P124

Promoter Engineering Platform at MeiraGTx

Josefa M. Sullivan[1], Dustin Lee[1], Chenjin Jin[1], Jessica Schmerler[1], Natalia Fabela[1], Enrico Mossotto[1], Edgar Hernandez[1], Kassiani Kytidou[2], Sabine Van der Sanden[2], Mostafa Zamatkesh[2], Diederik Lokhorst[2], Matthew J. During[1], Alexandria Forbes[1], & Ce Feng Liu[1]

[1]MeiraGTx New York [2]MeiraGTx Amsterdam

Promoter engineering can improve strength, specificity, and safety of gene therapies

Benefits of Promoter Engineering for Gene Therapies

- Precise control of therapeutic gene expression[1]
- Cell-specific expression independent of capsid[2]
- Increased potency potentially decreases immune responses and safety risks[3]
- Kinetics impact durability of gene therapy
- Reducing size without sacrificing strength or specificity allows efficient packaging or larger cargo[4]

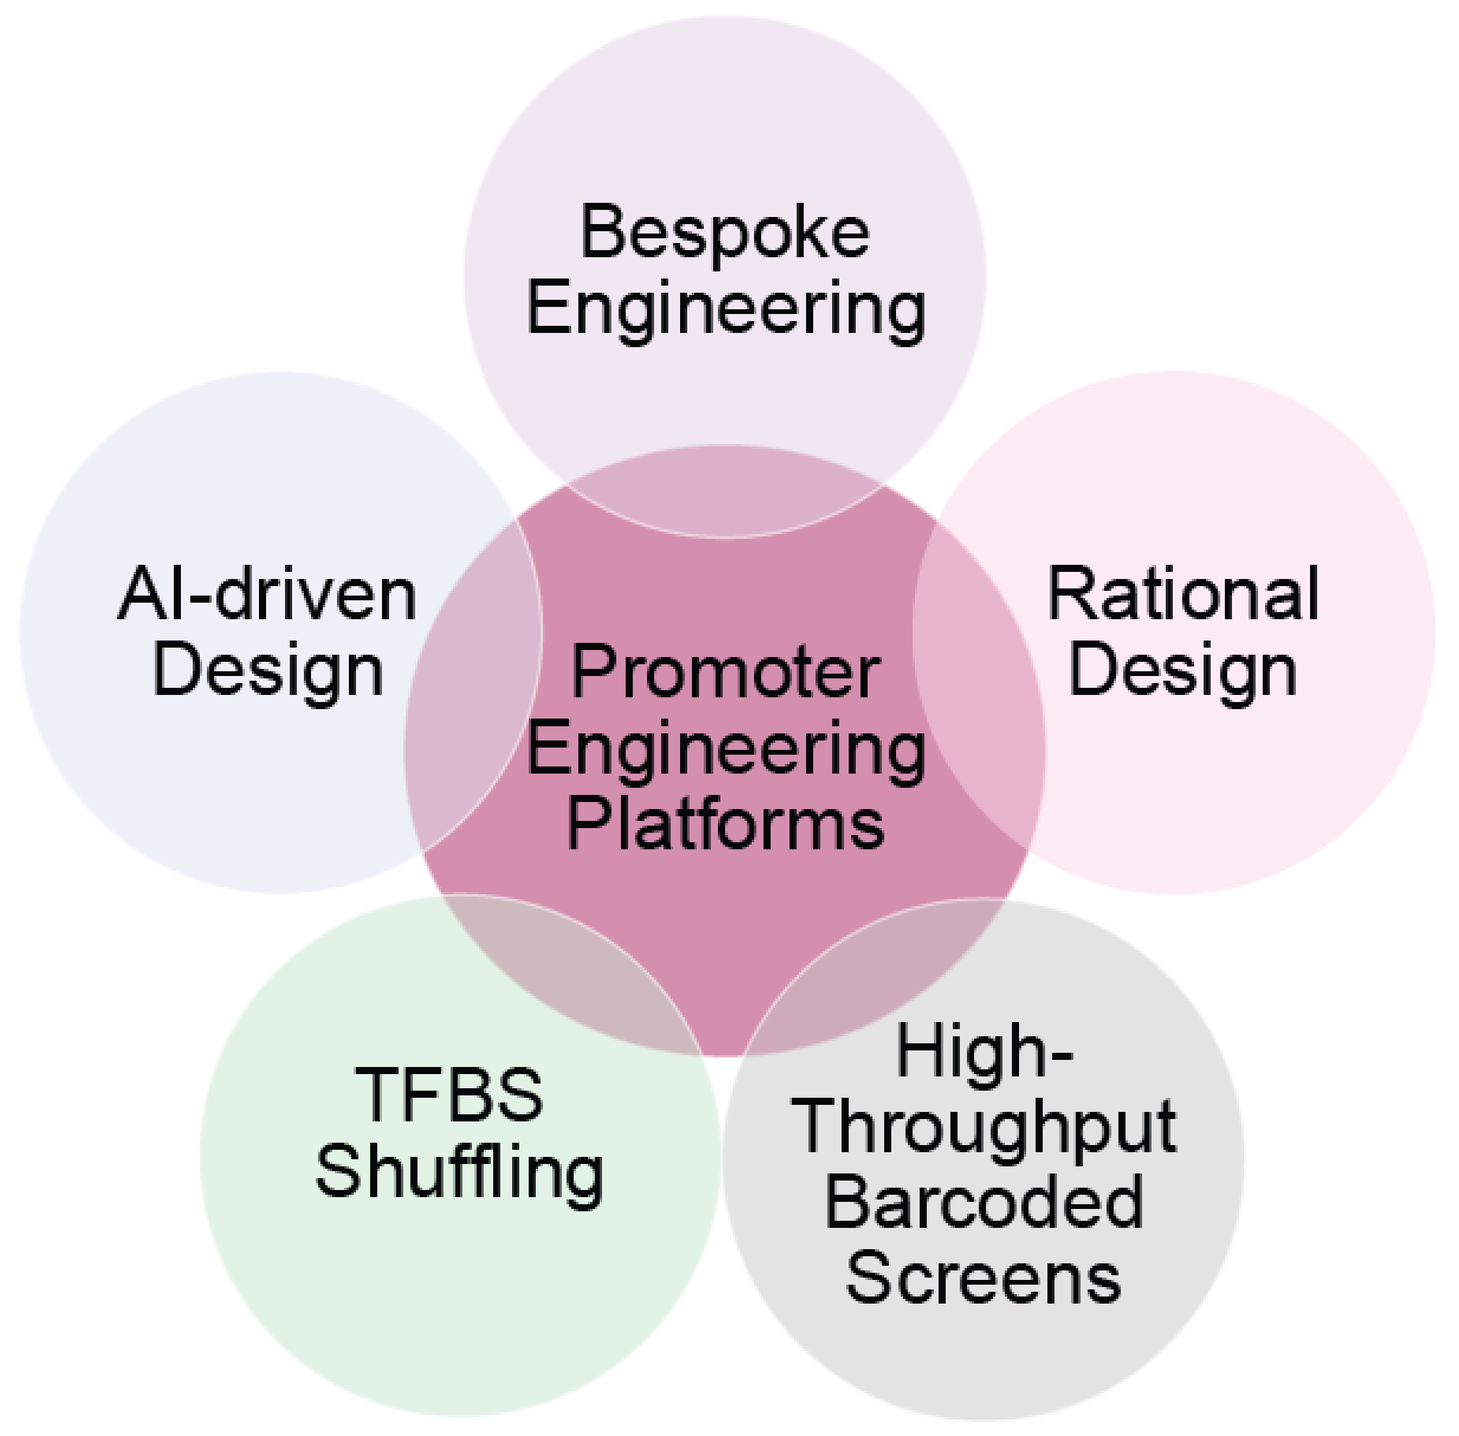
Bespoke Engineering
AI-driven Design
Rational Design
Promoter Engineering Platforms
TFBS Shuffling
High-Throughput Barcoded Screens

We have built libraries of synthetic promoters and promoter elements available for viral vector optimization. As these regulatory elements are context-dependent, different elements may be most appropriate in different gene sequence and cellular contexts.

Designing smaller or more potent CAG promoter variants

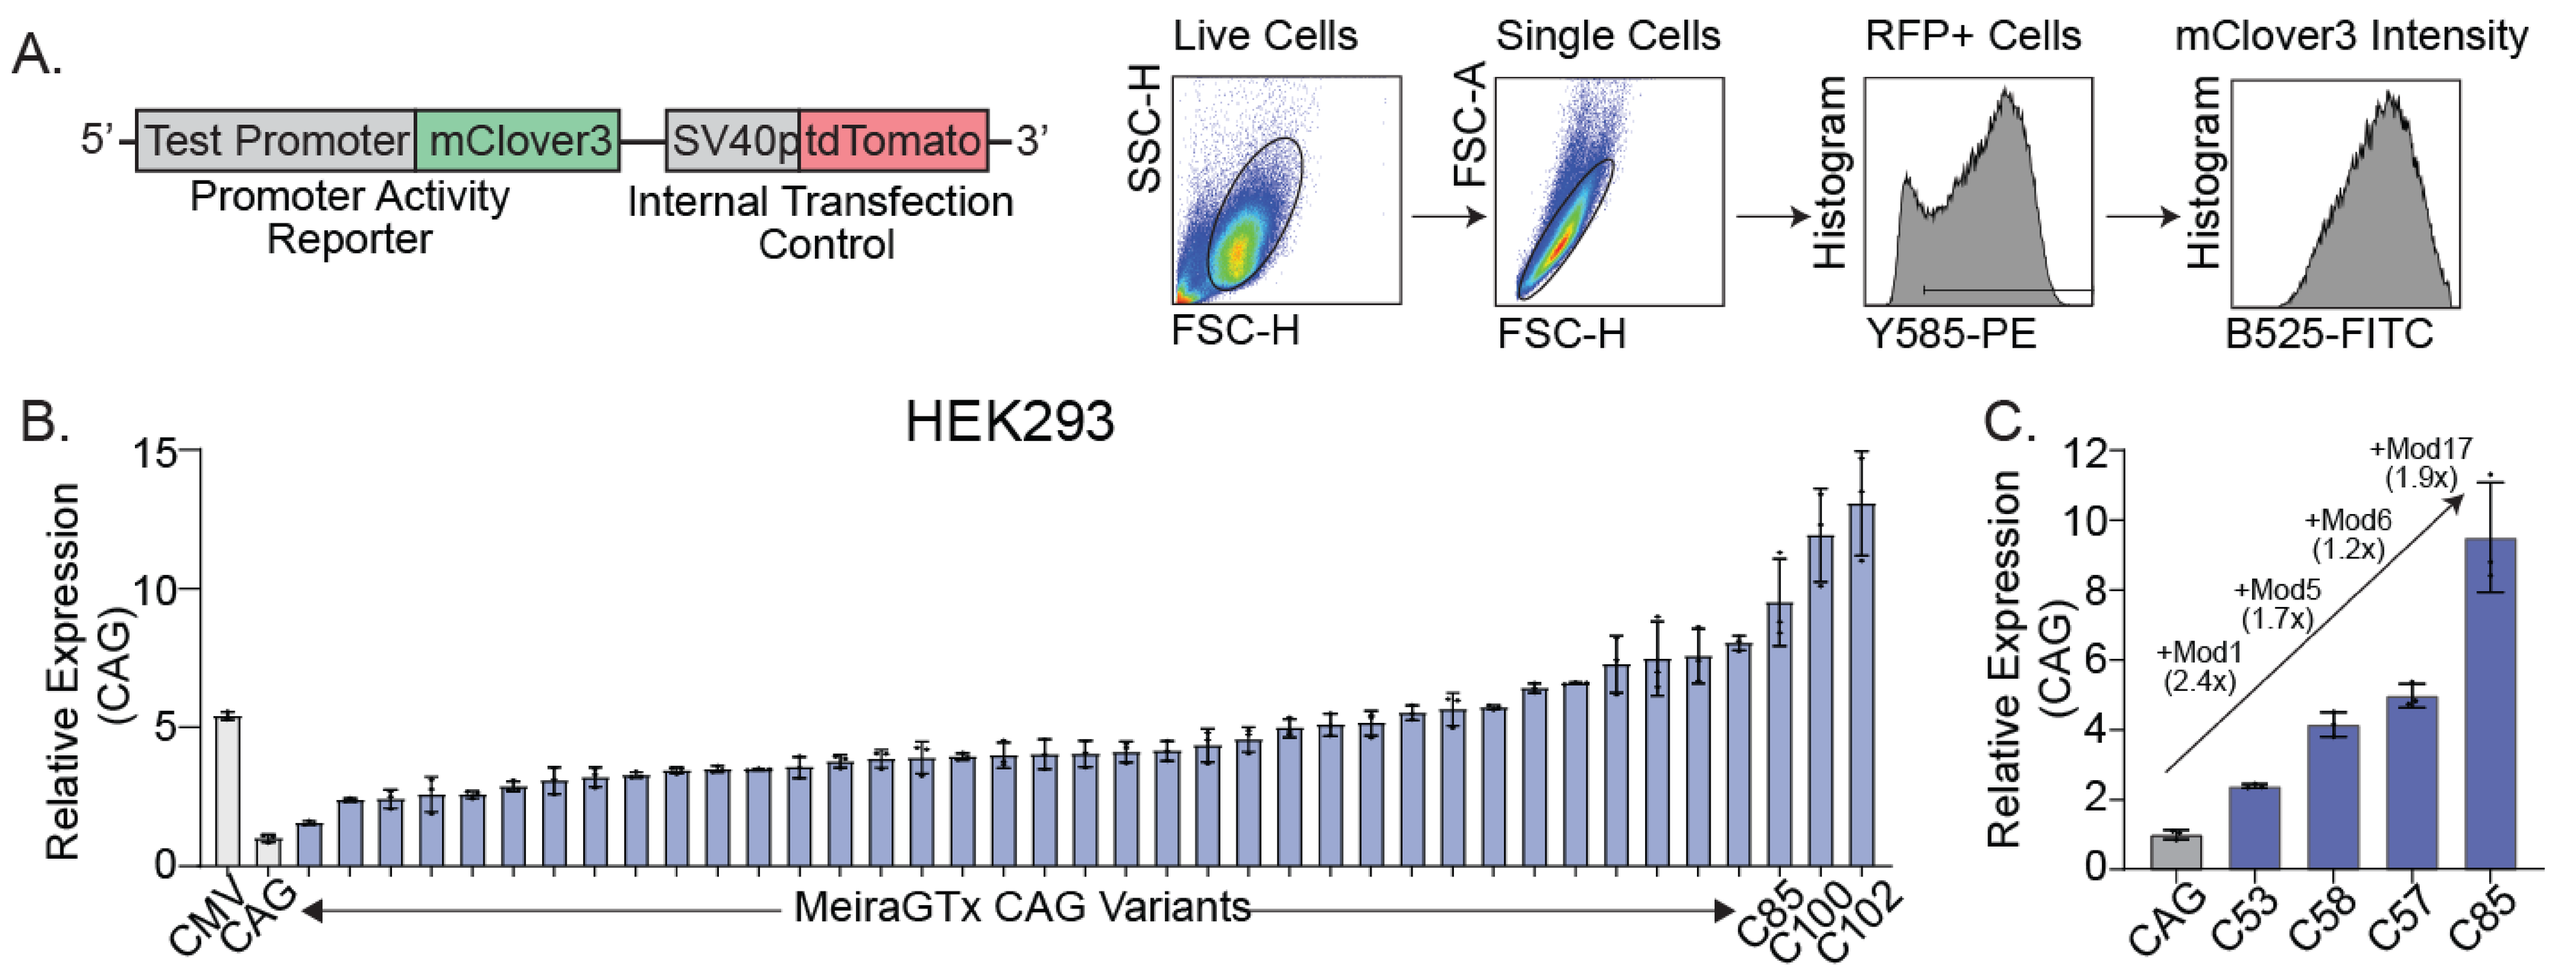
A.
5' Test Promoter mClover3 SV40p tdTomato 3'
Promoter Activity Reporter
Internal Transfection Control
Live Cells
Single Cells
RFP+ Cells
mClover3 Intensity
SSC-H
FSC-H
FSC-A
Histogram
Y585-PE
B525-FITC
B.
HEK293
Relative Expression (CAG)
CMV
CAG
MeiraGTx CAG Variants
C85
C100
C102
C.
+Mod1 (2.4x)
+Mod5 (1.7x)
+Mod6 (1.2x)
+Mod17 (1.9x)
CAG C53 C58 C57 C85

Promoter	CMV	CAG	C53	C54	C55	C56	C57	C58	C63	C64	C65	C66	C67	C68	C69	C70	C83	C84	C85	C86	C87	C88	C89	C90	C91	C99	C100	C101	C102	C108
Size (kb)	0.7	1.8	1.7	2.1	1.9	1.7	1.9	1.9	2.0	1.7	1.5	1.1	1.7	1.4	1.2	1.1	1.9	2.0	2.0	2.0	2.0	1.9	2.0	2.0	2.2	2.2	2.0	2.0	2.0	1.7
Potency (HEK293T)	5.3	1.0	2.4	3.1	2.9	2.4	5.0	4.2	5.5	3.4	4.0	3.2	4.0	3.6	3.8	4.0	6.4	4.1	9.5	5.2	4.0	8.0	3.5	7.6	5.1	4.4	11.9	7.3	13.1	7.5
Potency (C2C12)	5.2	1.0	1.8	2.7	2.9	1.4	3.9	3.8	3.4	2.5	2.1	1.4	1.5	1.4	1.1	1.3	3.9	3.4	5.1	3.3	2.1	5.3	2.3	5.1	1.6	1.7	3.8	2.1	3.9	2.0

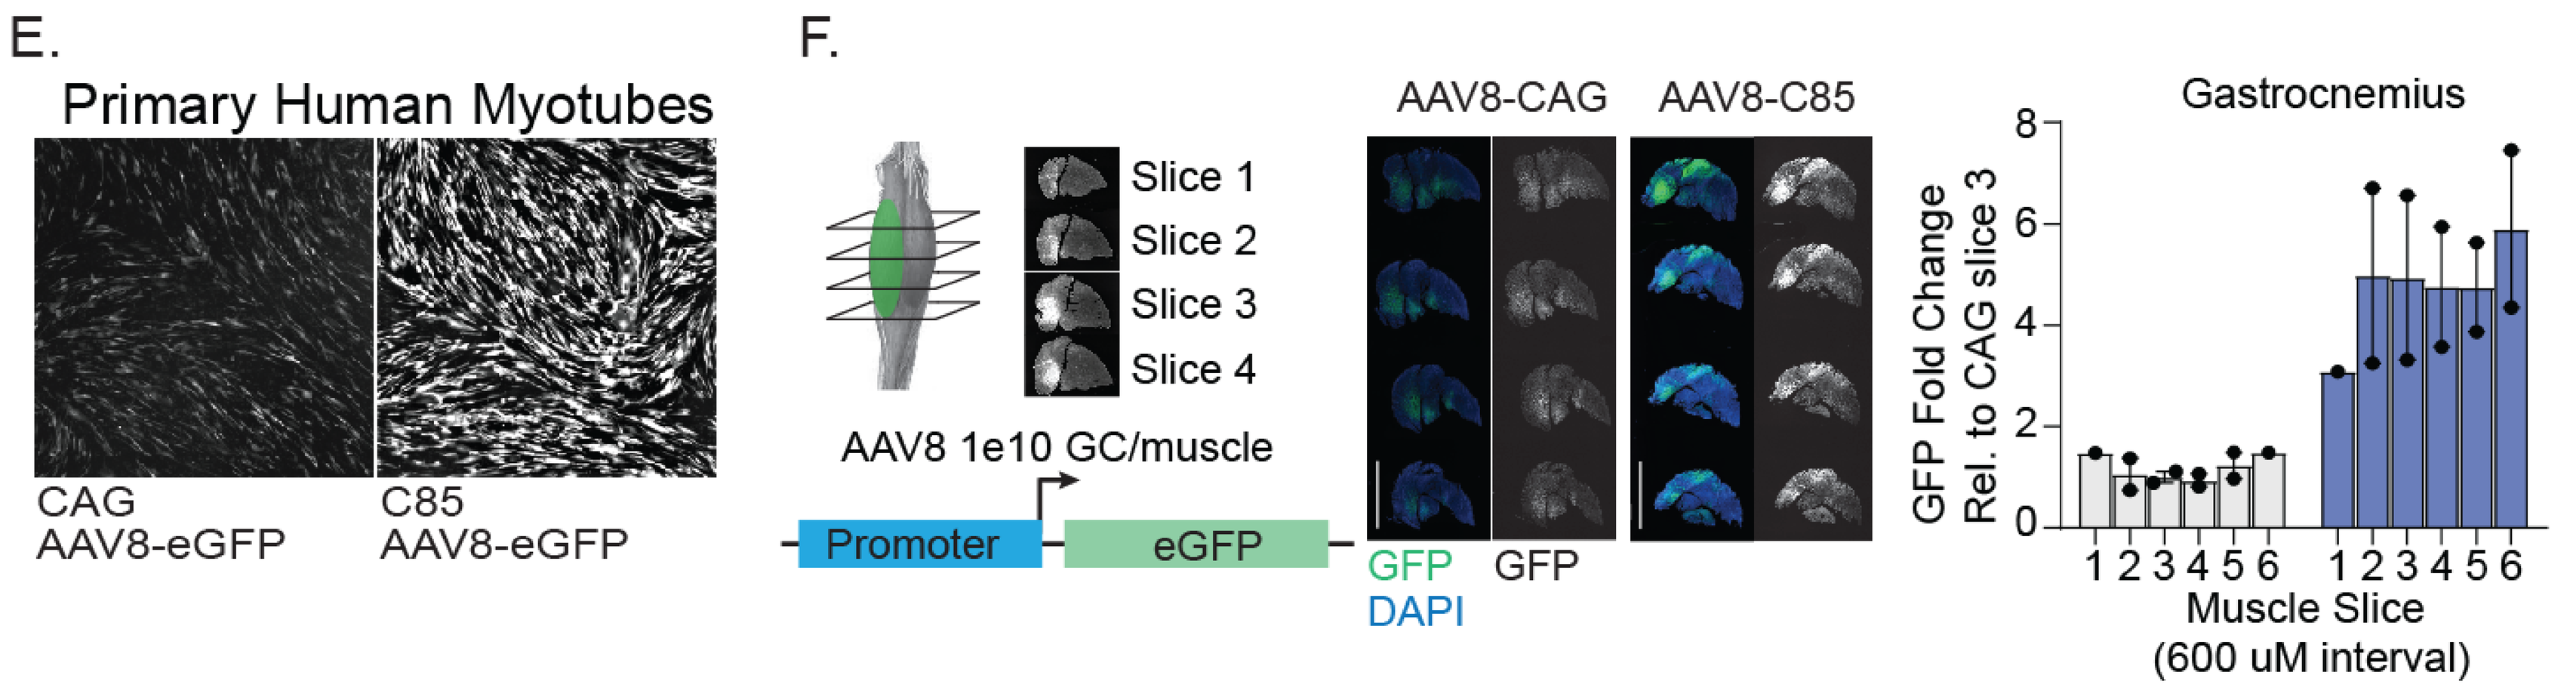
E.
Primary Human Myotubes
CAG AAV8-eGFP
C85 AAV8-eGFP
F.
Slice 1
Slice 2
Slice 3
Slice 4
AAV8 1e10 GC/muscle
Promoter
eGFP
AAV8-CAG
AAV8-C85
GFP DAPI
GFP
Gastrocnemius
GFP Fold Change Rel. to CAG slice 3
Muscle Slice (600 uM interval)

(A) FACS-based assay to screen promoter activity in vitro. (B) Screening of CAG variants in transiently transfected HEK293 cells identifies candidates that are up to 13-fold stronger than the original CAG promoter. (C) Example of the step-wise evolution of one top CAG variant. (D) Heatmap shows size (green) and activity of CAG variants in HEK293 and C2C12 cells in vitro. (E) Transduction of primary human myotubes shows increased expression from MGTx CAG variant, C85. (F) C85 drives stronger expression in vivo as indicated by native eGFP fluorescence in the mouse gastrocnemius. Scale: 5 mm

Evolution of strong, muscle-specific promoters

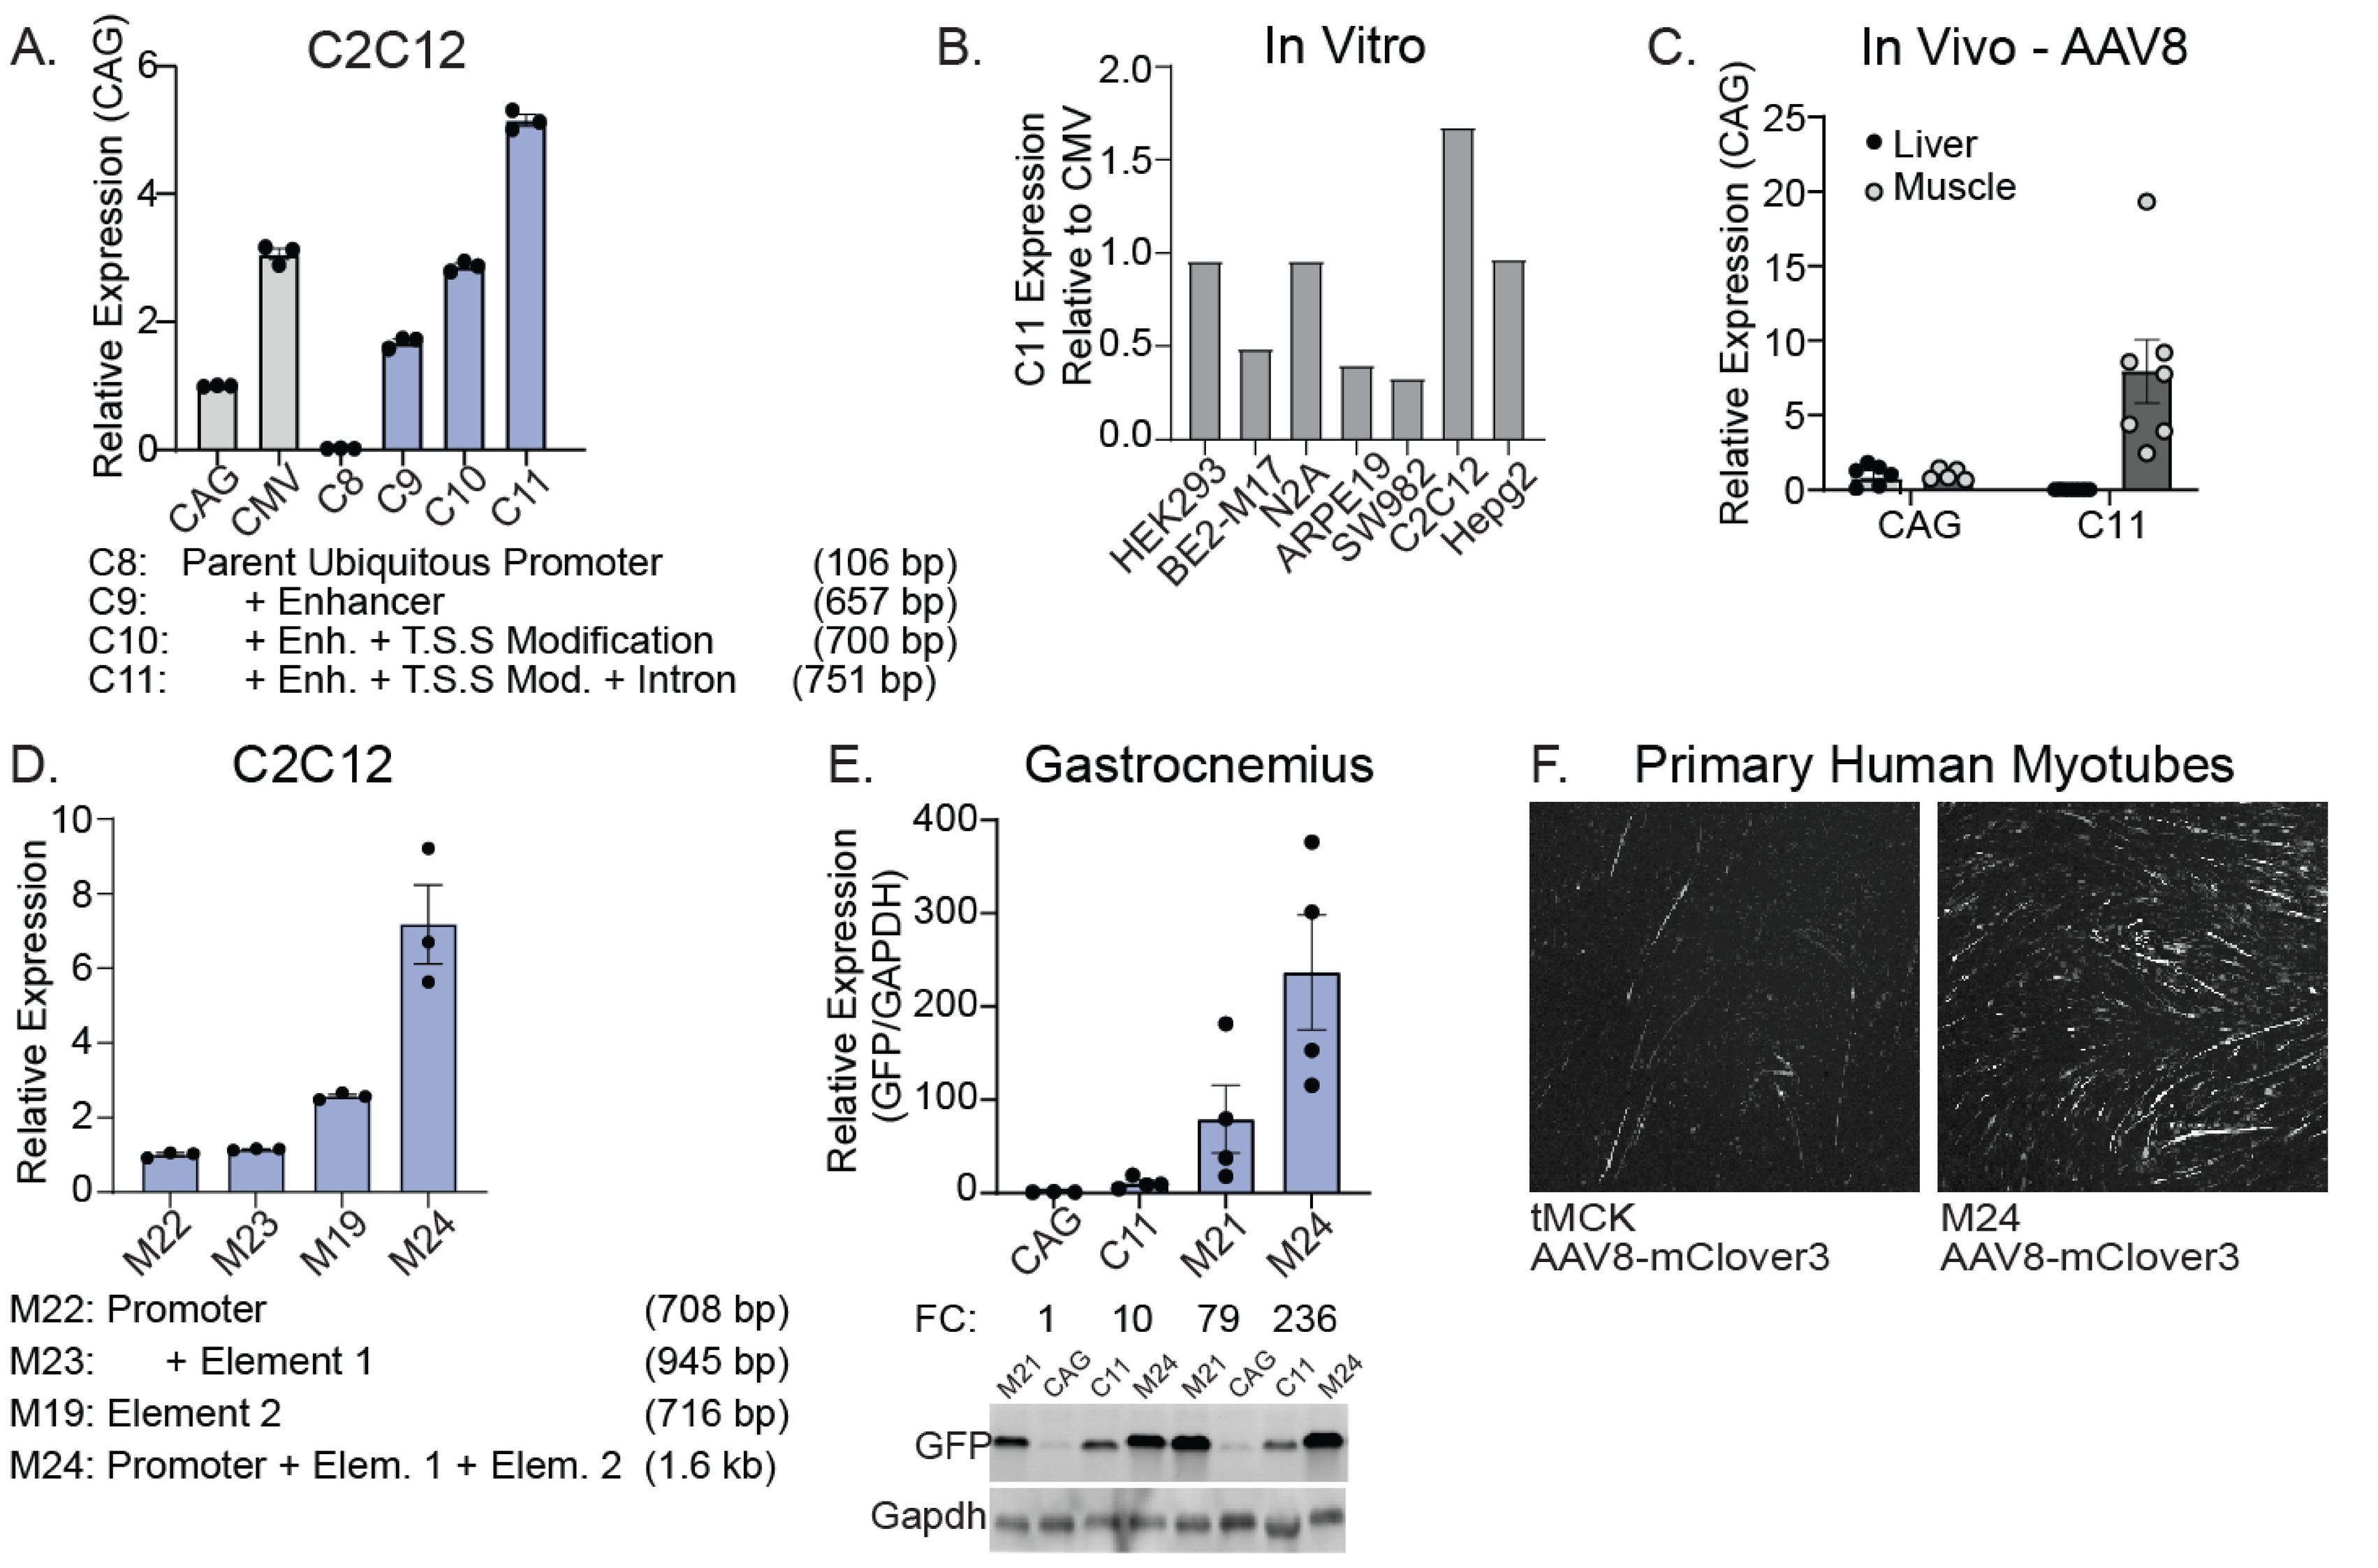
A.
C2C12
Relative Expression (CAG)
CAG CMV C8 C9 C10 C11
C8: Parent Ubiquitous Promoter (106 bp)
C9: + Enhancer (657 bp)
C10: + Enh. + T.S.S Modification (700 bp)
C11: + Enh. + T.S.S Mod. + Intron (751 bp)
B.
In Vitro
C11 Expression Relative to CMV
HEK293 BE2-M17 N2A ARPE19 SW982 C2C12 Hepg2
C.
In Vivo - AAV8
Liver
Muscle
CAG C11
D.
C2C12
Relative Expression
M22 M23 M19 M24
M22: Promoter (708 bp)
M23: + Element 1 (945 bp)
M19: Element 2 (716 bp)
M24: Promoter + Elem. 1 + Elem. 2 (1.6 kb)
E.
Gastrocnemius
Relative Expression (GFP/GAPDH)
CAG C11 M21 M24
FC: 1 10 79 236
GFP
Gapdh
F.
Primary Human Myotubes
tMCK AAV8-mClover3
M24 AAV8-mClover3

(A) Step-wise evolution of a small, constitutive promoter driving expression in transfected C2C12 cells. (B) C11driven expression in various transfected cell lines. (C) Transduction of the liver (i.v.) and muscle (i.m.) with AAV8 in wild-type, 8 week-old male mice shows C11 promoter activity is stronger in the muscle. (D) Step-wise evolution of a muscle-specific promoter (M22 to M24) rather than CAG derived promoter, in transfected C2C12 cells. (E) Western blot of transduced mouse muscle tissue. (F) Transduction of primary human myotubes with AAV8-mClover3-WPRE-SV40pA

MGTx promoters are highly potent and durable in the mouse muscle

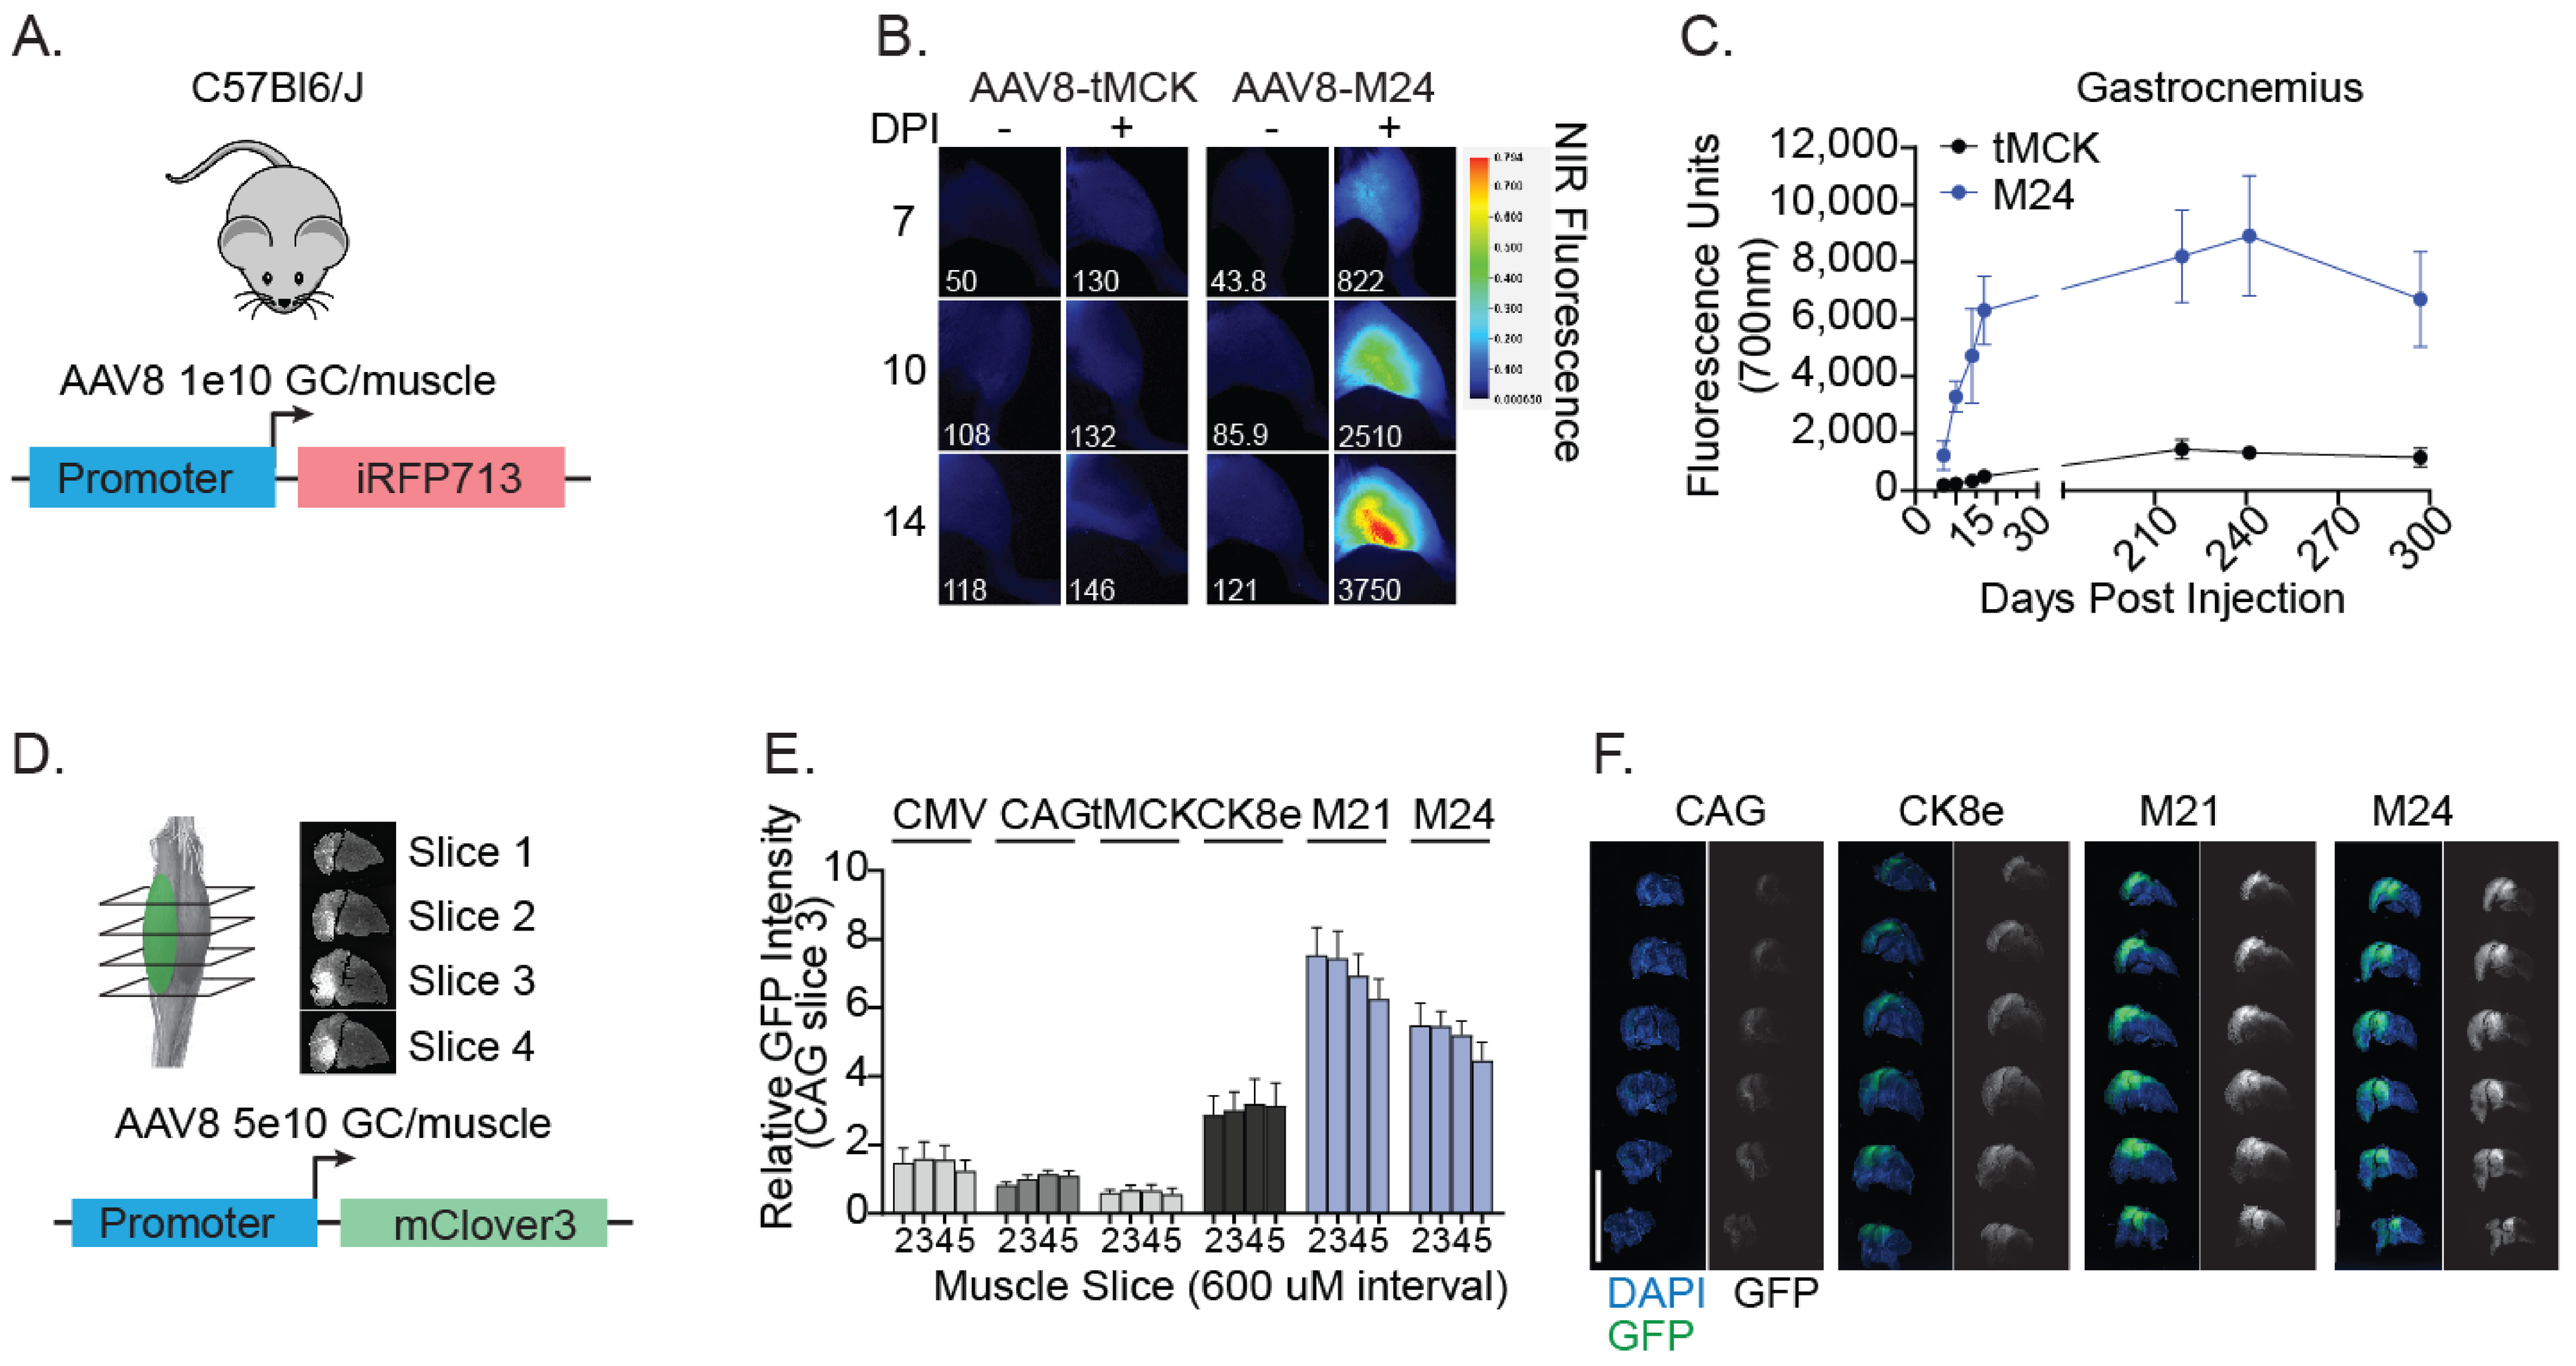
A.
C57Bl6/J
AAV8 1e10 GC/muscle
Promoter
iRFP713
B.
AAV8-tMCK
AAV8-M24
DPI
NIR Fluorescence
C.
Gastrocnemius
tMCK
M24
Fluorescence Units (700nm)
Days Post Injection
D.
Slice 1
Slice 2
Slice 3
Slice 4
AAV8 5e10 GC/muscle
Promoter
mClover3
E.
CMV CAG tMCK CK8e M21 M24
Relative GFP Intensity (CAG slice 3)
Muscle Slice (600 uM interval)
F.
CAG CK8e M21 M24
DAPI GFP
GFP

(A) AAV8-iRFP713 under control of tMCK or M24 promoter was injected intramuscular to male C57Bl6 mice at 8 weeks of age and imaged using the LICOR Pearl. (B) Representative imaging data shows uninjected (-) and injected (+) hindlimbs for AAV8-tMCK and AAV8-M24. (C) Quantification of fluorescence shows M24 has at least 7-fold higher expression than tMCK that is maintained out to 300 days post injection. (D) AAV8-eGFP was injected into the gastrocnemius of male C57Bl6 mice at 8 weeks of age under the control of CMV, CAG, TMCK, CK8e, and two strong MGTx muscle specific promoters M21 and M24. After 4 weeks, the muscle was harvested and sectioned for native eGFP fluorescence. (E) Quantification and (F) Representative images, scalebar 10 mm, show MGTx promoters M21 and M24 are stronger than CAG or CK8e in the mouse gastrocnemius

High-throughput screening of synthetic elements to identify sequences that enhance promoter activity in muscle

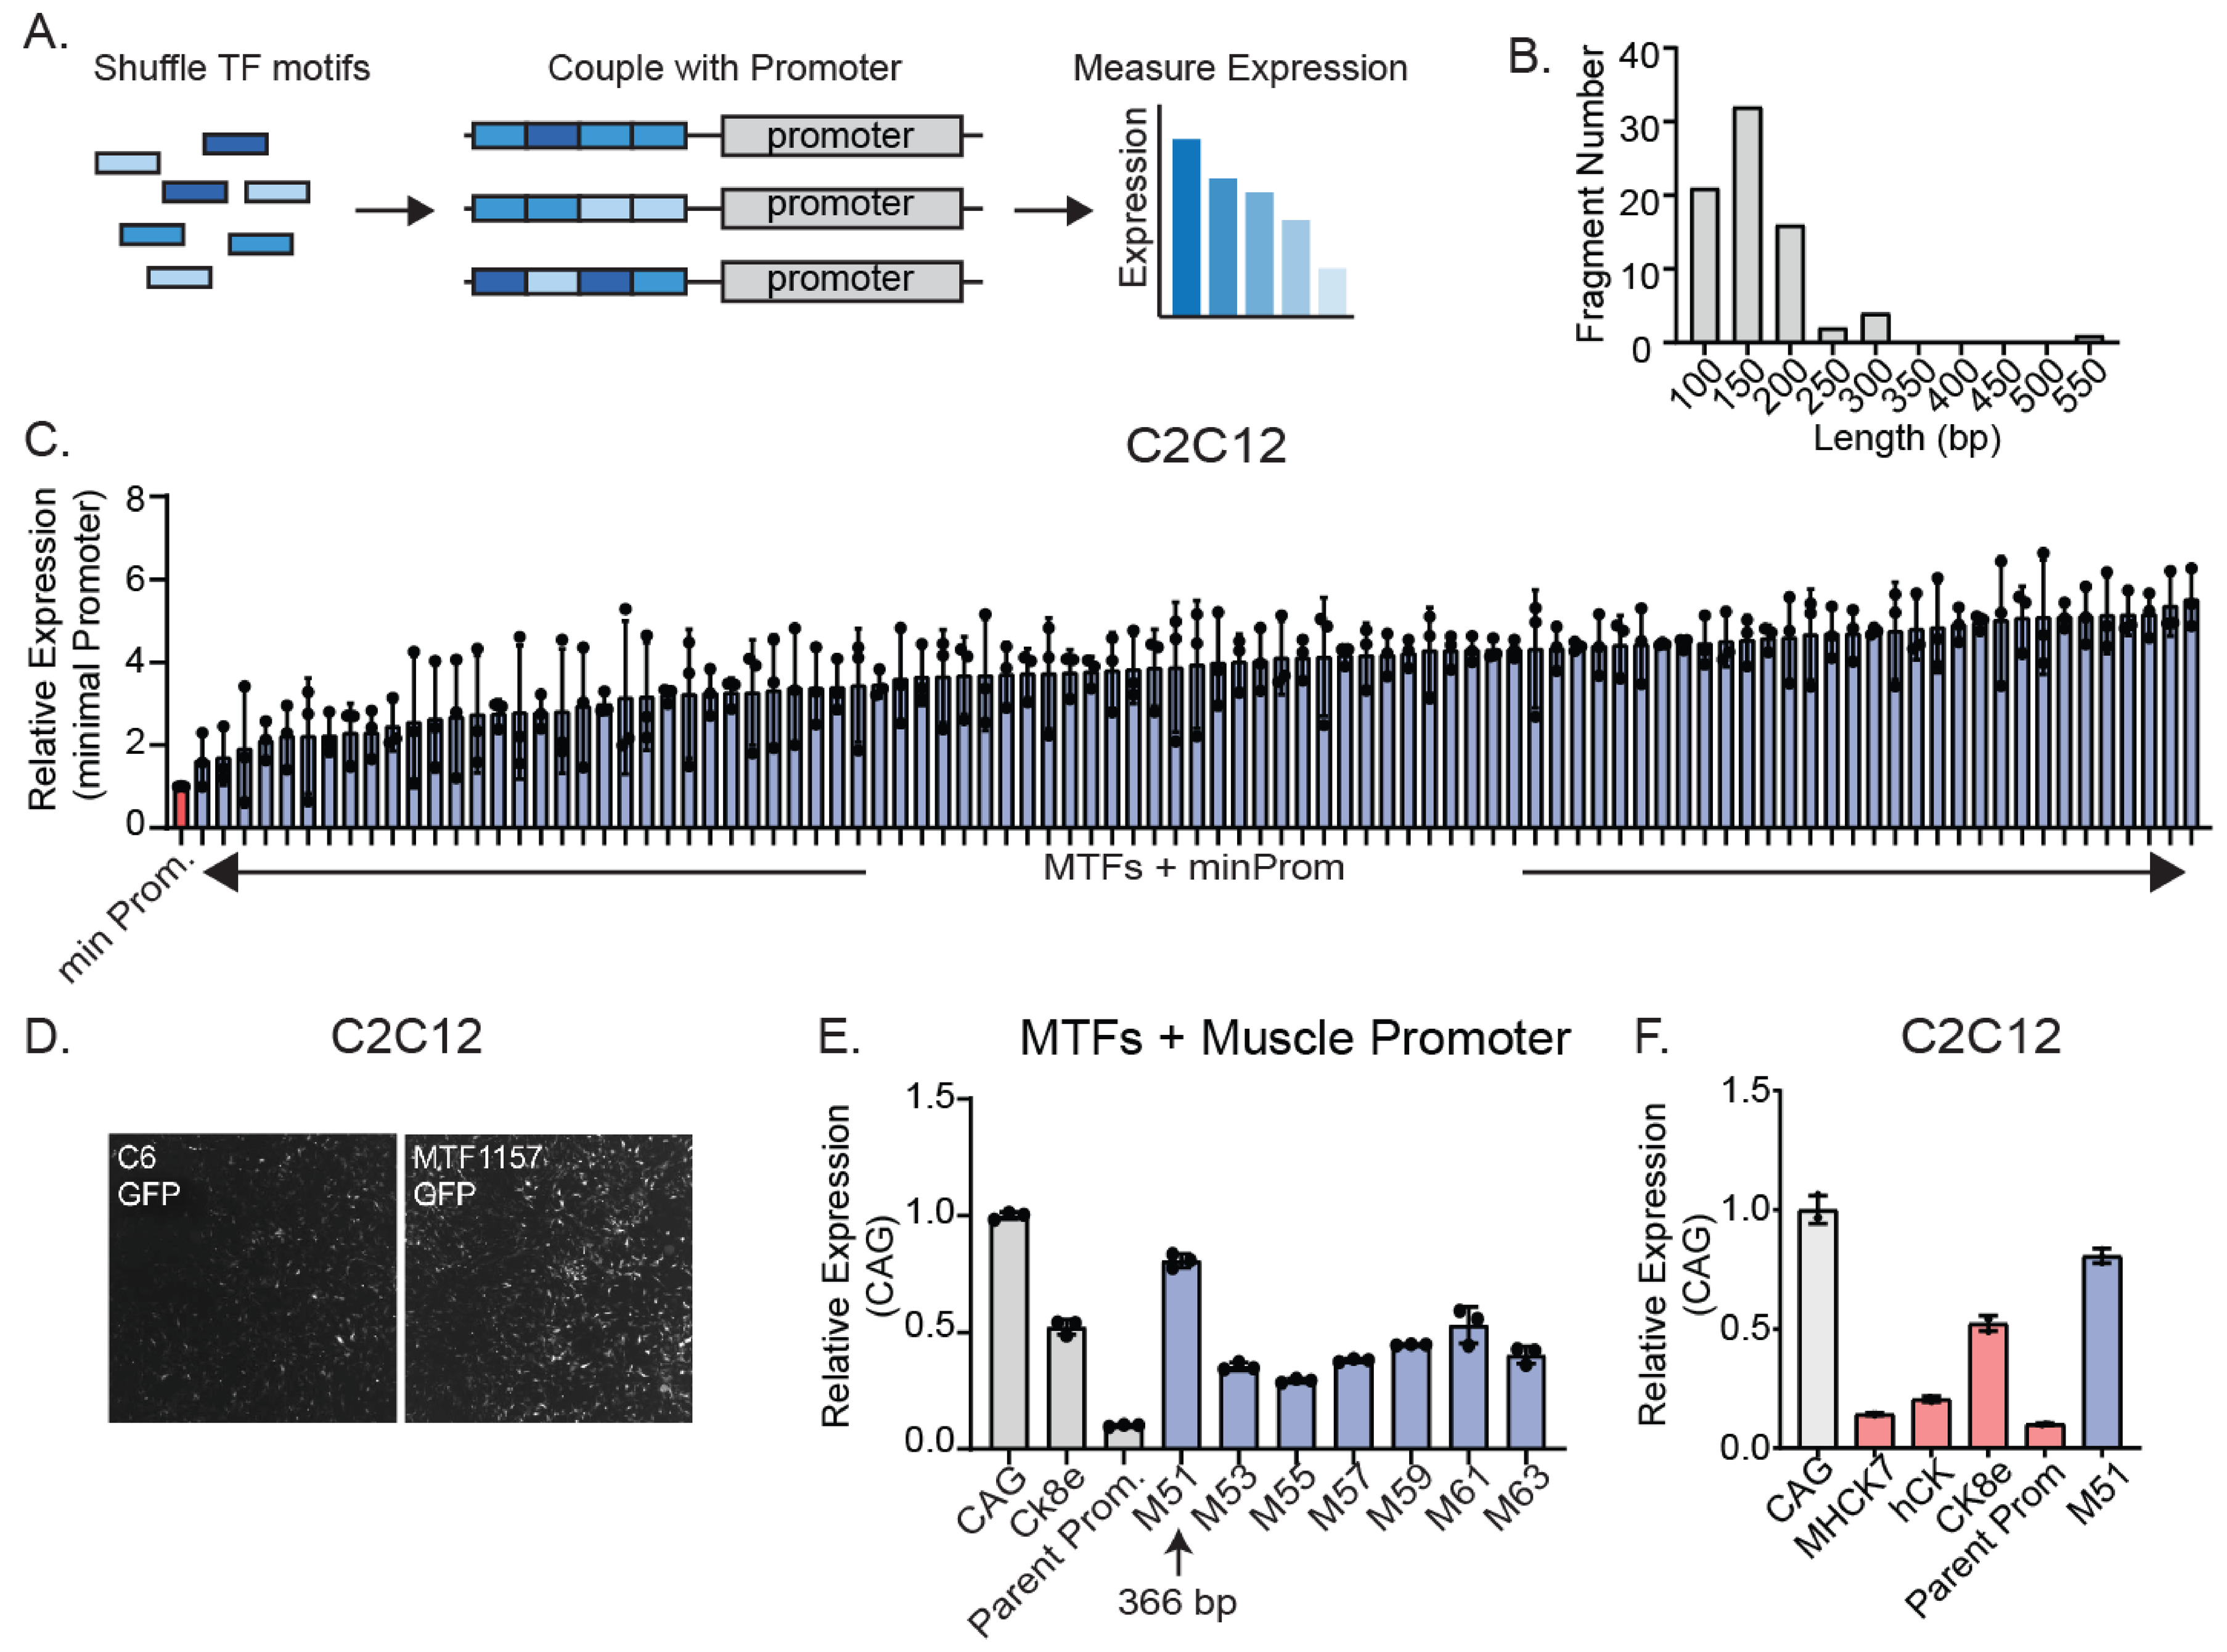
A.
Shuffle TF motifs
Couple with Promoter
Measure Expression
promoter
Expression
B.
Fragment Number
Length (bp)
C.
C2C12
Relative Expression (minimal Promoter)
min Prom.
MTFs + minProm
D.
C2C12
C6 GFP
MTF1157 GFP
E.
MTFs + Muscle Promoter
Relative Expression (CAG)
CAG Ck8e Parent Prom. M51 M53 M55 M57 M59 M61 M63
366 bp
F.
C2C12
CAG MHCK7 hCK CK8e Parent Prom M51

(A) TF motifs are randomly ligated together and then cloned upstream of a minimal, ubiquitous promoter. Expression is measured in a microscopy-based assay. (B) Representative sequencing shows majority of synthetic sequences are 200 bp or less. (C) Synthetic fragments increase expression from a minimal promoter up to 8-fold in transfected, mouse C2C12 muscle cell line. (D) Representative image of a top hit identified by the screen. (E) Fragments can be used in combination with other promoters to generate potent, novel muscle promoters. (F) MGTX-M51 is smaller and more potent than gold-standard muscle promoters currently used in the clinic.

Conclusions

- We have generated libraries of multiple novel composite promoters driving strong expression in muscle cell lines that are >>CAG and a subset that are >CMV.
- MeiraGTx CAG variants are 13-fold stronger than the original CAG *in vitro*.
- Evolution of a small, ubiquitous promoter, C11, results in 5-fold higher activity than CAG specifically in mouse muscle.
- Muscle-specific M24 promoter leads to stronger sustained expression than tMCK *in vivo*
- Our high-throughput platforms have yielded several synthetic enhancers which we have used to make new muscle promoters that are stronger and smaller than CK8e and other clinically-relevant promoters.
- These results highlight our diverse platform for promoter engineering at MeiraGTx which we have applied to other tissues and cell-types, including the liver, central nervous system and the salivary gland.

References

1. Wang et al. Nat Rev Drug Discov. 2019 May;18(5):358-378.
2. Au et al. Front Med (Lausanne). 2022 Feb 9;8:809118
3. Cordier et al. Hum Gene Ther. 2001 Jan 20;12(2):205-15
4. Wu et al. Mol Ther. 2010 Jan; 18(1): 80-86.