Poster #1245

Towards Ultra Scale-Down AAV Production in Microtiter Plates

Authors: Vincent Wiegmann, Florian Dziopa & Bastiaan Leewis

MSAT Upstream Development, MeiraGTx, London, UK

ABSTRACT

High-throughput screening methods have become an integral part of research and development in the biopharmaceutical industry. Due to their low cost, ease of operation and high degree of parallelization, microtiter plates remain an important format for early-stage process development with a strong track-record of bioprocessing applications. Yet, few examples of viral vector production in microtiter plates have been published to date.

This work describes the scale-down of a transient transfection-based AAV production process from 10 L stirred tank reactor (STR) to 24 deep square well microtiter plates (24 DSW), using an AAV2-based product.

Initially, a range of working volumes was evaluated and shown to result in comparable HEK293 growth kinetics to the 10 L STR-scale. Subsequently, a Design of Experiment (DoE) approach was employed to further the understanding of the transfection step at the micro-scale. Finally, process variability and scalability of the optimized process were assessed.

1. Material and Methods

Cell Expansion

HEK293 cells were seeded at the same cell density of 3x10⁵ VC/mL in DSW and STR vessels. Microtiter plate cultivations were performed either in polystyrene or polypropylene square 24-deepwell microplates with low-evaporation sandwich covers (all Enzyscreen BV, NL) in a S41i shaker incubator (Eppendorf, Germany) set to a shaking speed of 240rpm (1" orbital throw). A process overview is detailed in Figure 1. The temperature shift mimics the biphasic temperature control at bioreactor scale, while the CO_2-ramp mimics bioreactor CO_2 demand with the aim to maintain a pH of 6.9±0.4 in well plates. Bioreactor cultures were performed in BioBLU 14c vessels (Eppendorf, Germany).

Transfection

Cells were transfected 96h post-inoculation, using TransIT-VirusGEN transfection reagent (Mirus Bio LLC, USA). Plates were immediately returned to the incubator shaker after transfection mix addition. In some instances (see Table 1), the shaking speed was reduced to 200 rpm for 30 min after transfection mix addition.

Harvest

72h post-transfection cells were lysed using a Triton X-100 based lysis buffer, incubated for 2h at 350 rpm and 37°C.

Analytics

Viable cell concentration and viability were measured using a NucleoCounter NC-250 (Chemometec, Denmark). Viral genome (VG) titers were determined using qPCR, while virus particle (VP) titer was measured by immunoassay in the Gyrolab platform (Gyros Protein Technologies, Sweden).

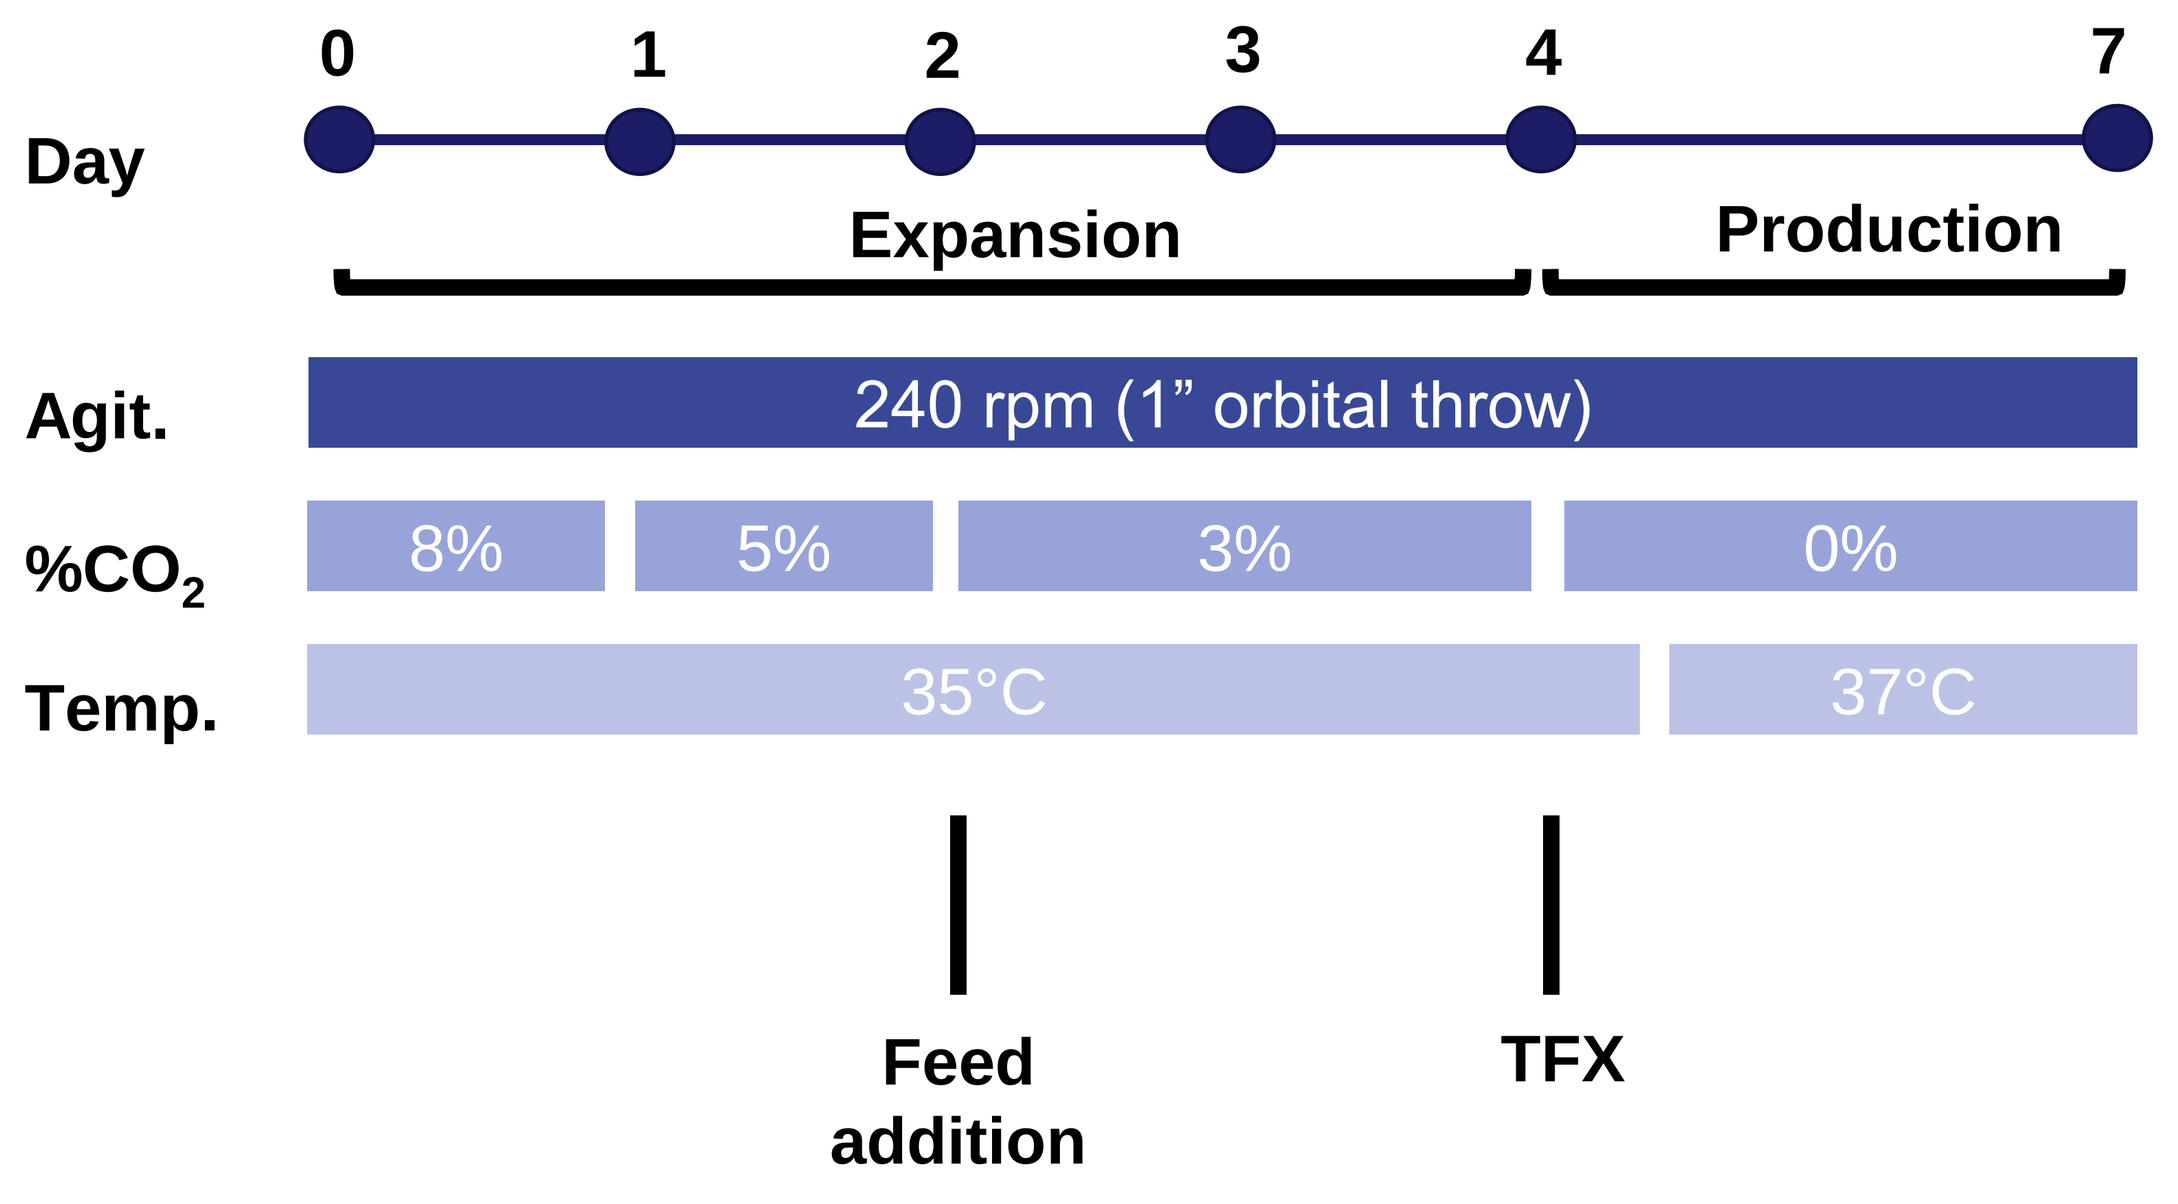
Day
0
1
2
3
4
7
Expansion
Production
Agit.
240 rpm (1" orbital throw)
%CO2
8%
5%
3%
0%
Temp.
35°C
37°C
Feed addition
TFX

Figure 1: Ultra scale-down AAV manufacturing process overview.

2. Cell Expansion in Microtiter Plates

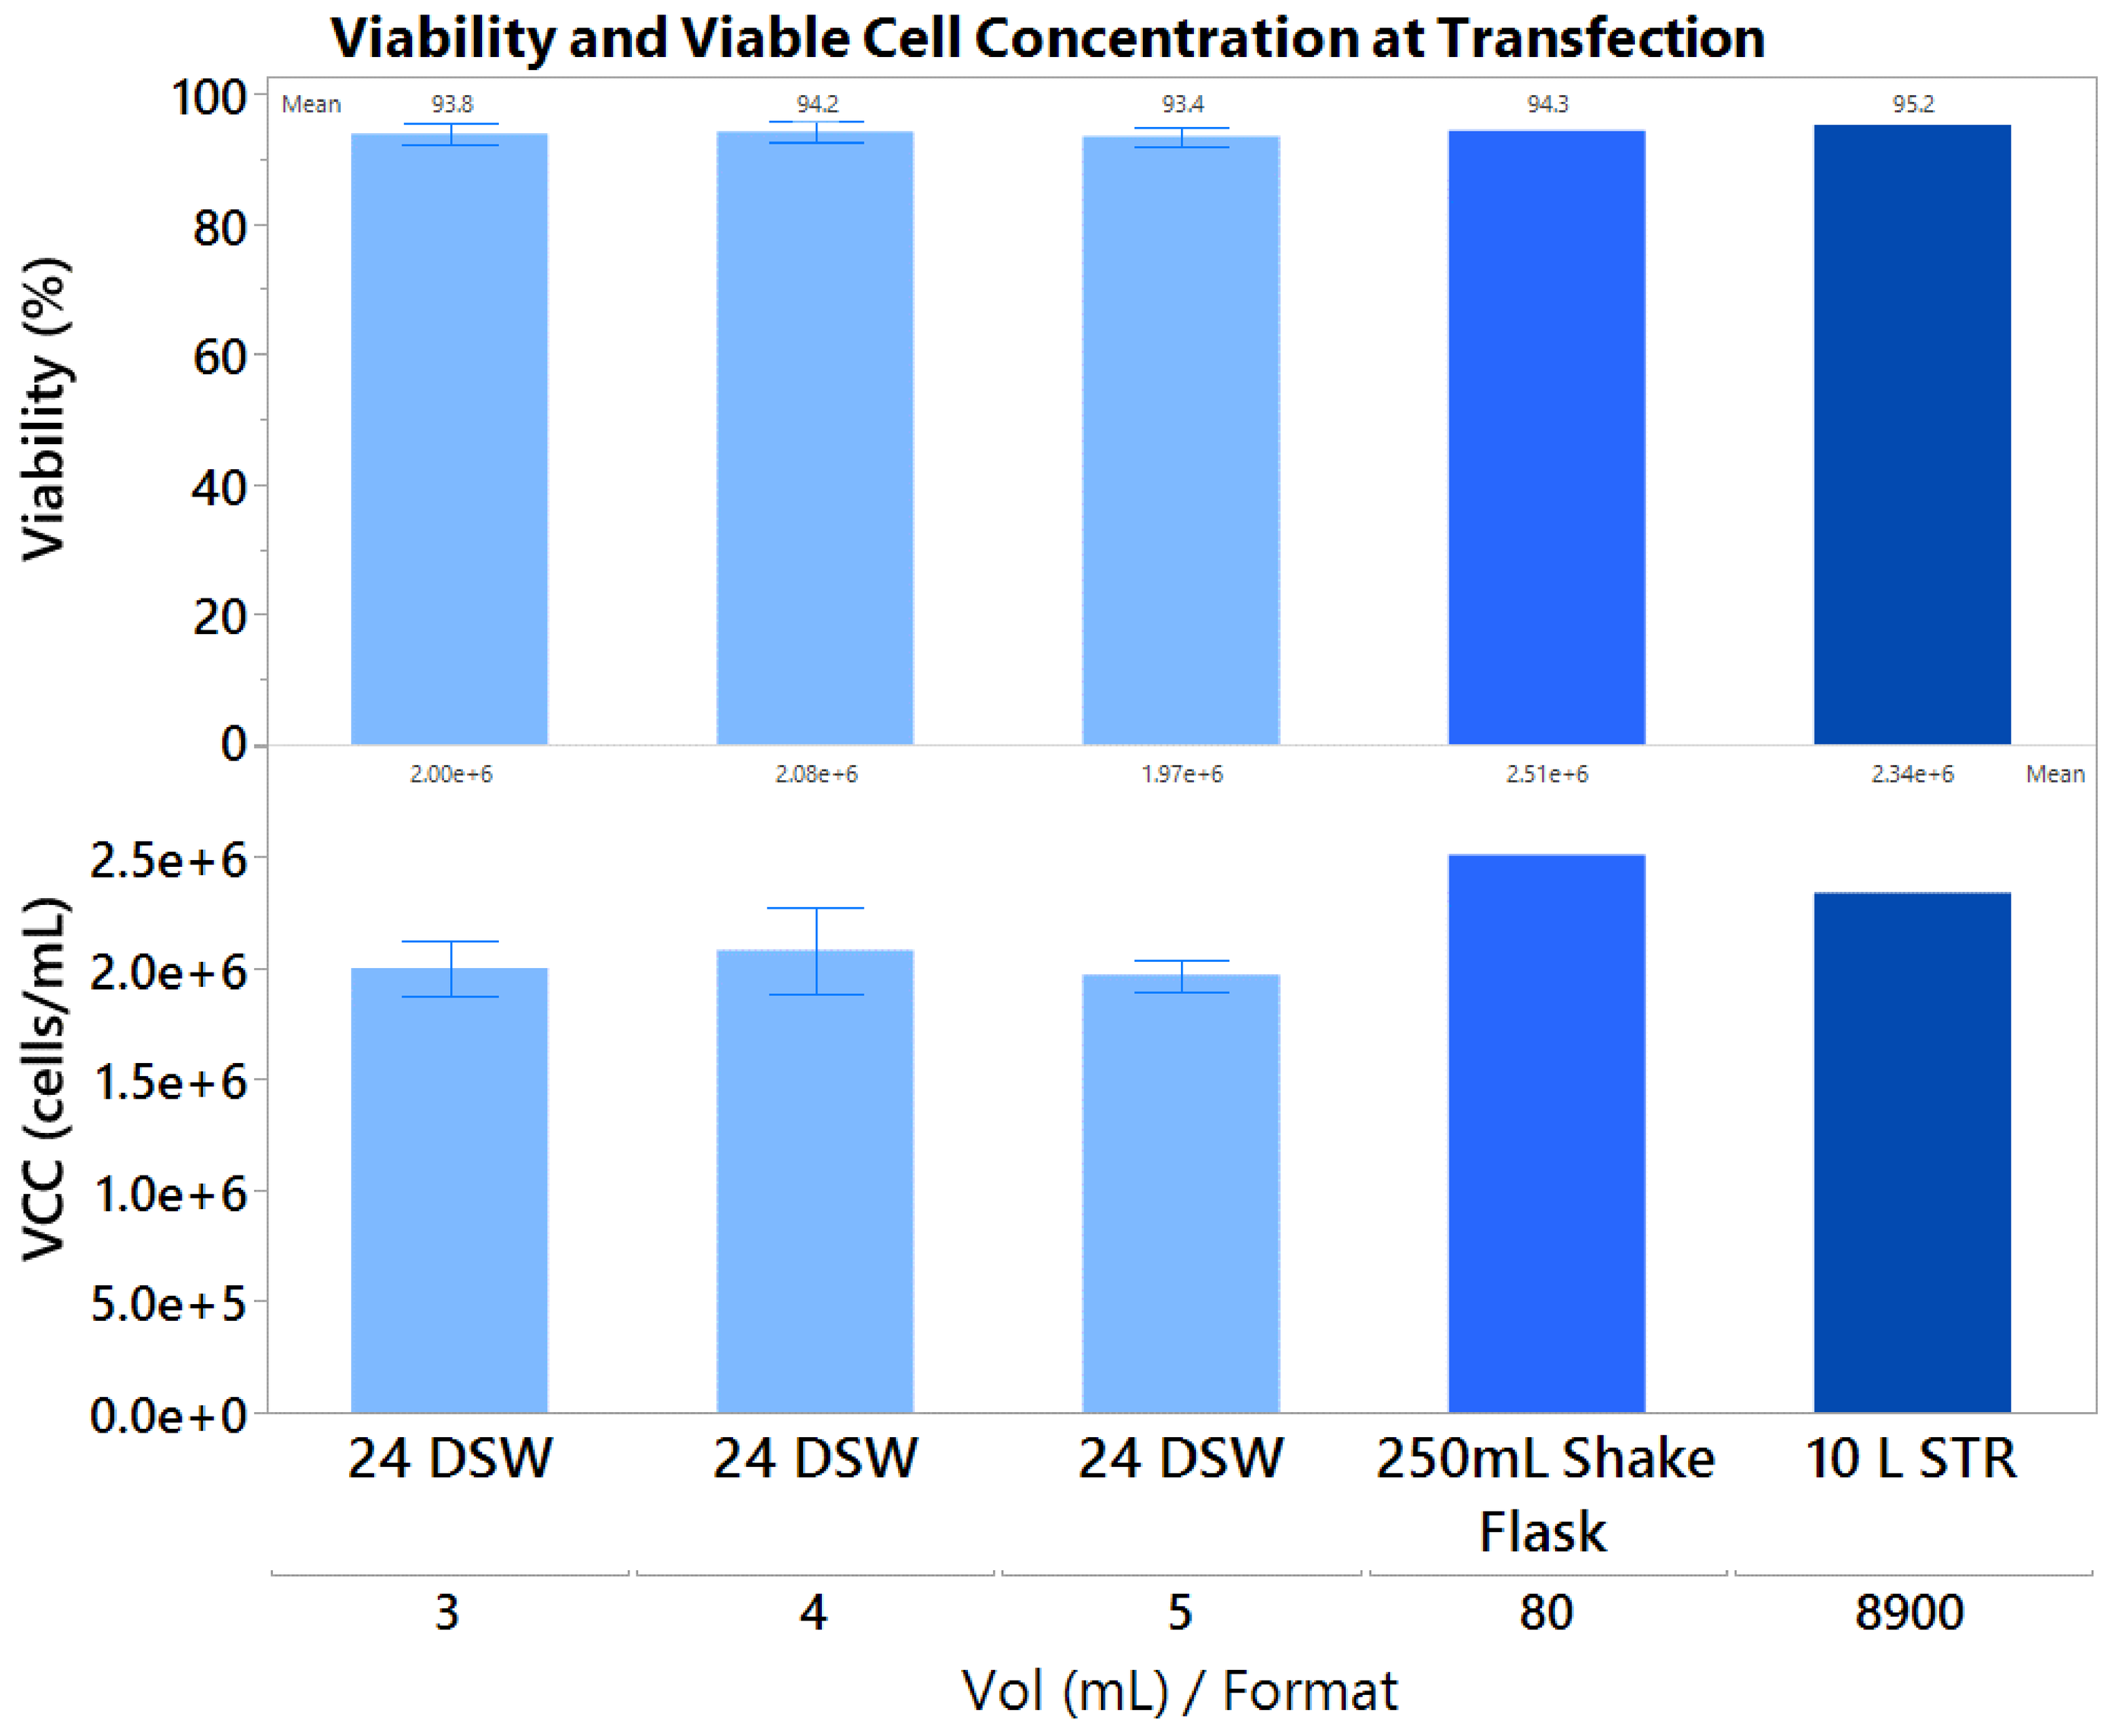
Viability and Viable Cell Concentration at Transfection
Viability (%)
Mean
93.8
94.2
93.4
94.3
95.2
2.00e+6
2.08e+6
1.97e+6
2.51e+6
2.34e+6
VCC (cells/mL)
24 DSW
24 DSW
24 DSW
250mL Shake Flask
10 L STR
3
4
5
80
8900
Vol (mL) / Format

Figure 2: Cell Viability and Viable Cell Concentration at transfection in 24DSW (■), Shake Flask (■), and 10L STR (■). Error bars represent Mean ± S.D. (n = 3).

- Although cell growth was slightly slower in 24 DSW compared to 250 mL shake flask and 10 L STR, the cell concentration at transfection was within the expected range of 2.0 – 3.5x10⁶ VC/mL.
- Cell viability at transfection was above 90% and highly comparable between cultivation formats.
- A working volume of 5 mL was deemed ideal due to a lower relative evaporation and larger available volume.

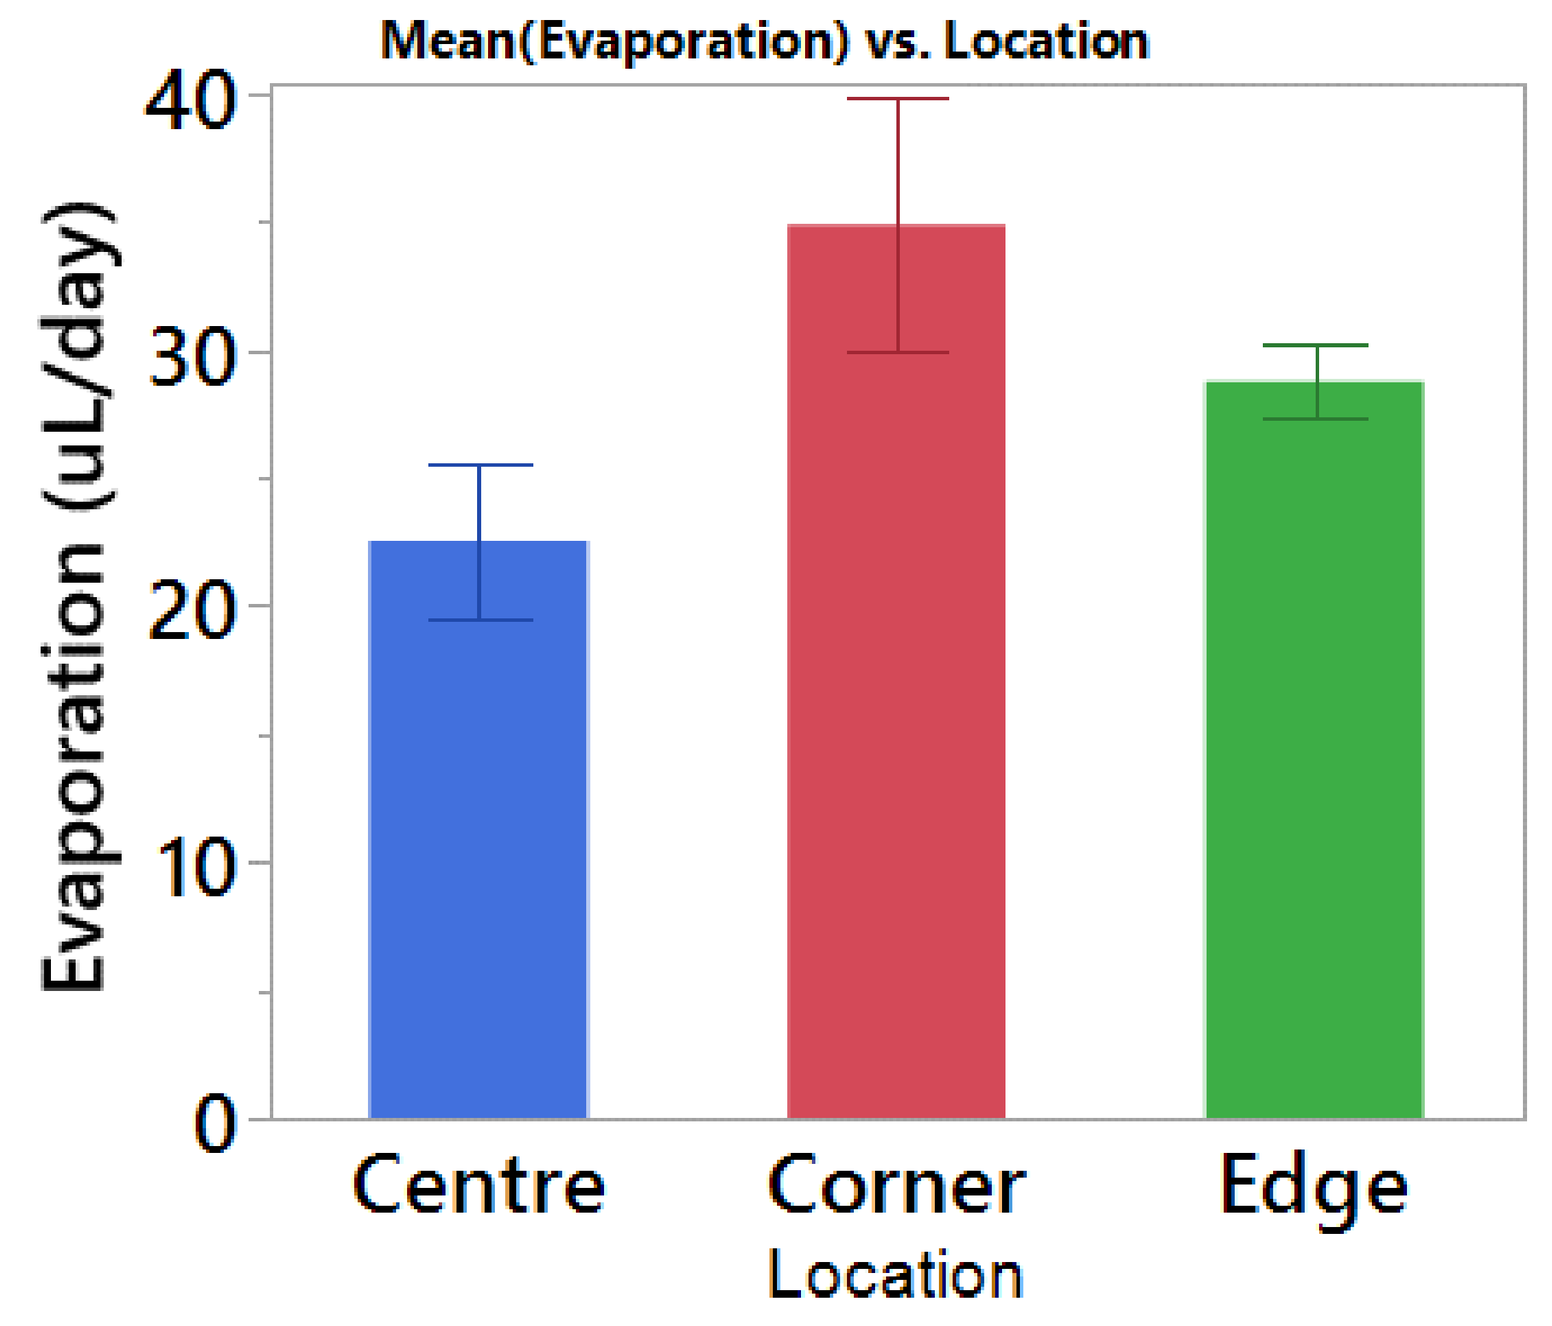
Mean(Evaporation) vs. Location
Evaporation (uL/day)
Centre
Corner
Edge
Location

Figure 3: Daily evaporation in 24 DSW in combination with a low-evaporation sandwich cover.

3. Factor Screening using Design of Experiment

- A Design of Experiment approach was employed to investigate the importance of factors that may affect process productivity (see Table 1).
- Choice of 24 DSW material and vessel in which the transfection mix was prepared did not significantly affect the VG titer.
- Returning the transfected culture to a shaking speed of 240 rpm and mixing of the cell broth immediately after transfection mix addition via aspiration proved pivotal in maintaining a VG titer comparable to bioreactor productions.
- Optimal process conditions resulted in a productivity within the VG titer range obtained at 10 L STR scale of 1.1 - 2.4x10¹¹ VG/mL (n=7).

Table 1: Design of Experiment Factors and Limits.

Factor Name	Lower Limit (-)	Upper Limit (+)	Units
TFX mix preparation vessel	Eppendorf tube	Glass bottle	N/A
24 DSW Material	Polypropylene	Polystyrene	N/A
Aspiration after TFX mix addition	No	Yes	N/A
Agitation post-TFX mix addition	200	240	RPM

Table 2: Summary of significant model effects.

Source	Logworth	PValue
Aspiration after TFX mix addition	2.463	0.00344
TFX agitation	2.208	0.00619

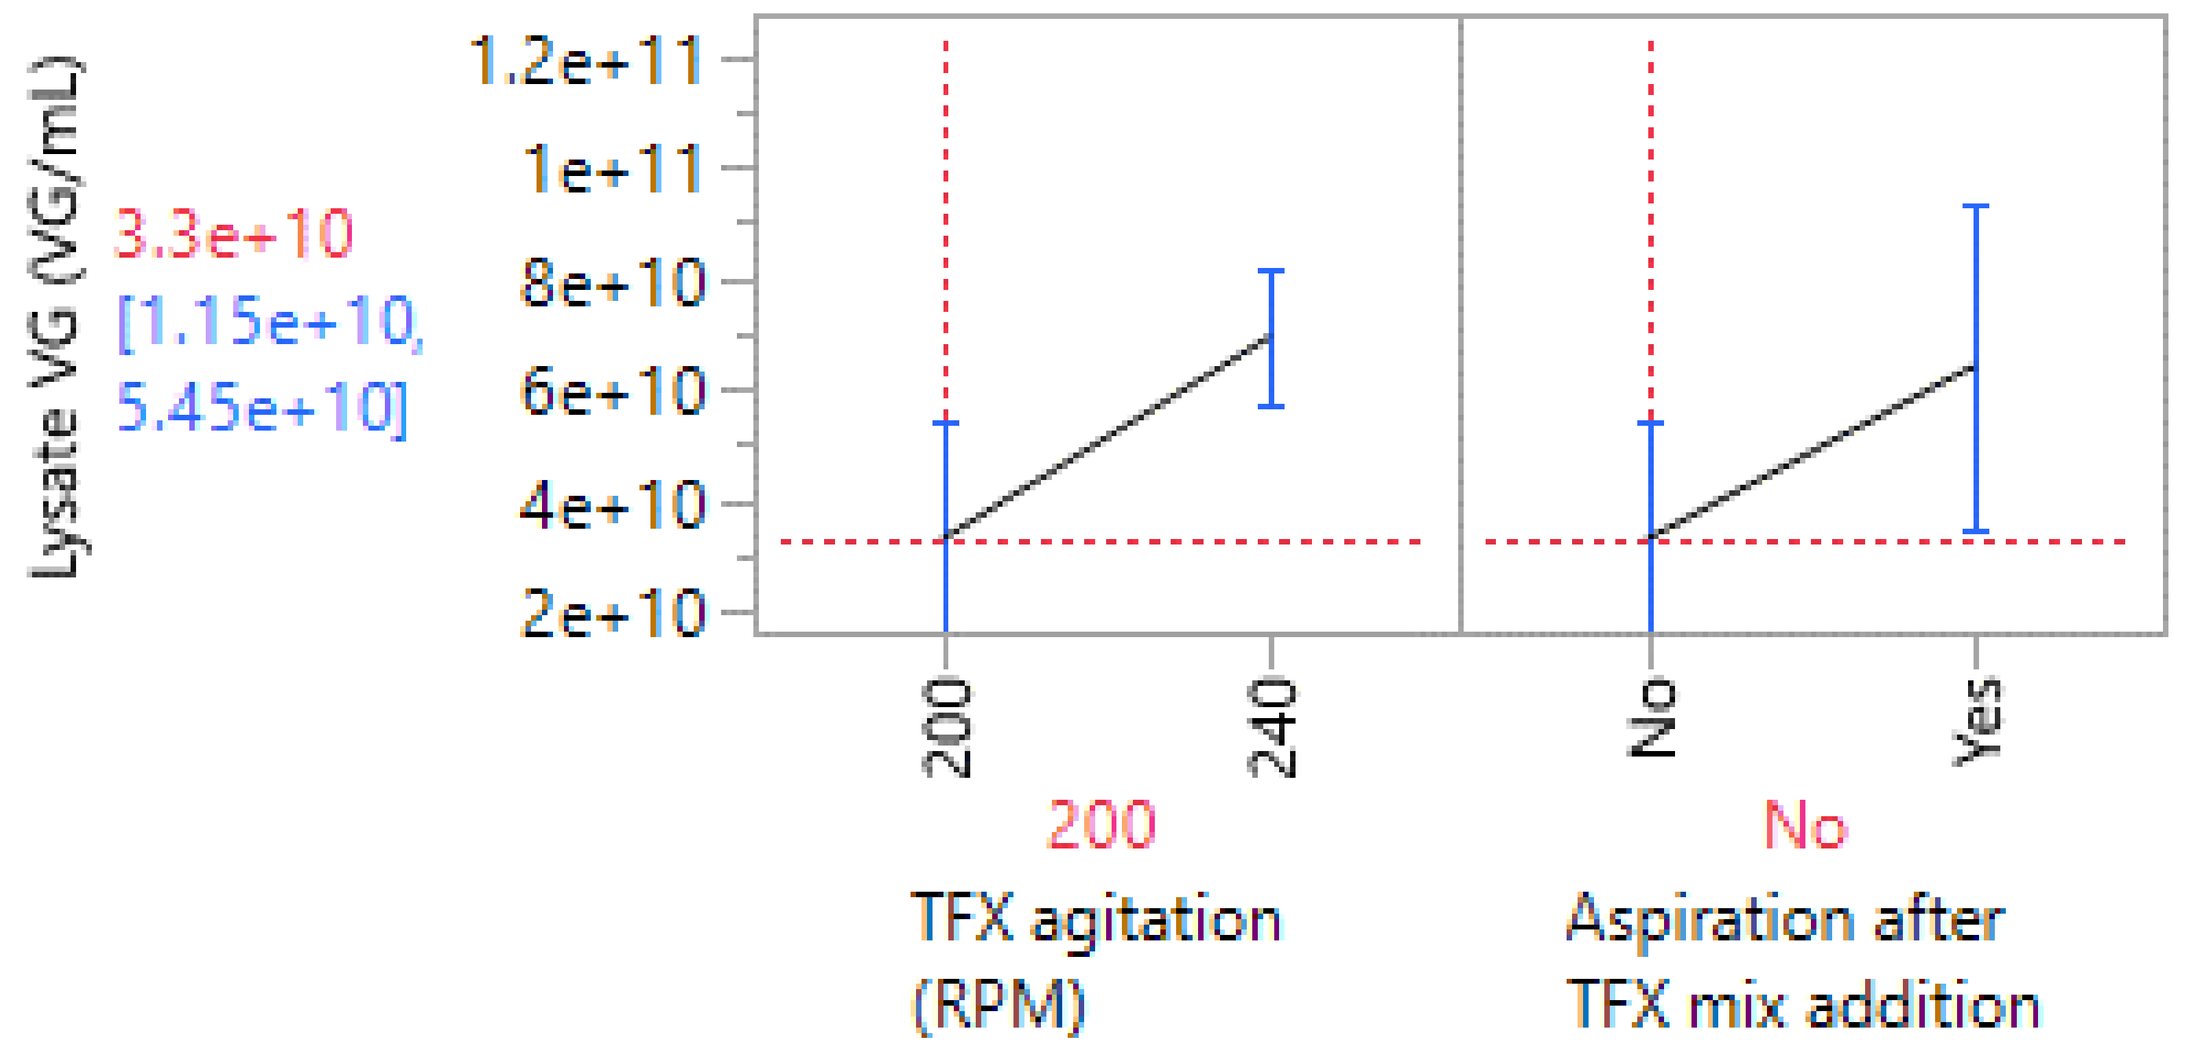
Lysate VG (VG/mL)
3.3e+10
[1.15e+10, 5.45e+10]
200
TFX agitation (RPM)
No
Aspiration after TFX mix addition

Figure 4: Model prediction profile for non-optimal conditions.

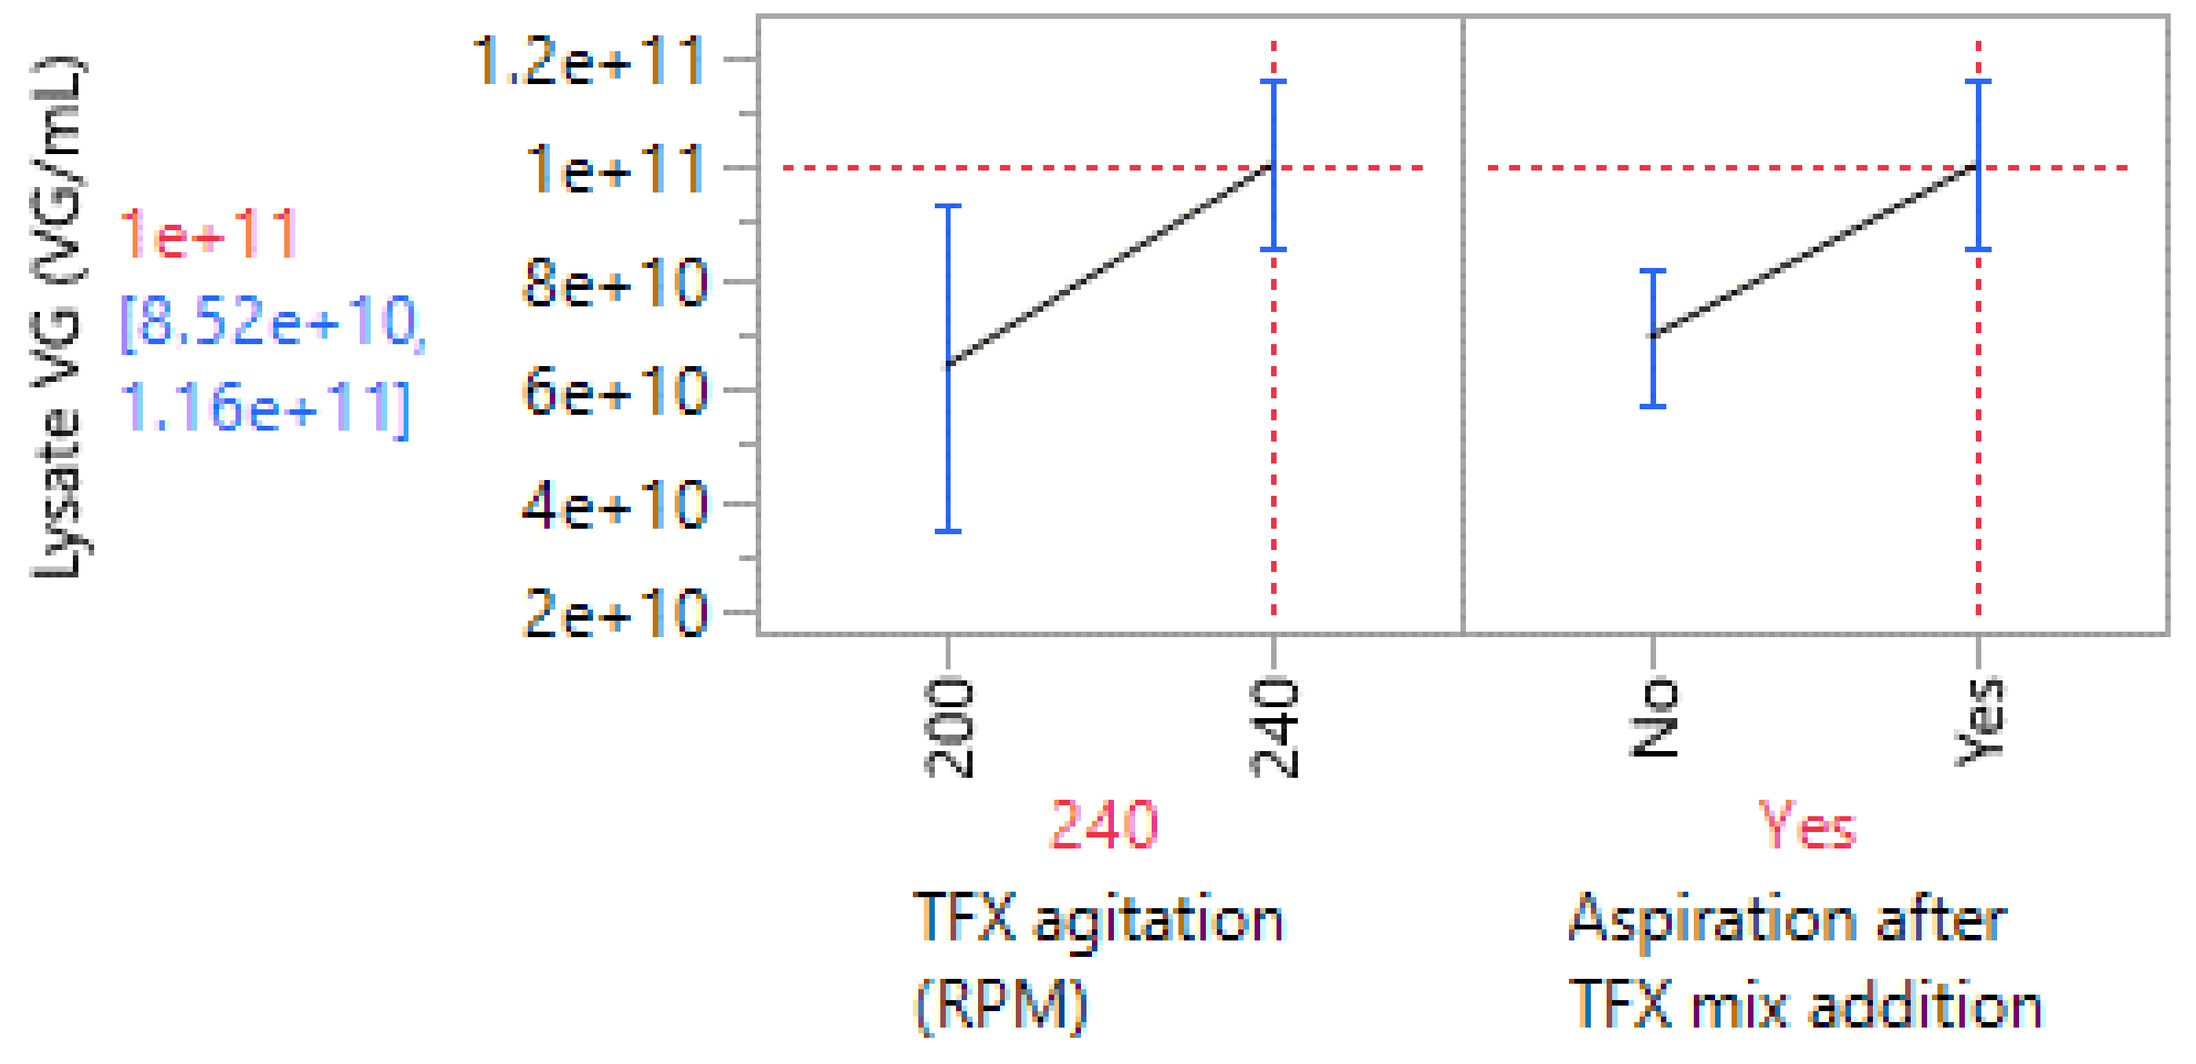
Lysate VG (VG/mL)
1e+11
[8.52e+10, 1.16e+11]
240
TFX agitation (RPM)
Yes
Aspiration after TFX mix addition

Figure 5: Model prediction profile for optimal conditions.

4. Assessment of Scalability and Intra-plate Repeatability

- To assess intra-plate process repeatability, the scale-down production process was repeated in 12 wells. The VG titer was compared to the established range at 10 L STR scale.
- The VG productivity was shown to fall within the range established for the AAV2-based product for all wells.
- The VG titer was subject to some degree of variability (cv: 26.6%), which was consistent with the 10 L STR scale process reproducibility (n=7).
- No significant effect (*p*=0.69) of the well location on titer was established, suggesting that the entire 24 DSW can be utilized for screening experiments.

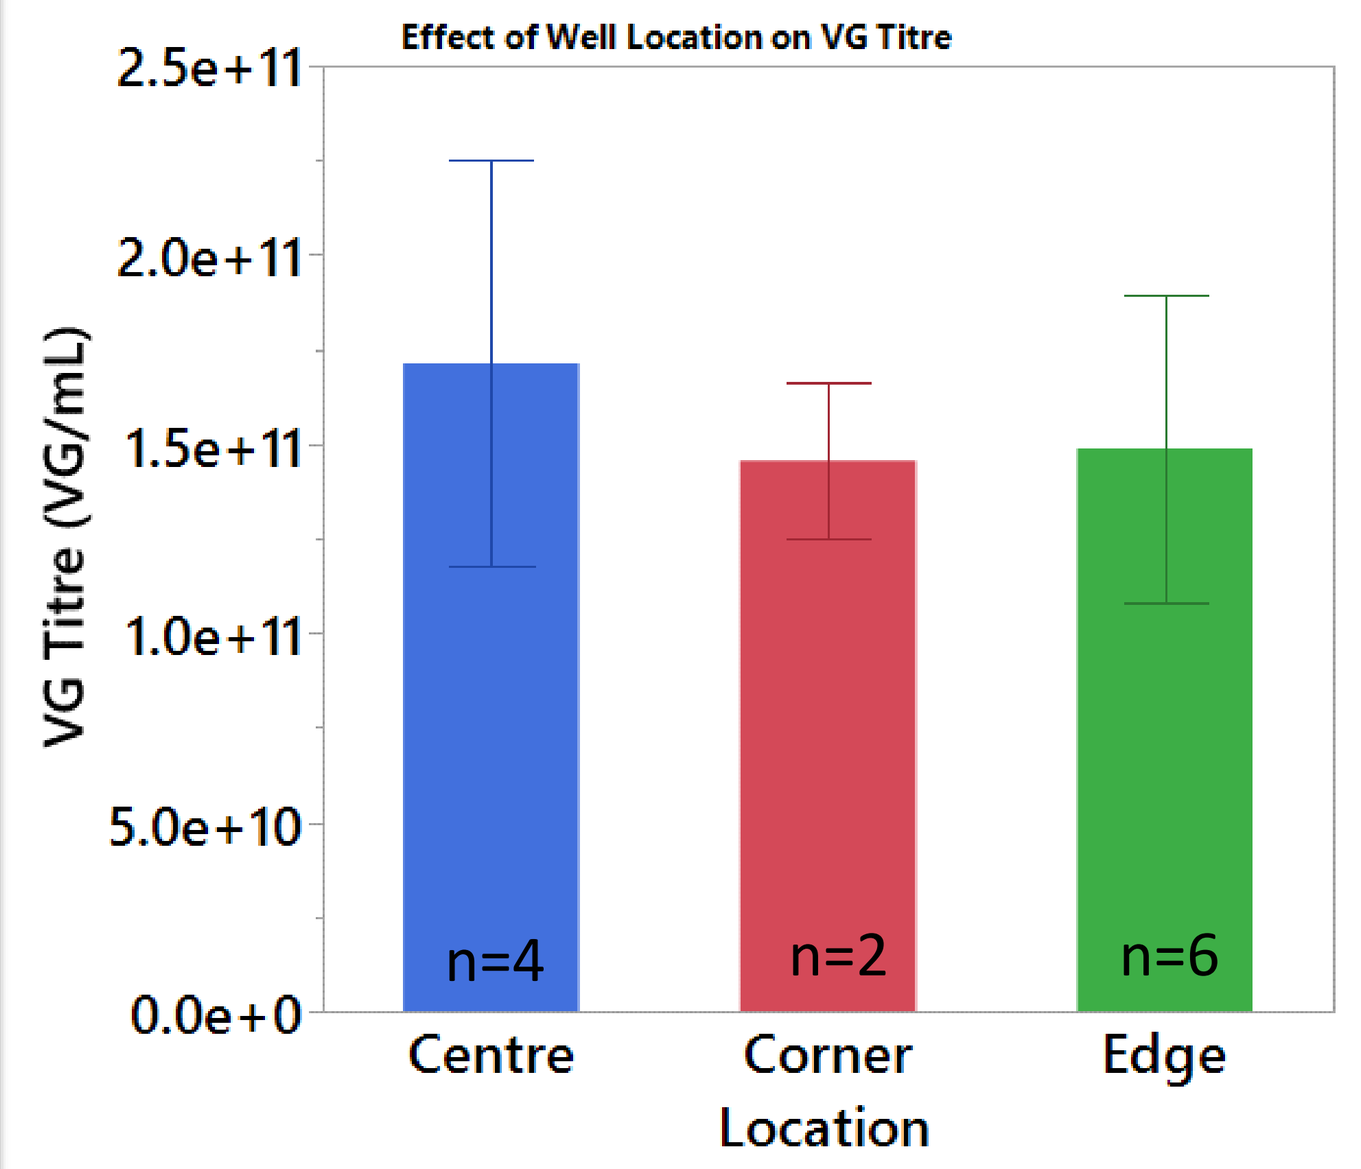
Effect of Well Location on VG Titre
VG Titre (VG/mL)
n=4
n=2
n=6
Centre
Corner
Edge
Location

Figure 7: VG Titer by location. Error bars represent Mean ± S.D.

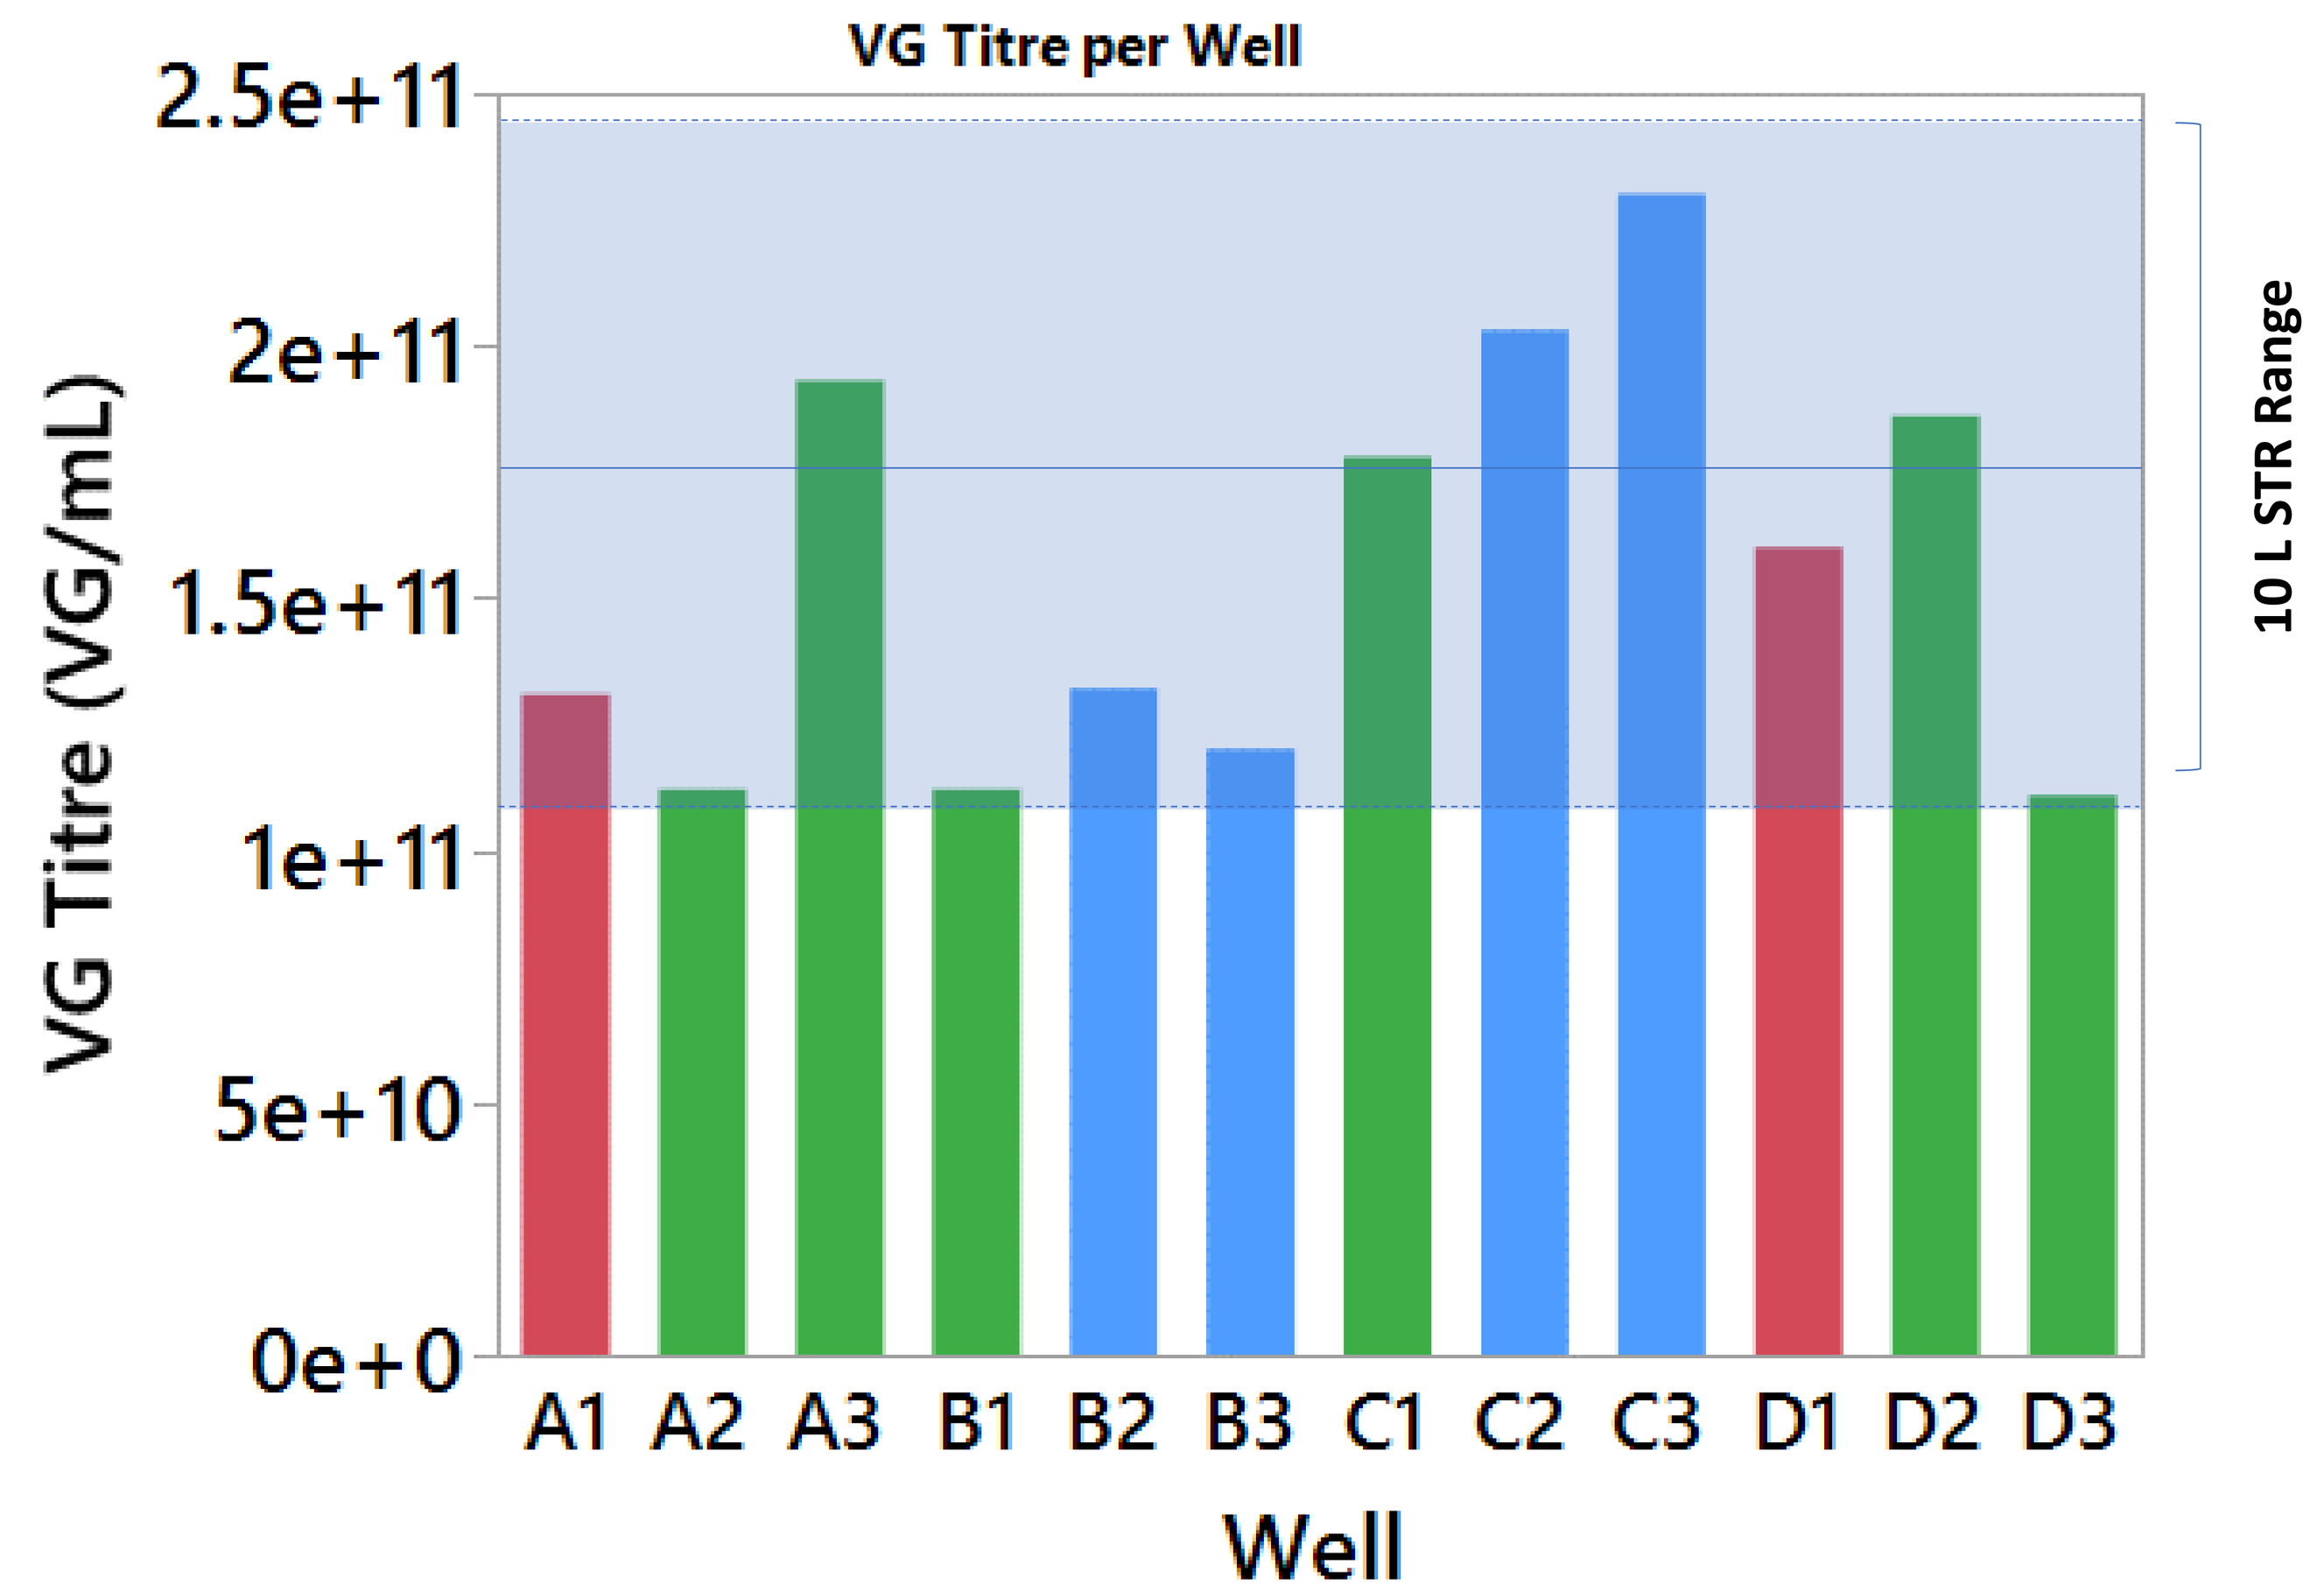
VG Titre per Well
VG Titre (VG/mL)
A1 A2 A3 B1 B2 B3 C1 C2 C3 D1 D2 D3
Well
10 L STR Range

Figure 6: VG Titer per well in comparison to the expected range at 10L STR scale. Color-coded well locations entail center (■), corner (■), and edge (■).

5. Conclusions

Using a microtiter plate scale-down model can significantly accelerate development timelines. However, fundamental differences to stirred tank bioreactors may impede scalability.

This work has demonstrated that cell expansion and productivities comparable to the 10 L scale can be reached using the 24 DSW format cultured in an incubator shaker. A tightly controlled transfection step with sufficient mixing was identified as playing a decisive role in achieving scalable productivity.

Overall, these findings illustrate a viable method for high-throughput early-stage AAV process development.