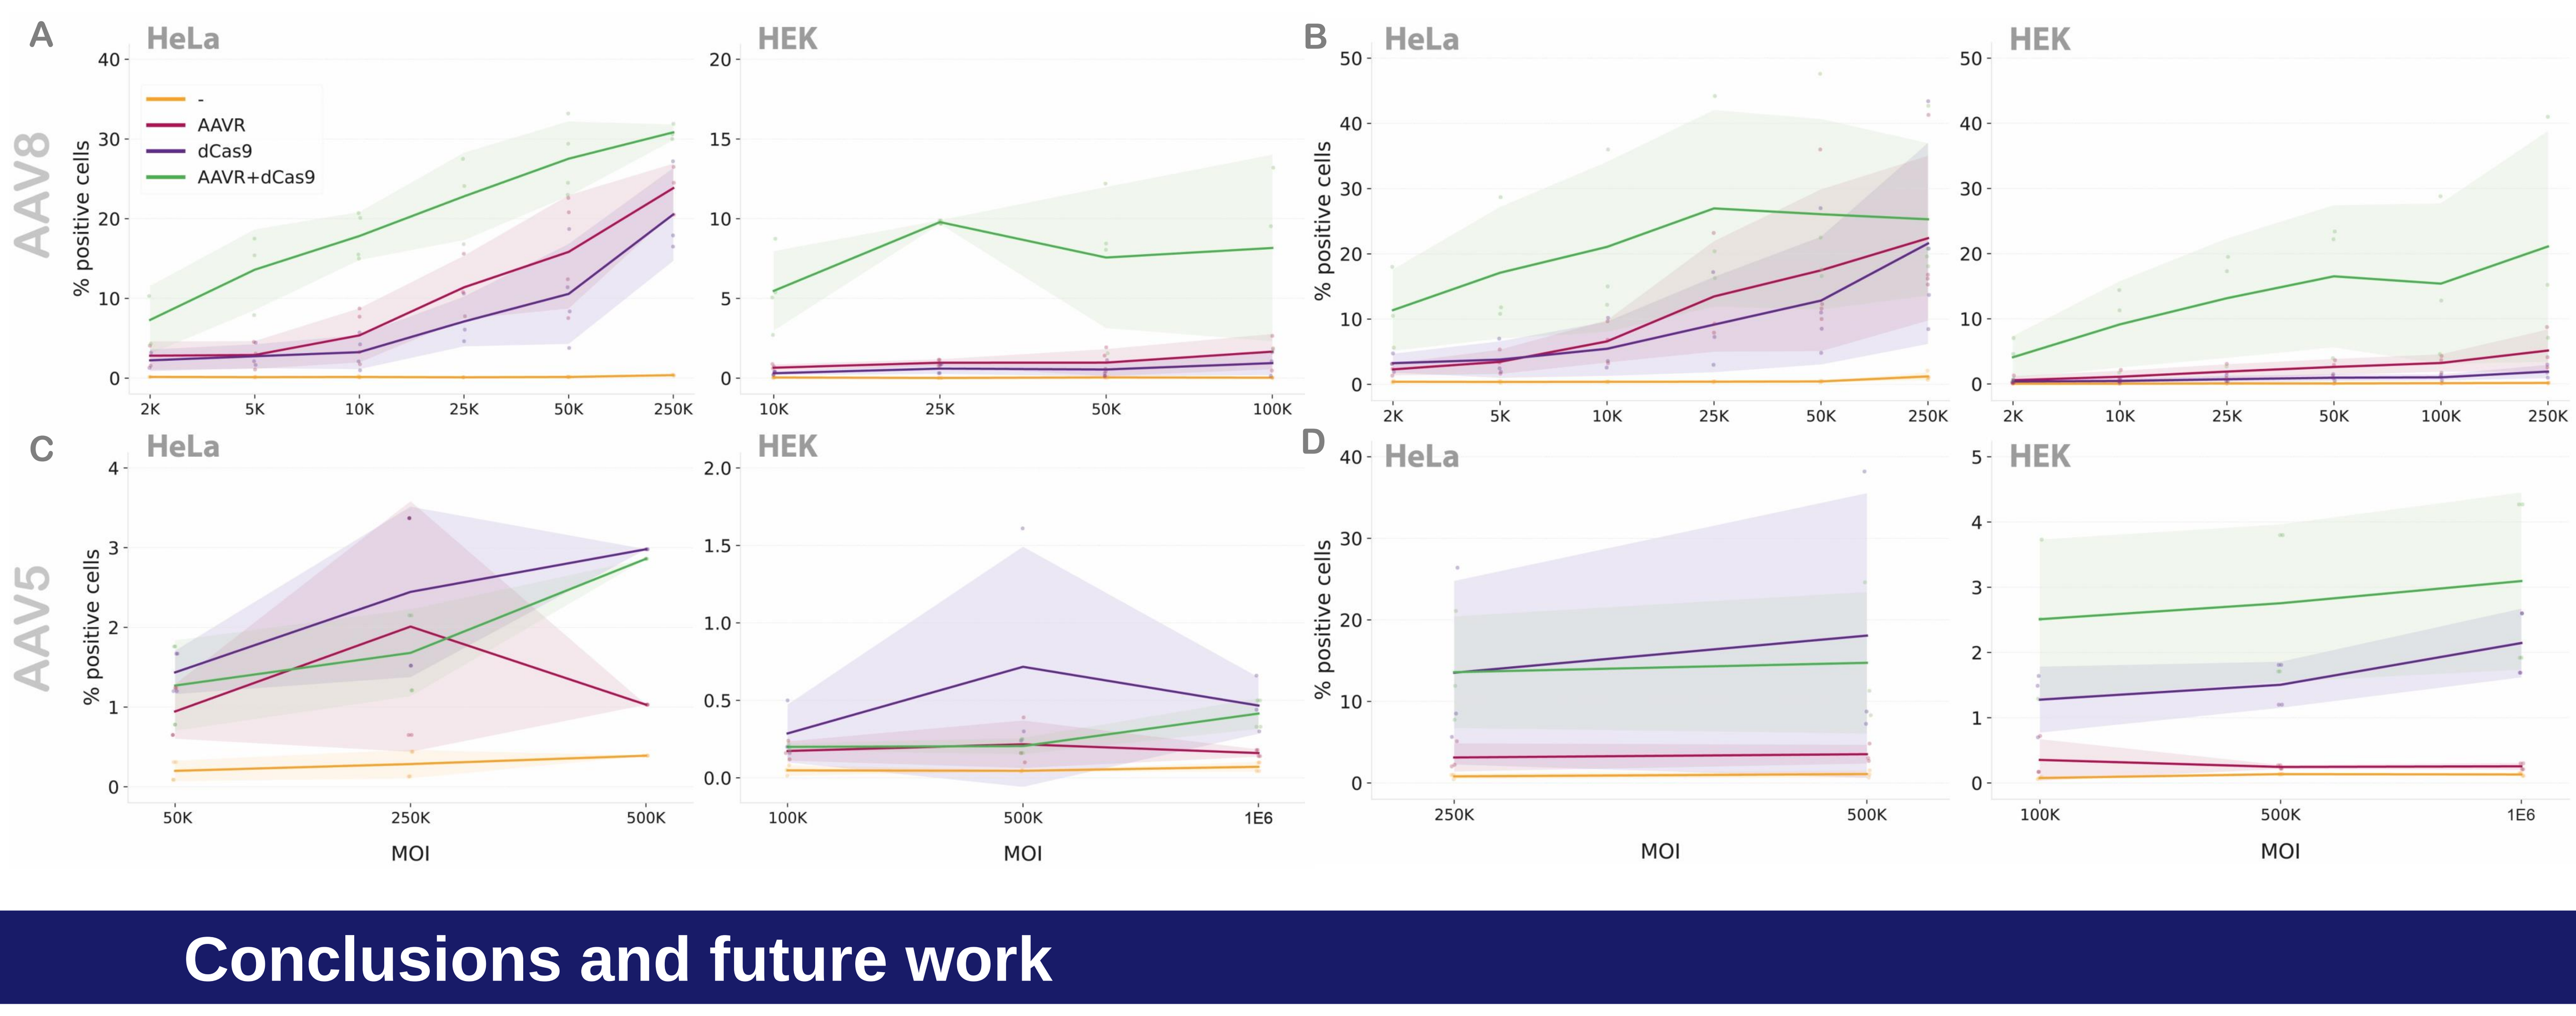
A
B
C
D
AAV8
AAV5
HeLa
HEK
-
AAVR
dCas9
AAVR+dCas9
% positive cells
MOI

Conclusions and future work

Over-expression of AAVR results in higher transduction for AAV8 but not for AAV5. Doxorubicin however, seems to have a more generic effect, enhancing transgene expression for both serotypes. Our preliminary results show that the combination of AAVR and doxorubicin has additive effects. Although the fold difference is both serotype and cell line dependent, higher levels were achieved in all cases tested. We have also been able to achieve trans-activation of two ocular promoters in both cell lines with AAV8 and AAV5, although with lower efficiency with the latter. When combining the system with AAVR, trans-activation is enhanced and higher levels of transgene expression can be achieved. Our next steps include exploring further combinations, establishing stable cell lines and starting tests with therapeutic transgenes.

This work paves the way towards establishing potency assays for ocular vectors *in vitro*, with the ability of working at low MOIs and with cell lines that are inexpensive, easy to maintain and that provide very quick readout times.

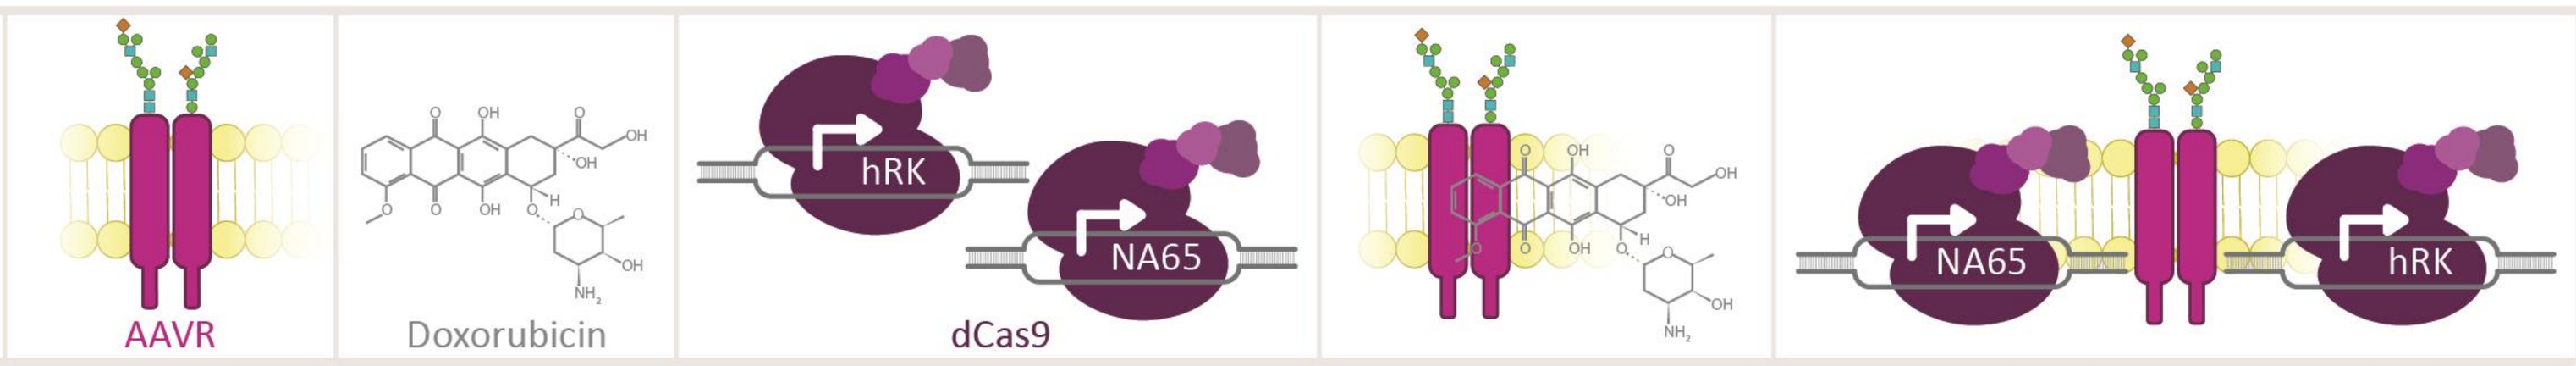
hRK
NA65
NA65
hRK

		AAVR	Doxorubicin	dCas9		
HeLa	8	High increase	enhanced expression	efficient trans-activation	additive effects	enhanced trans-activation
	5	No effect	enhanced expression	trans-activation achieved at low efficiency	not additive	combination not synergistic
HEK	8	High increase	enhanced expression	trans-activation achieved at low efficiency	additive effects	enhanced trans-activation
	5	No effect	enhanced expression	no successful trans-activation	additive effects	combination not synergistic, slight increase in NA65

References

(1) Pillay, 2016
(2) Dudek, 2018
(3) Ju, 2004
(4) Kanazawa, 2004
(5) Nicolson, 2016
(6) Yan, 2004
(7) Gong, 2021
(8) Russell, 1995
(9) Rambhai, 2020
(10) Zhang, 2009
(11) Zhang, 2012
(12) Chavez, 2015
(13) McDougald, 2019