P630

Improving AAV in vitro transducibility for cell-based potency assay development

Gemma Estrada Girona, Anastasios Georgiadis

MeiraGTx London

Introduction

When producing new viral vector tools for gene delivery, the ability to assess their efficacy is an essential part of the research process. This can be done using a variety of assays and measuring different parameters, but experimental setup aside, there is an initial common step: transduction. Two critical aspects of gene therapy development depend on transduction to be efficient: cell-based potency assays are a regulatory requirement for commercialisation of AAV gene therapies but poor AAV in vitro transducibility is hindering the development of efficient and robust assays, and pre-clinical research is often limited in basic expression testing by low transducibility in easy-to-implement assays that would allow faster paced experiments and iterations. Finding tools to enhance this process would therefore be greatly beneficial. We set out to screen different AAV serotypes, cell lines and treatments and asses the effect of these in the transduction process with the aim of finding enhancements that could be easily transferable between platforms.

Metal ion supplementation

Metal ions are critical components in many cellular processes. A 2020 study[1] showed that the combination of zinc and cobalt media supplementation led a 30-fold enhancement of AAV transduction in HeLa with scAAV3. To our knowledge, no further work on the use of metals has been published.

Our initial toxicity screens revealed a high susceptibility to the treatment for all cell lines and concentrations had to be greatly reduced (Fig. 1) to obtain minimal robustness (with respect to the published ones, even for the same cell line).

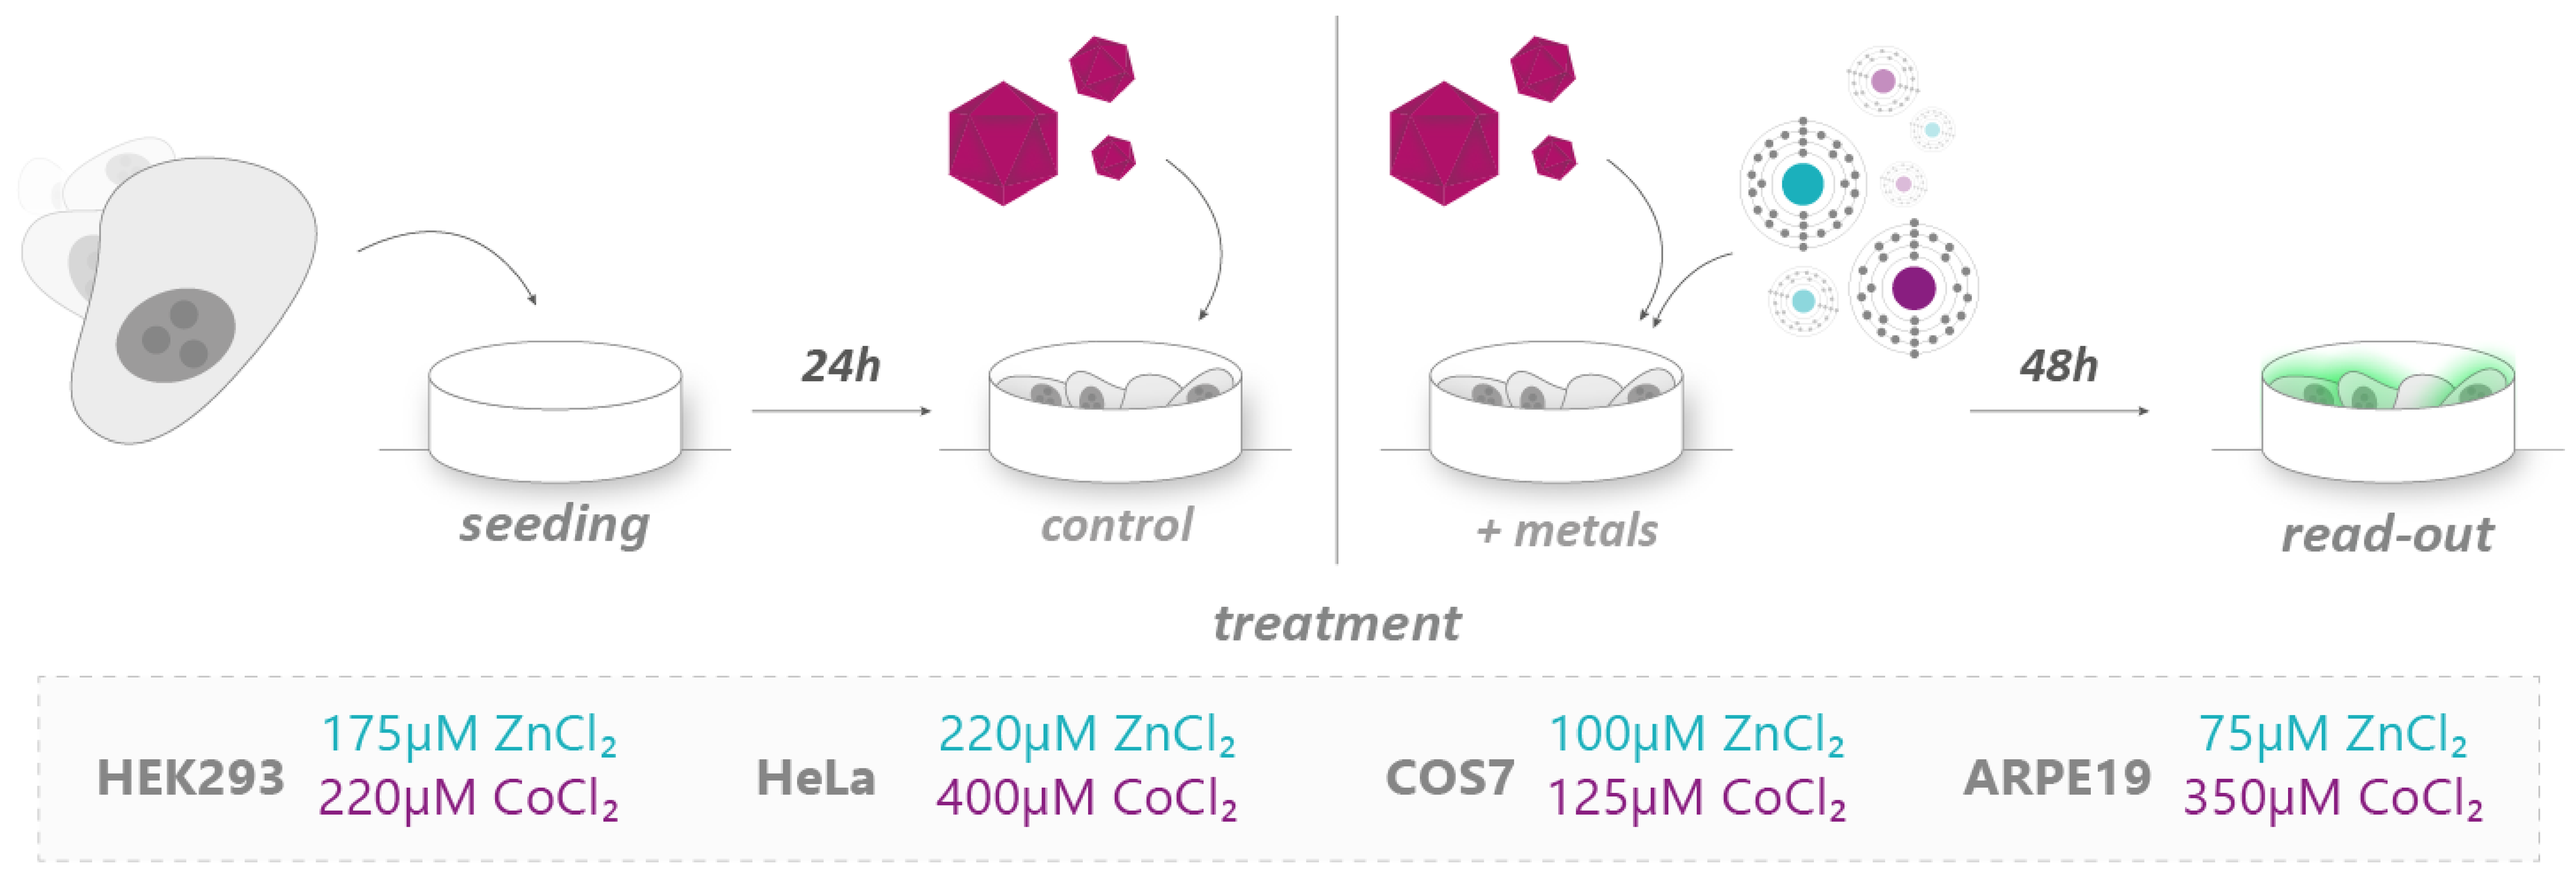
seeding
24h
control
+ metals
48h
read-out
treatment
HEK293 175µM ZnCl₂ 220µM CoCl₂
HeLa 220µM ZnCl₂ 400µM CoCl₂
COS7 100µM ZnCl₂ 125µM CoCl₂
ARPE19 75µM ZnCl₂ 350µM CoCl₂

Figure 1. Schematic of the protocol for assessing the effect of metal ion treatment and summary of the final concentrations of zinc and cobalt chloride solutions used for each cell line.

In HEK293 metal treatment results in a slight signal increase for AAV8 (Fig. 2). In COS and HeLa the effect is negligible. Higher concentrations in COS showed a tendency to higher increase but toxicity was higher, often killing the cells. Preliminary data in ARPE19 cells *(data not shown)* trends towards a longer time-dependent consistent increase in transduction.

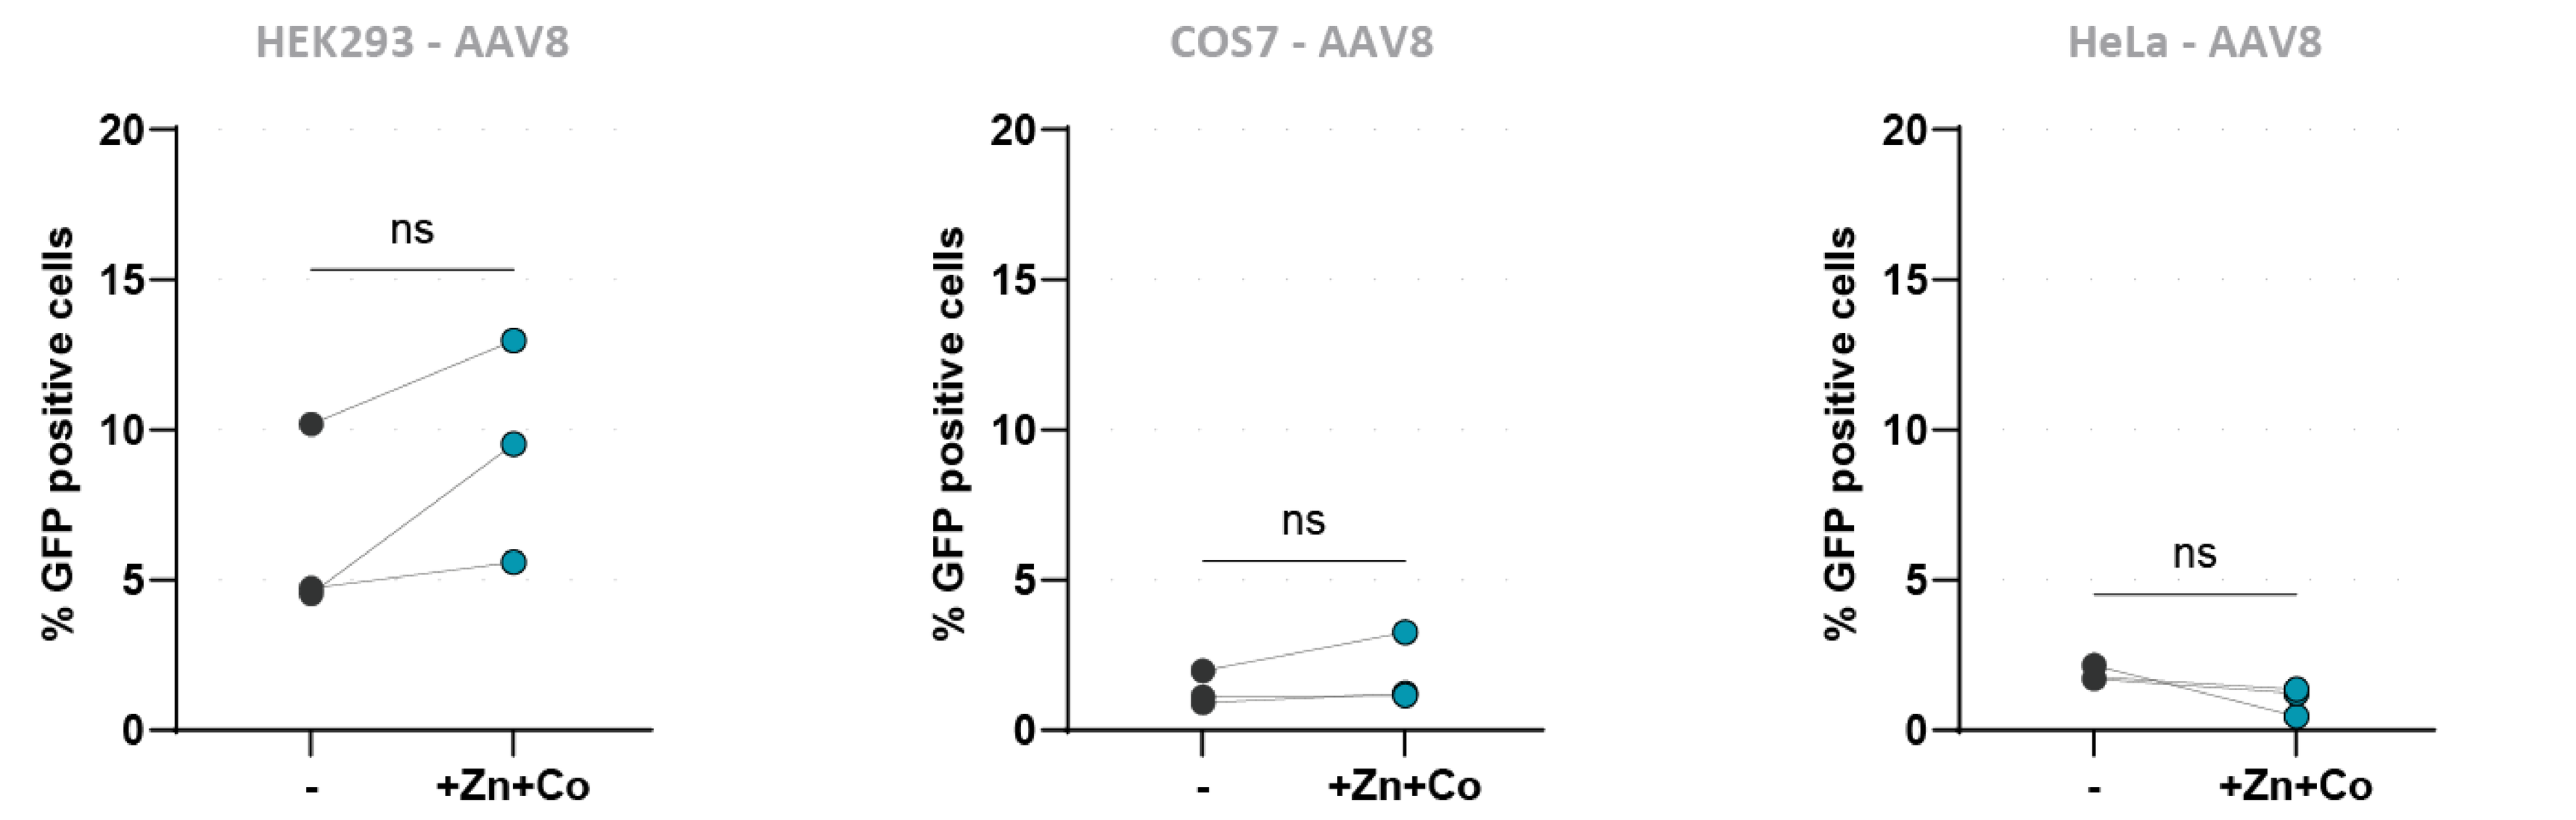
HEK293 - AAV8
COS7 - AAV8
HeLa - AAV8
% GFP positive cells
ns
-
+Zn+Co

Figure 2. Effect of zinc and cobalt in AAV8-CAG-EGFP transduction. Each pair of points represents the average of triplicates per experiment. MOI (vg/cell): 5000 (HEK), 1000 (COS), 200 (HeLa). ns = not significant vs untreated (paired t-test).

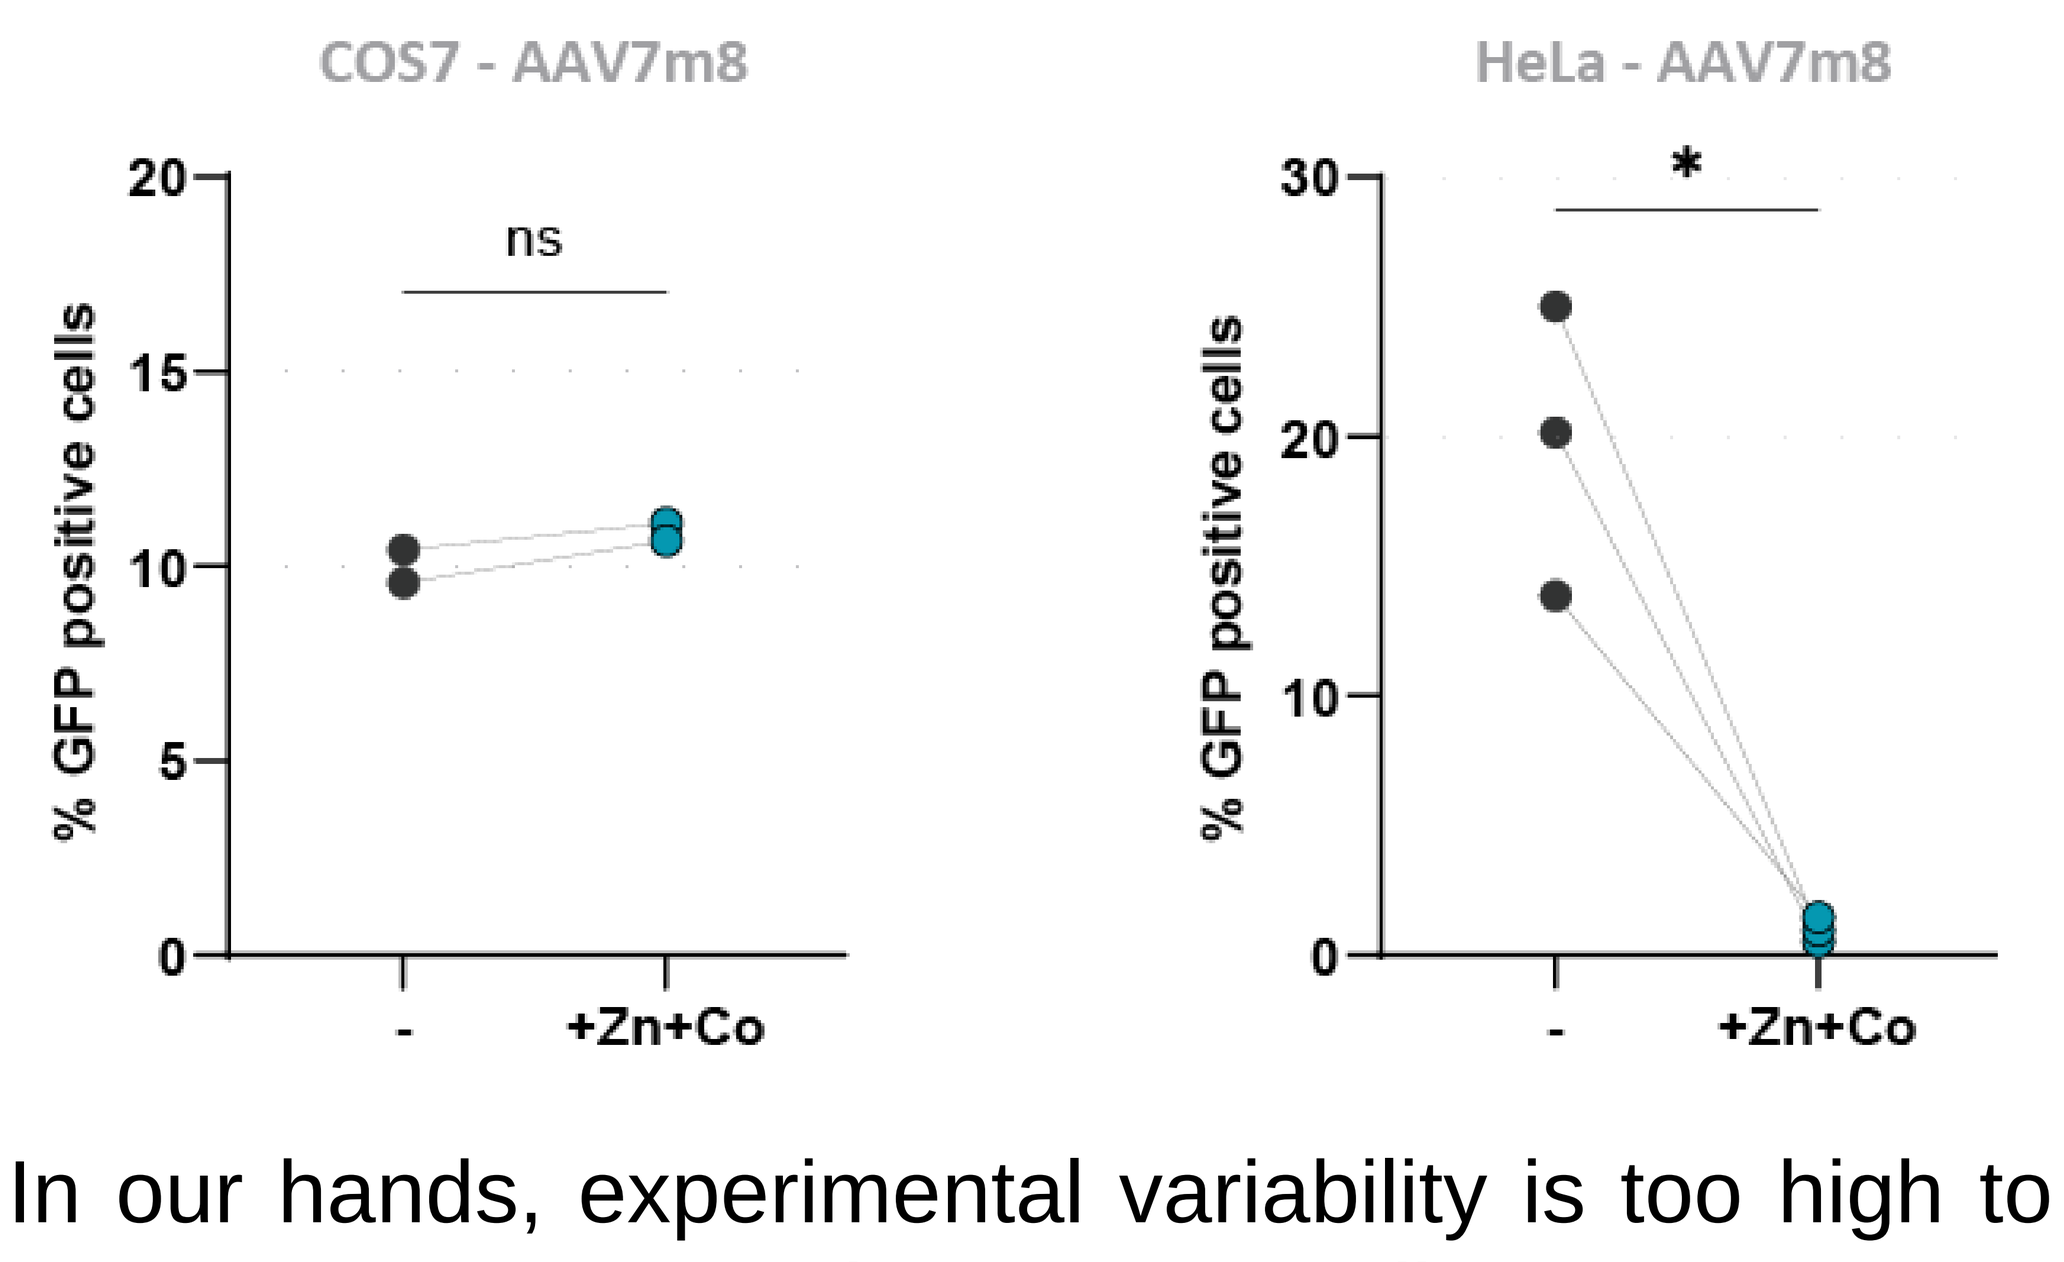
COS7 - AAV7m8
HeLa - AAV7m8
% GFP positive cells
ns
*
-
+Zn+Co

The same treatment has also no effect in COS cells transduced with 7m8 whereas it seems to even impair transduction for 7m8 (Fig.3).

Figure 3. Effect of zinc and cobalt in AAV7m8-CAG-EGFP transduction. Each pair of points represents the average of triplicates per experiment. MOI (vg/cell) 50. * $p<0.05$ vs untreated (paired t-test).

In our hands, experimental variability is too high to ensure reproducible conditions, with experiments often failing due to cell death. Even though media supplementation has the attraction of being easily transferable between platforms, metal ion treatment relies on stress induction and results in unreliable outcomes. The most promising trend was observed in ARPE19 cells and over longer periods of time, suggesting that this treatment might only be suitable for slow dividing, contact inhibited and/or suspension cell lines.

Conclusions and outlook

The mechanism by which metal ions and AAVR enhance transduction is unknown. For metals, a stress response was hypothesized[1], whereas AAVR is believed to be act on a post-attachment step related with trafficking rather than membrane attachment[3,5].

Other molecules have been explored in the enhancement of AAV transduction: 1) Topoisomerase inhibitors: enhance vector-mediated transgene expression by increasing efficiency of the second-strand synthesis, in particular several reports exist for the effect of etoposide[6,7,8] and doxorubicin[9,10] 2) DNA synthesis inhibitors such as hydroxyurea[11,6] 3) Proteasome inhibitors like LLnL[9].

Although studies on the effects of these molecules being vector or cell line dependent can be found, the work on topoisomerase inhibitors points towards a more general enhancement and it should therefore be further explored. In addition, we would like to investigate whether combining different compounds would have additive effects.

AAVR over-expression

KIAA0319L was identified as the most important gene involved in AAV infection[2]. It encodes a transmembrane protein known as “multiserotype AAV receptor” or AAVR. Several studies focused on the effect of AAVR knock-out, reporting no to <10 fold AAV infection rate for different serotypes and cell lines[2,3]. We therefore hypothesized whether over-expression of the protein could lead to enhanced transduction and tested this effect in transient transfections (Fig. 4).

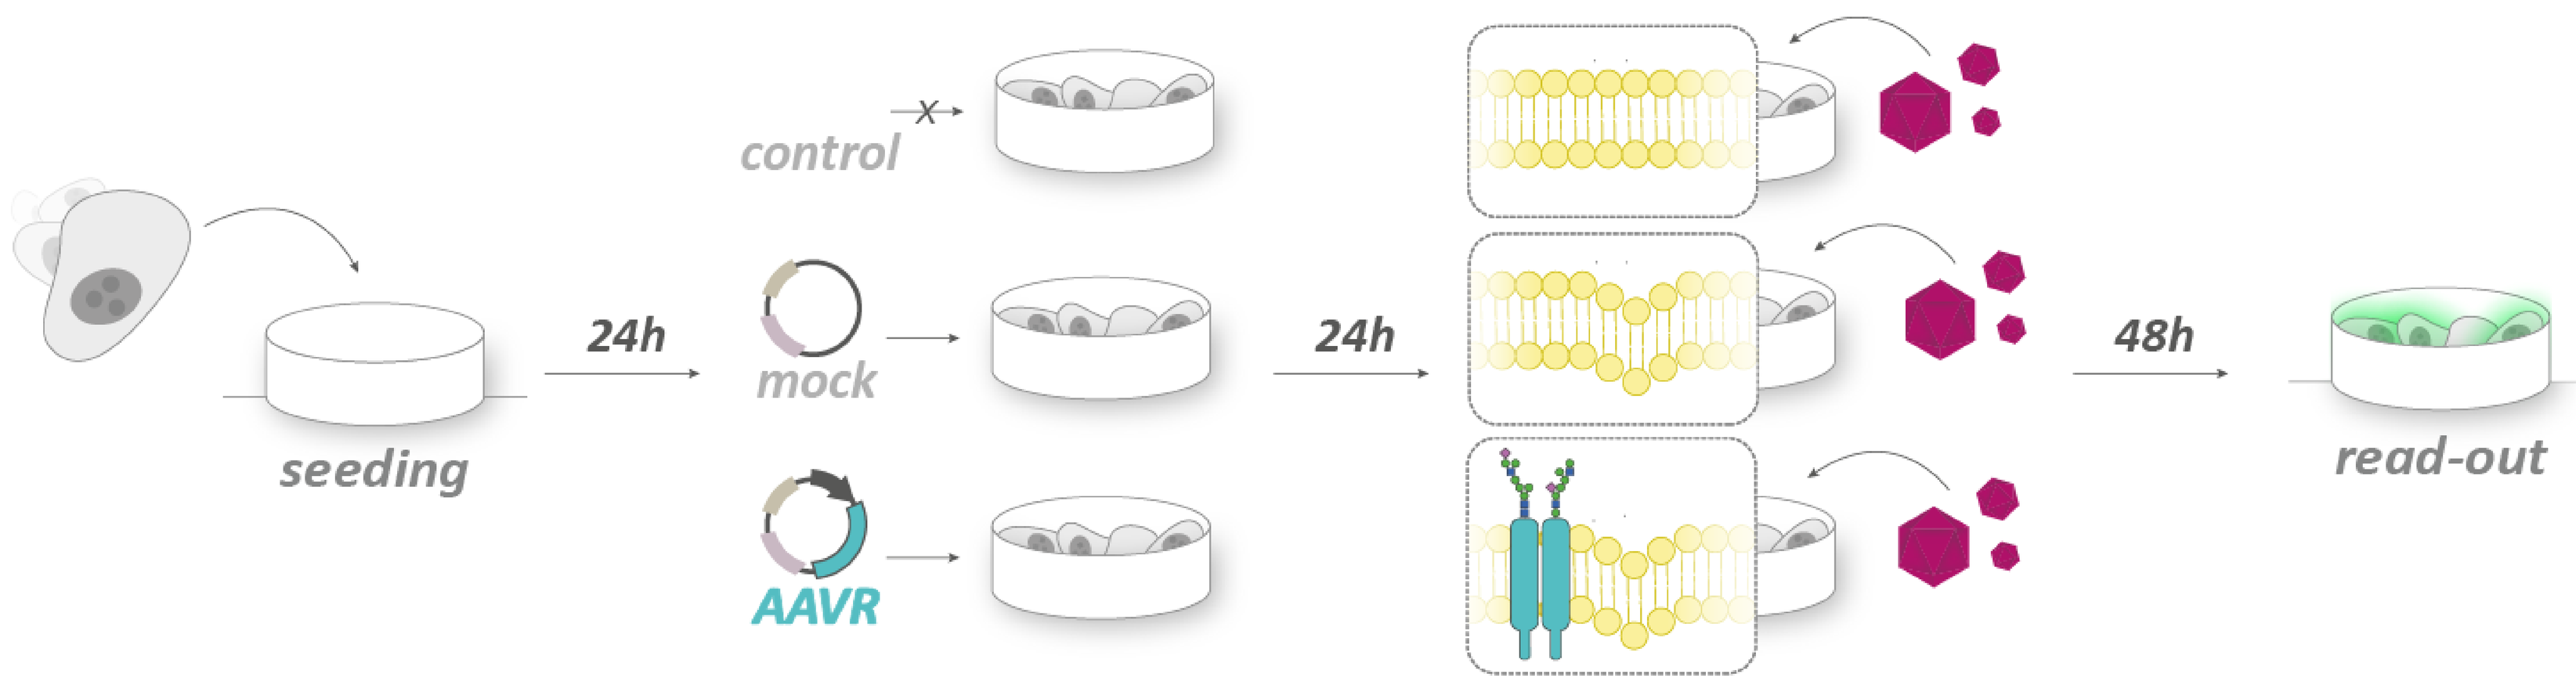
seeding
24h
control
mock
AAVR
24h
48h
read-out

Figure 4. Schematic of the protocol for assessing the effect of AAVR over-expression via transient transfection.

Expression of AAVR prior to AAV addition resulted in a significant increase in AAV8 transducibility in HEK cells (> 3.5 fold), although it yielded no differences in HeLa nor COS cells (Fig.5). Trends remained the same with different MOIs *(data not shown).*

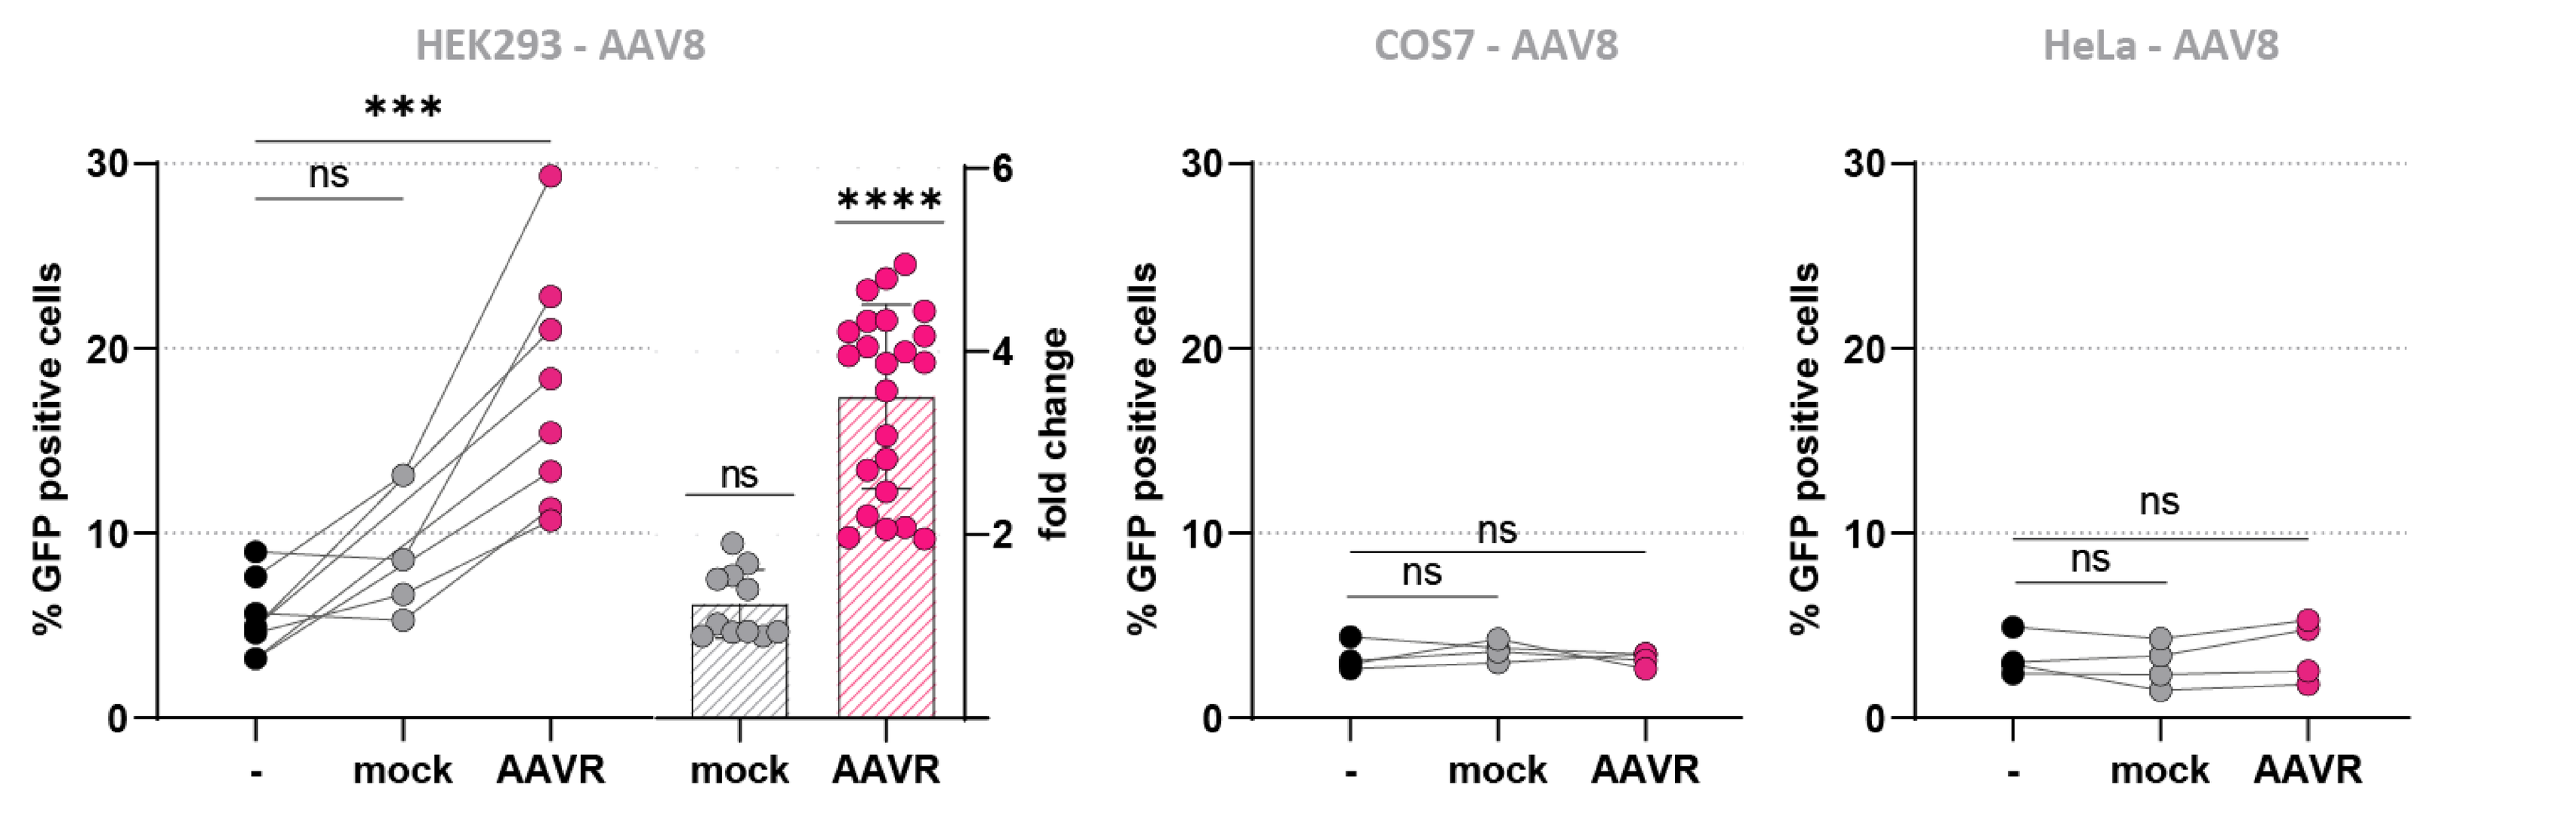
HEK293 - AAV8
COS7 - AAV8
HeLa - AAV8
% GFP positive cells
fold change

ns
-
mock
AAVR

Figure 5. Effect of AAVR in AAV8-CAG-EGFP transduction. Each pair of points represents the average of triplicates per experiment. *** $p<0.001$ vs untreated (paired t-test). In HEK barplot: points represent individual data normalized against the average of the control triplicate per experiment. **** $p<0.0001$ (one-way ANOVA vs untreated). MOI: same as Fig.2.

Over-expression had no effect for AAV7m8 in HEK nor HeLa, but it showed a negative impact in COS cells (Fig. 6) (similar trend was reported for 84-31 cells[4]).
A small increase was observed when testing the effect on AAV5 transduction in HEK cells *(data not shown),* but variability of mock control was too large to be conclusive.

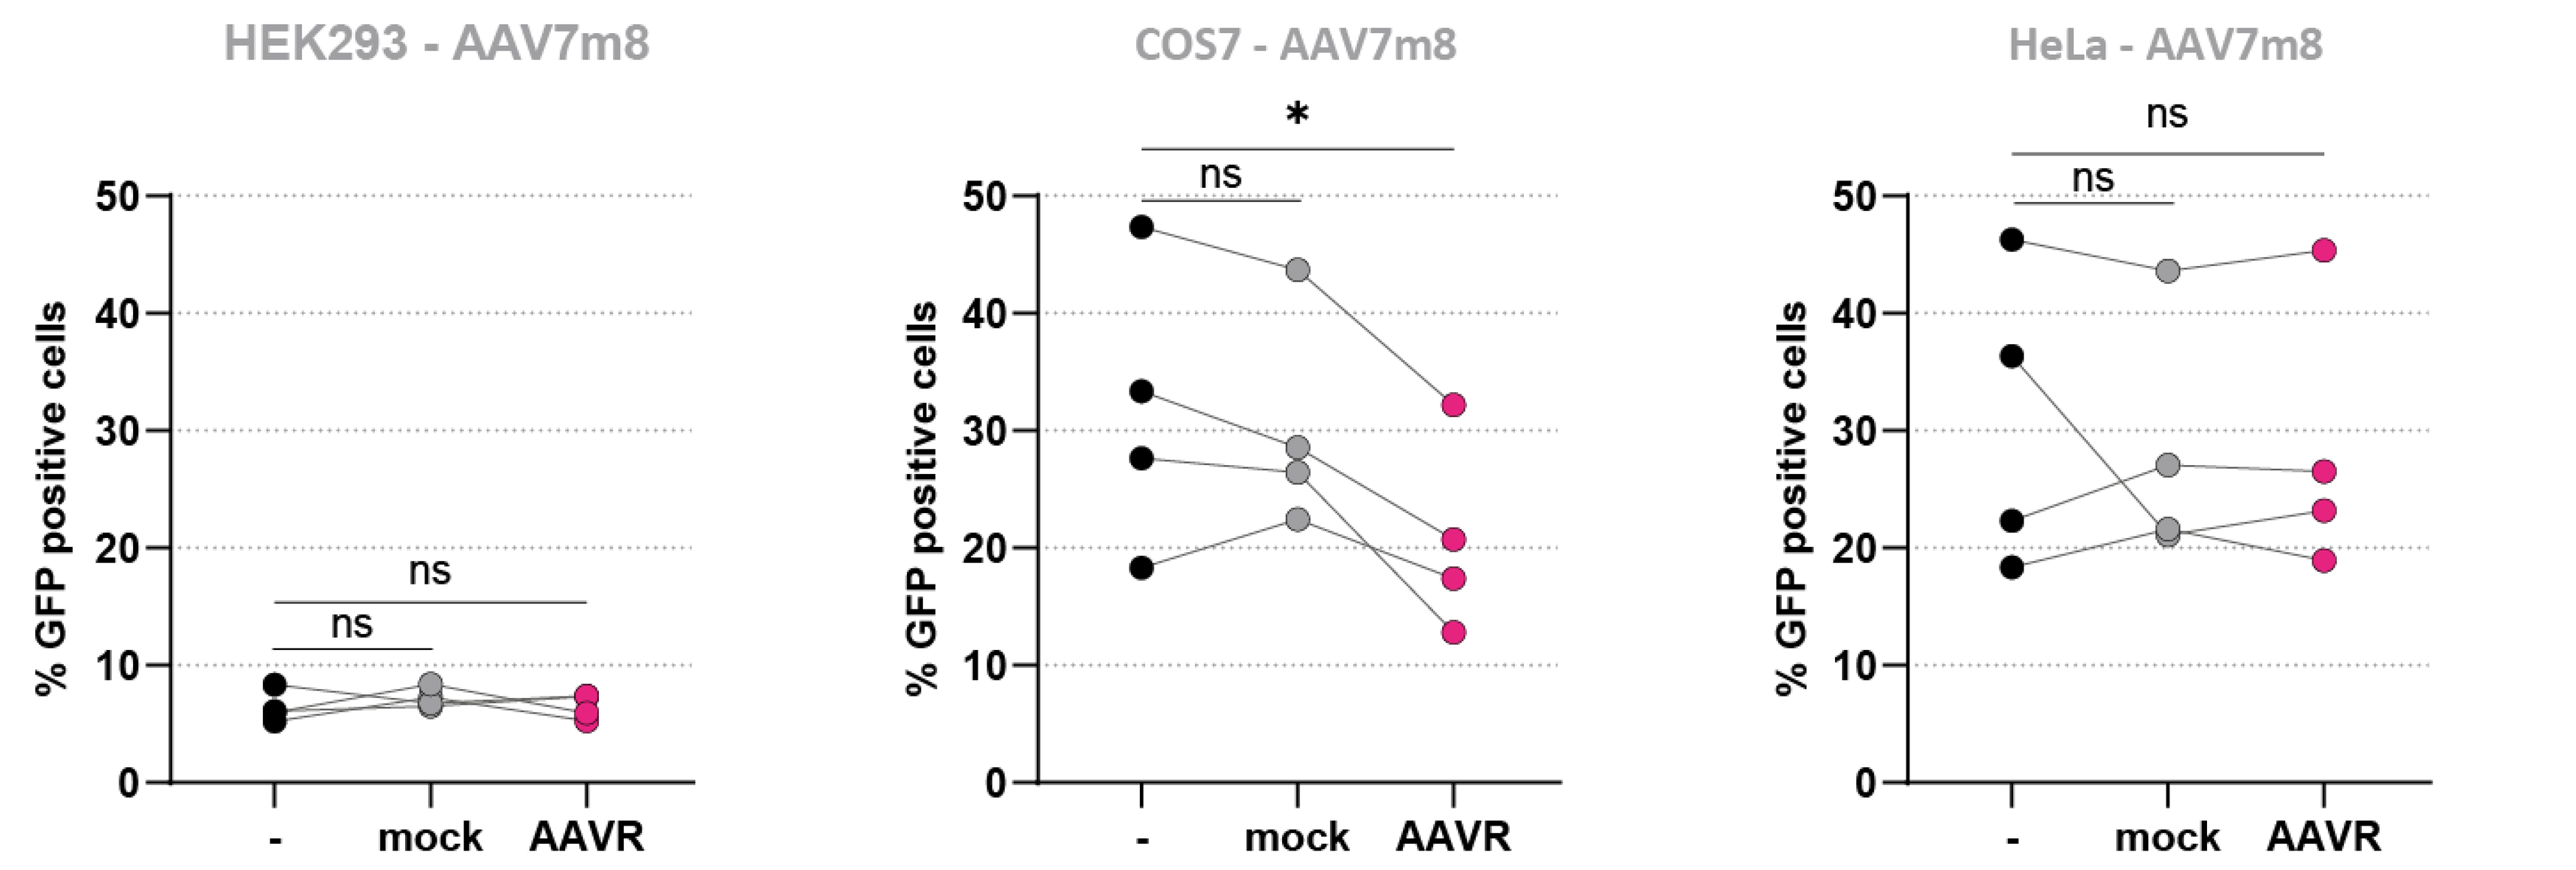
HEK293 - AAV7m8
COS7 - AAV7m8
HeLa - AAV7m8
% GFP positive cells
ns
*
-
mock
AAVR

Figure 6. Effect of AAVR in AAV7m8-CAG-EGFP transduction. Each pair of points represents the average of triplicates per experiment. * $p<0.05$ vs untreated (paired t-test). MOI (vg/cell) 50 for all cell lines.

Preliminary data on stable cell lines does not seem to indicate that these results are due to a transfection bias, since they show the same cell type/serotype trends and enhancement in HEK cells for AAV8 only increases to 5 fold *(data not shown).*
The effect of AAVR over-expression is cell line and serotype dependent and therefore does not represent a methodology universally applicable to enhance transducibility. In addition, cells need to be either transiently or stably manipulated, which might impose a limitation for implementation in more clinically relevant models.

Acknowledgements

To Sarah Wells for experimental assistance and to Tijmen Vermeule for AAV production

Methods

Cell lines HEK293, HeLa, COS-7 were chosen as “commonly used” and because they are easy to maintain, to expand, and to manipulate, allowing for inexpensive and accessible implementation of protocols. Seeded in 24-well plates at 120000(HEK)/60000(HeLa)/ 75000(COS) cells/well. Metal ion treatment: MOI adjusted by counting duplicates. 1 day after treatment 1V media was added on top, then full media exchange the day after. AAVR over-expression: Lipofectamine2000 transfections with endotoxin-free preps at 200 (HEK/HeLa) / 500ng DNA/well (COS). MOI adjusted by counting duplicates for each treatment. Flow cytometry: acquired in BD FACSLyric. Analysed in FlowJo. EGFP measured with 488 + 507LP(527/32) in cells gated by population (SSC-AxFSC-A) and singlets (FSC-WxFSC-A). AAV production: triple transfection in suspension HEK cells. Purification AAV-X chromatography. Buffer: PBS, 200mM NaCl, 0.001% Pluronic. Titration: TaqMan with BGHpA target.

References

(1) Rambhai, 2020 (2) Pillay, 2016 (3) Dudek, 2018 (4) McDougald, 2019 (5) Meyer, 2022 (6) Ju, 2004 (7) Kanazawa, 2004 (8) Nicolson, 2016 (9) Yan, 2004 (10) Gong, 2021 (11) Russell, 1995