Efficacy and safety of AAV2/5-hRKp.RPGR to treat X-linked retinitis pigmentosa

P226

AJ Smith[13], A Georgiadis[13], JD Hoke[1], J Ribeiro[1], M Basche[1], SJ Robbie[1], A Gonzalez-Cordero[1], P Ovando-Roche[1], S Naylor[3], JWB Bainbridge[123], RR Ali[123]

[1] UCL Institute of Ophthalmology
[2]NIHR Biomedical Research Centre at Moorfields Eye Hospital
[3] MeiraGTx Ltd

Introduction

X-linked retinitis pigmentosa due to recessive mutations in the *RPGR* gene is the commonest form of inherited retinal dystrophy with an incidence of ~1 in 15,000 males. Most patients present with loss of night vision before 10 years of age and progress to legal blindness by the third to fourth decade.

We developed a *RPGR* gene therapy vector, AAV2/5-hRKp.RPGR, based on the AAV2/5 serotype. It carries a stable deletion mutant of the retina-specific *RPGR* isoform, ORF15, driven by the human rhodopsin kinase promoter.

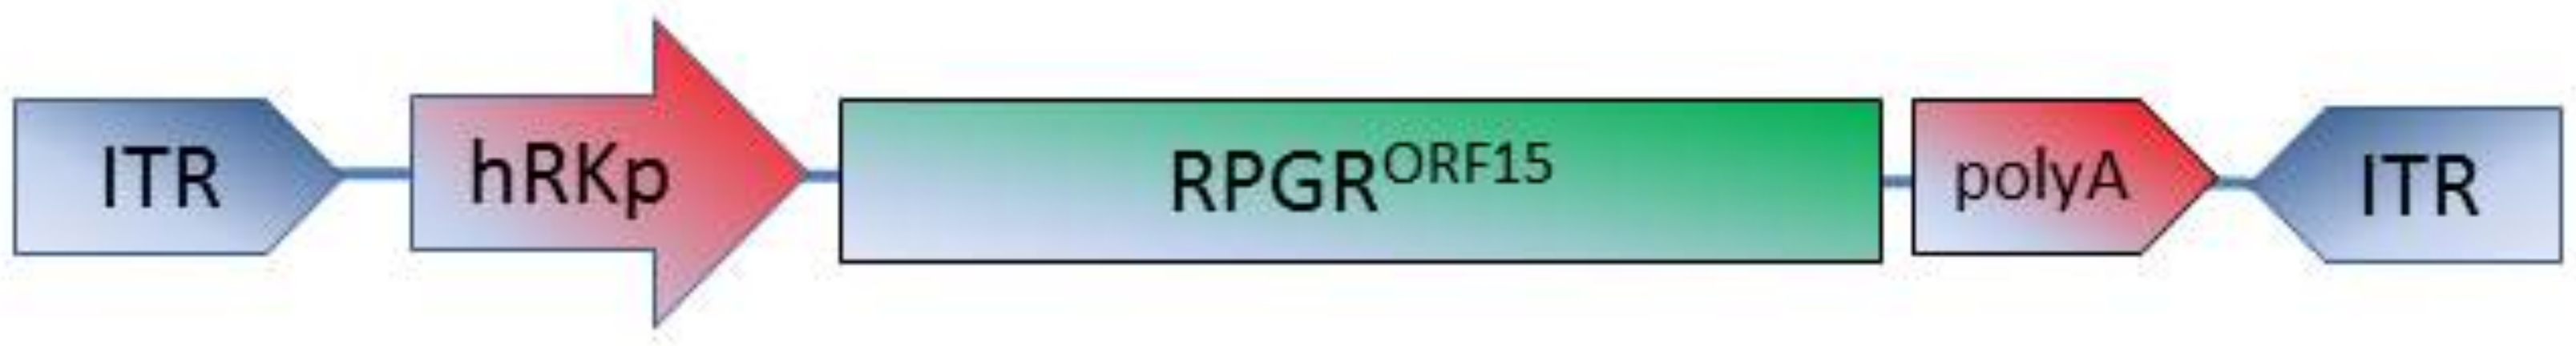
ITR
hRKp
RPGR ORF15
polyA
ITR

Efficacy in mice and hESC derived retina

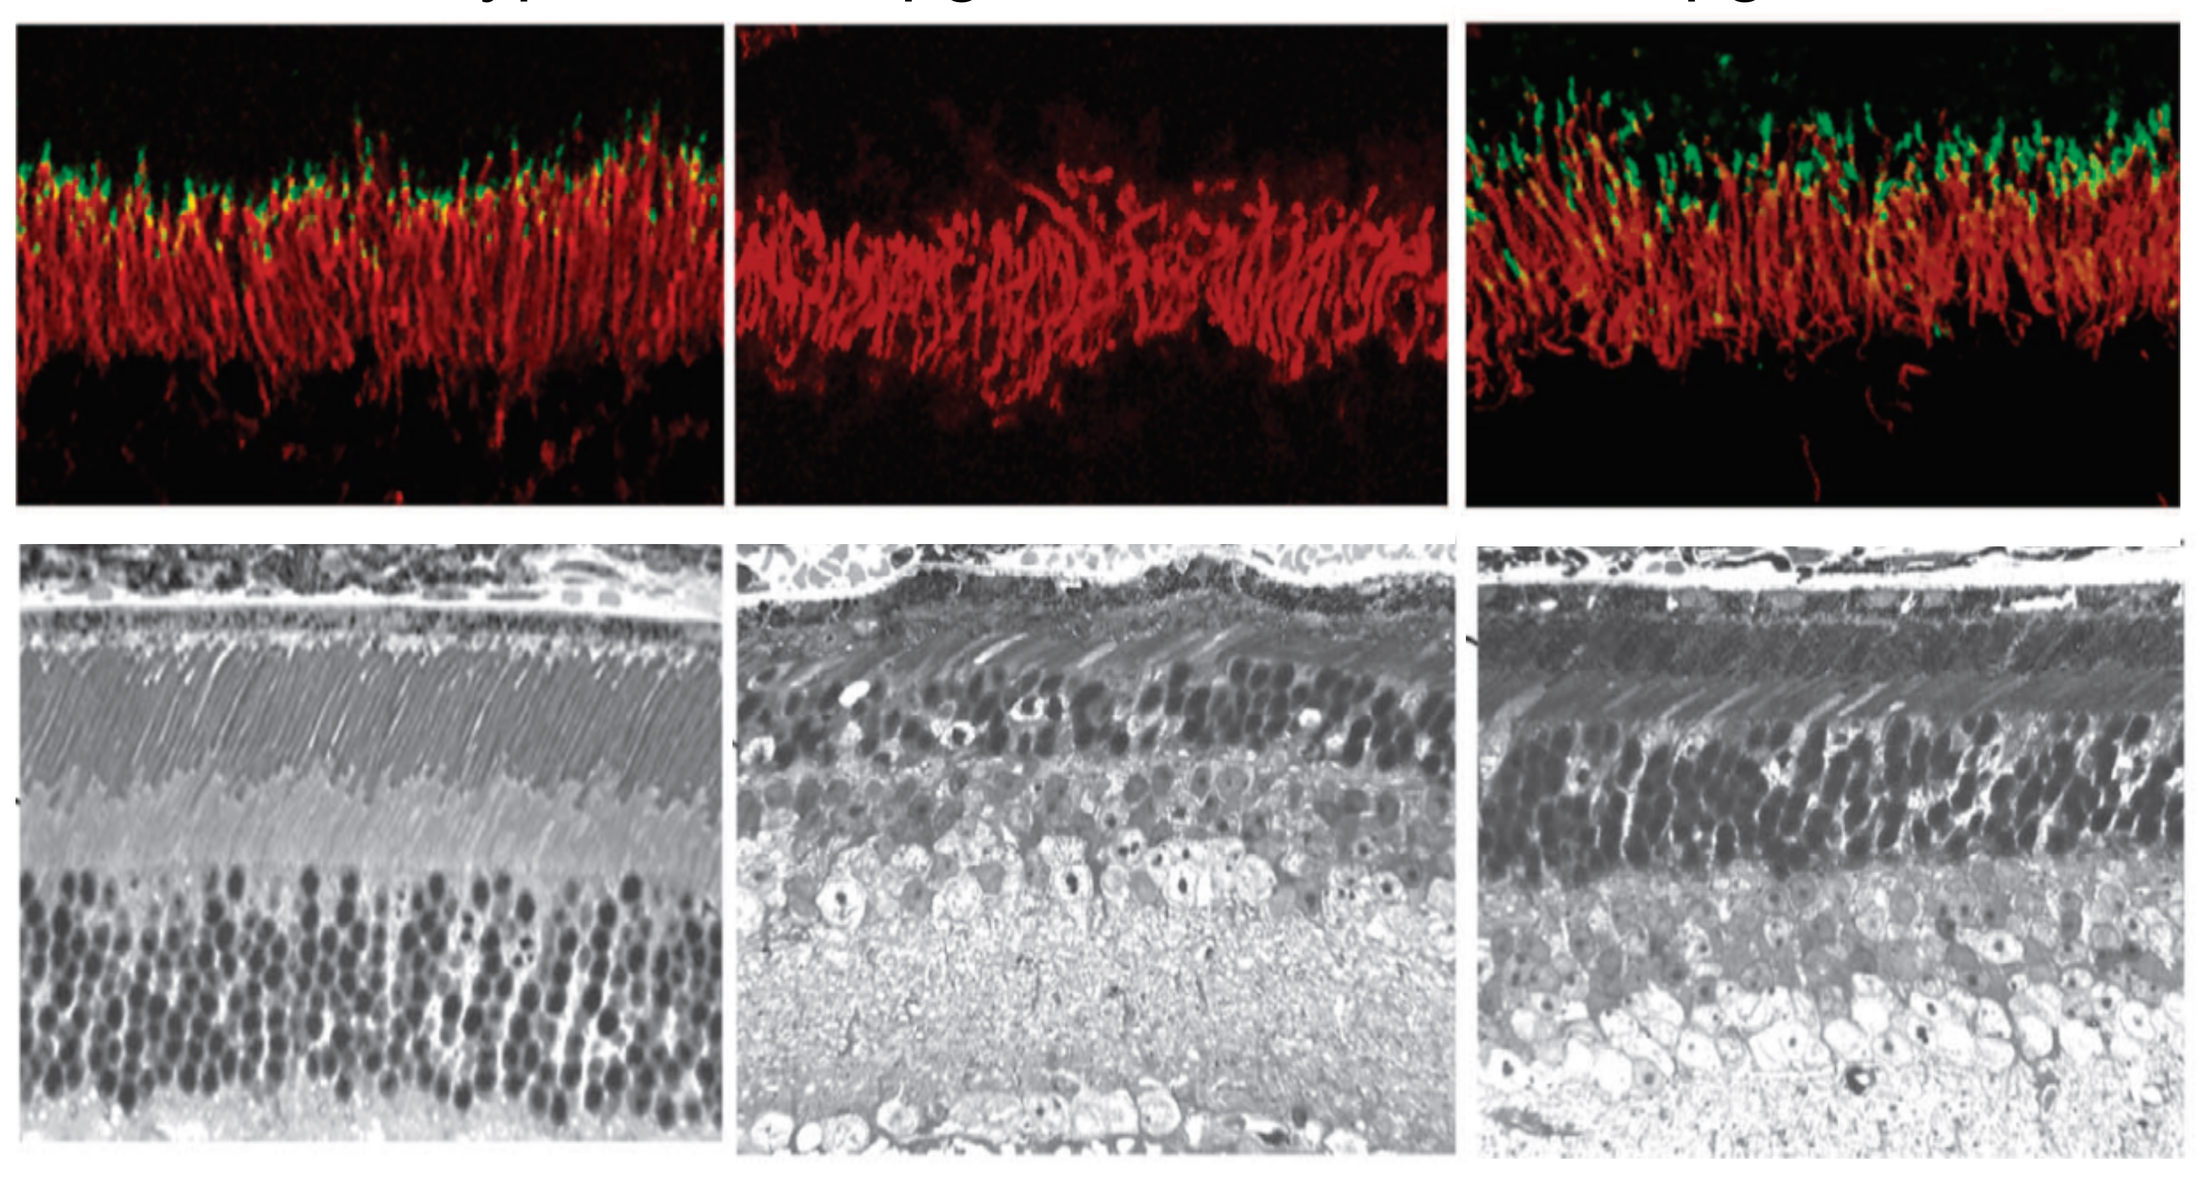
Wild type
Rpgr-/- untreated
Rpgr-/- treated

Subretinal administration of 2 x 10^9 vg of AAV2/8-hRKp.RPGR in *Rpgr*$^{-/-}$ mice restores RPGR protein to the connecting cilia of the photoreceptor cells (green in top panels), leading to a long-term preservation of photoreceptors (lower panels) and retinal function as measured by electroretinography (below).

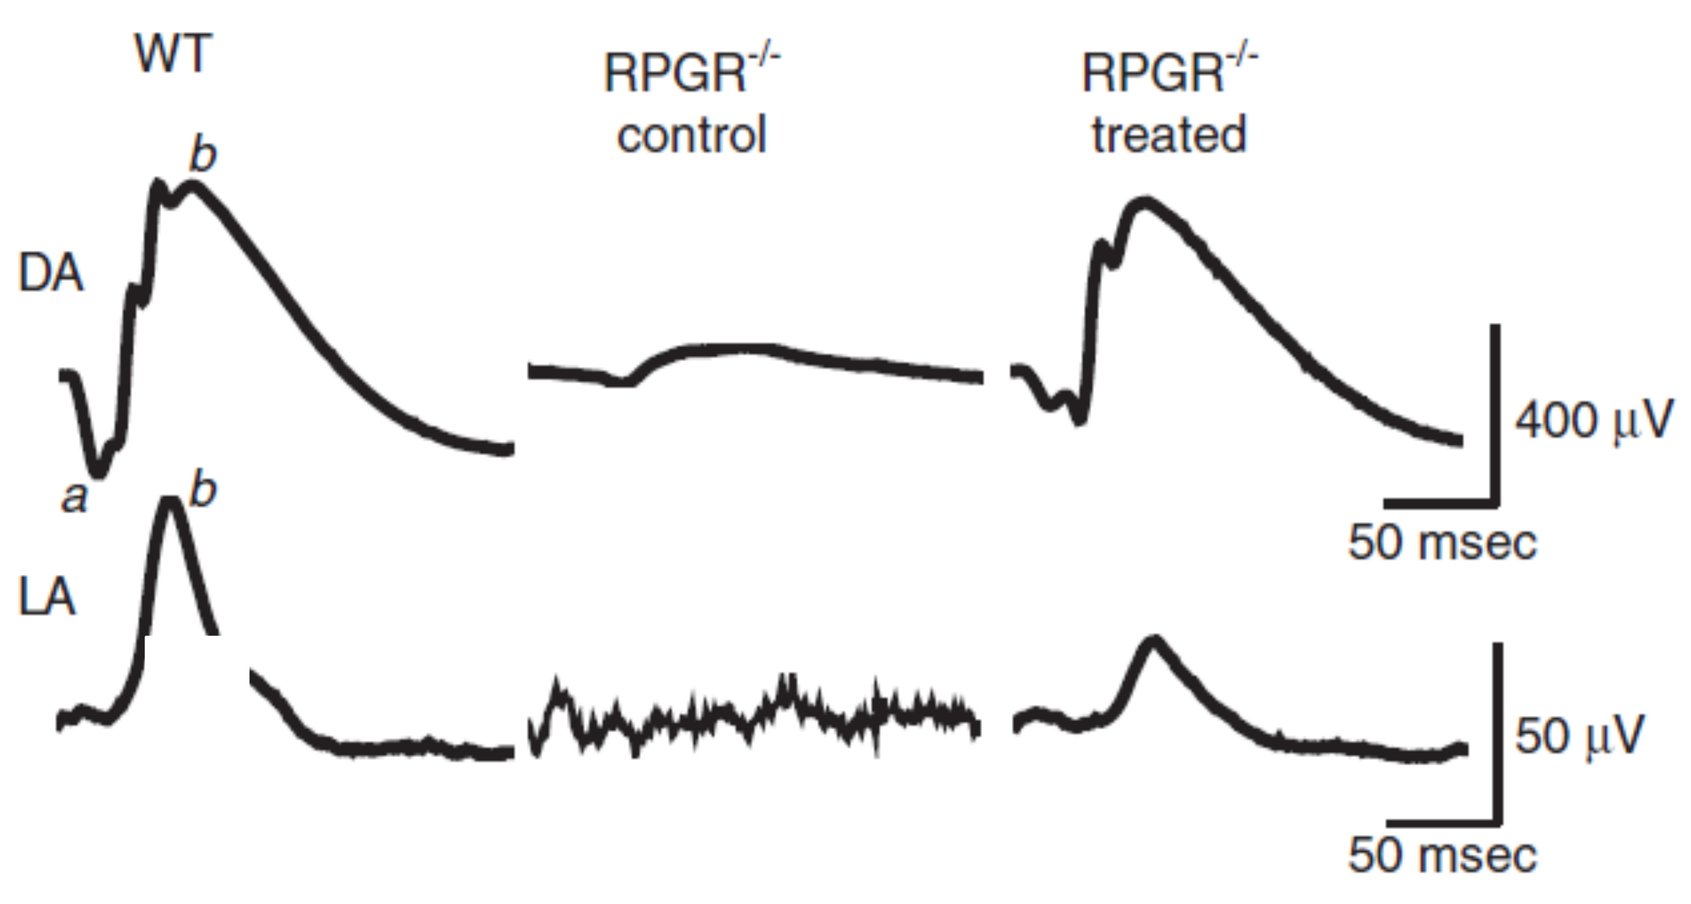
WT
RPGR-/- control
RPGR-/- treated
DA
LA
a
b
400 µV
50 msec
50 µV
50 msec

Restoration of cone opsin localisation to the outer segment in *Rpgr*$^{-/-}$ mice is equally efficient after subretinal administration of AAV2/5-hRKp.RPGR and AAV2/8-hRKp.RPGR

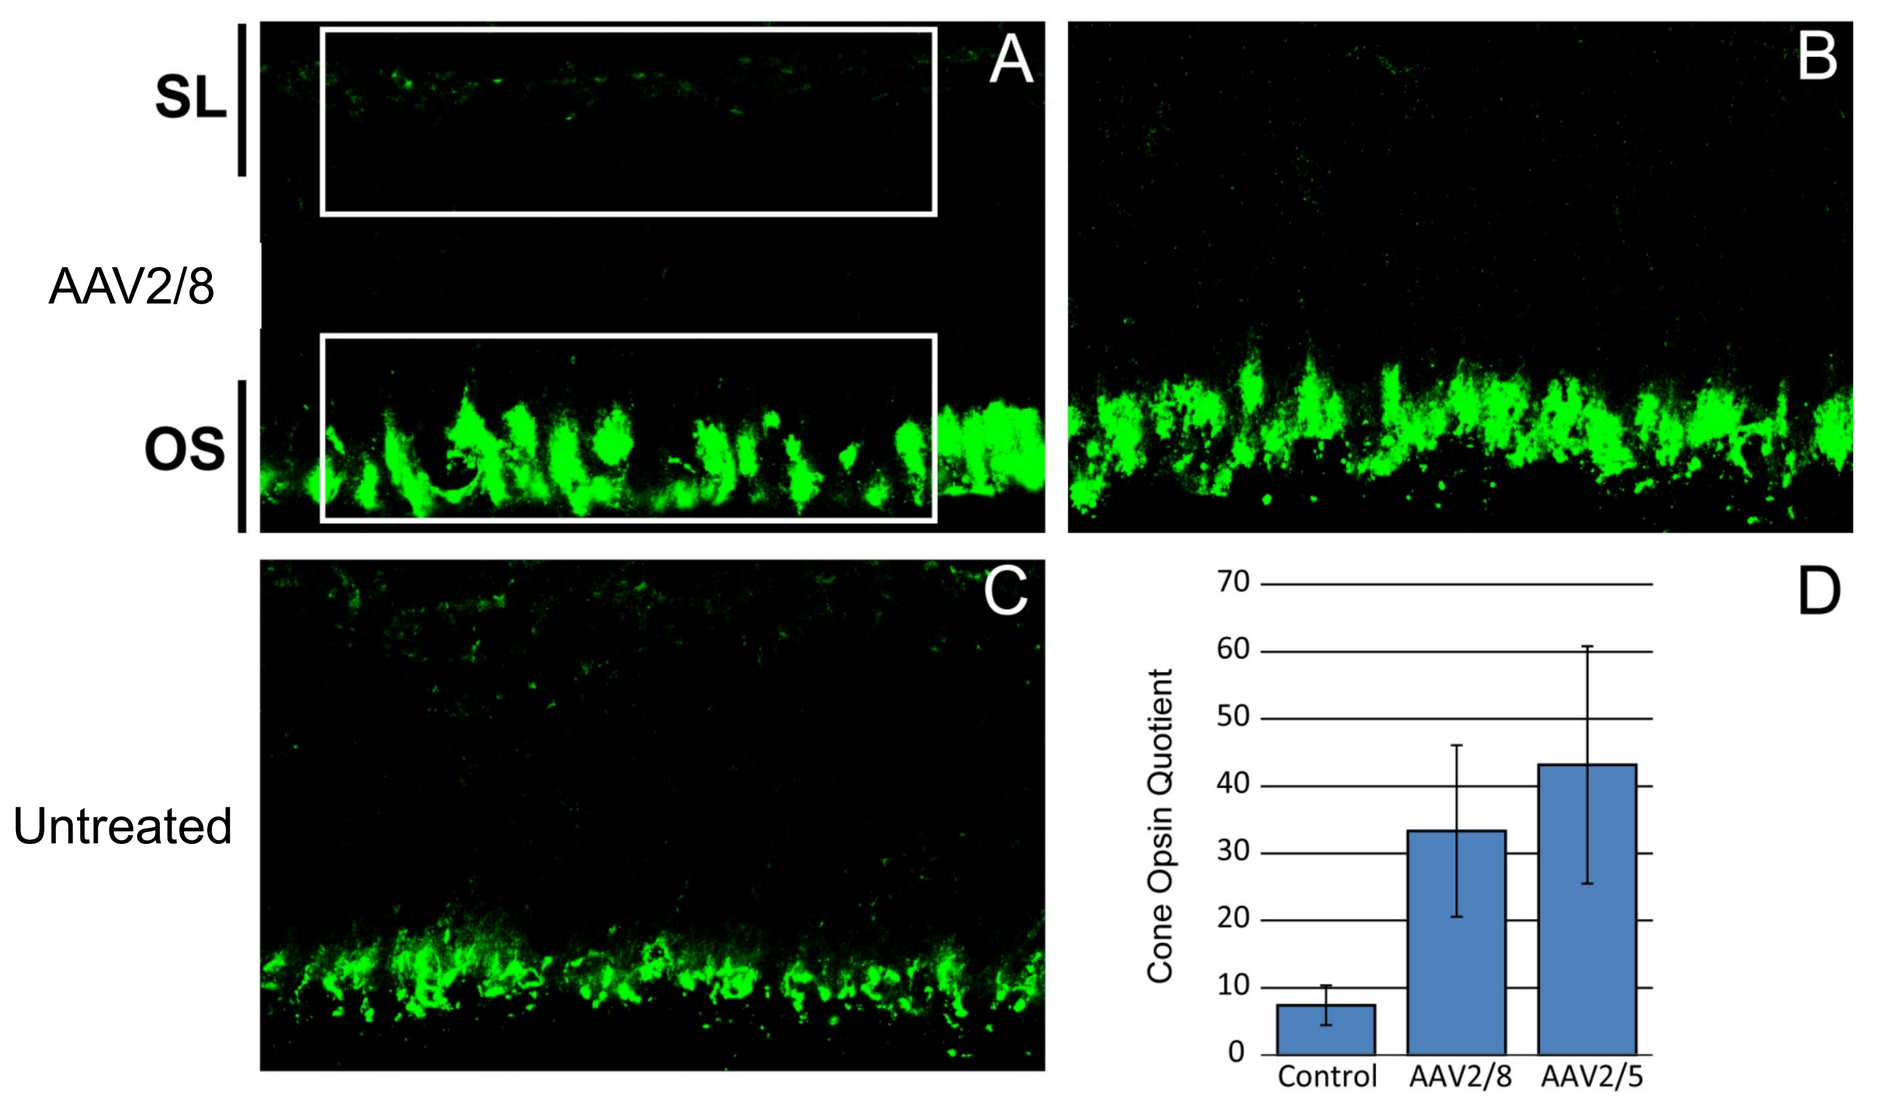
SL
OS
AAV2/8
AAV2/5
Untreated
A
B
C
D
Cone Opsin Quotient
Control
AAV2/8
AAV2/5

On RPGR$^{-/-}$ human iPSC-derived retinal organoids, *RPGR* gene transfer led to the restoration of RPGR protein at the connecting cilia of the photoreceptors (below, purple) and a dose-dependent recovery of poly-glutamylated tubulin, which is absent from the RPGR-/- retinal organoids (green).

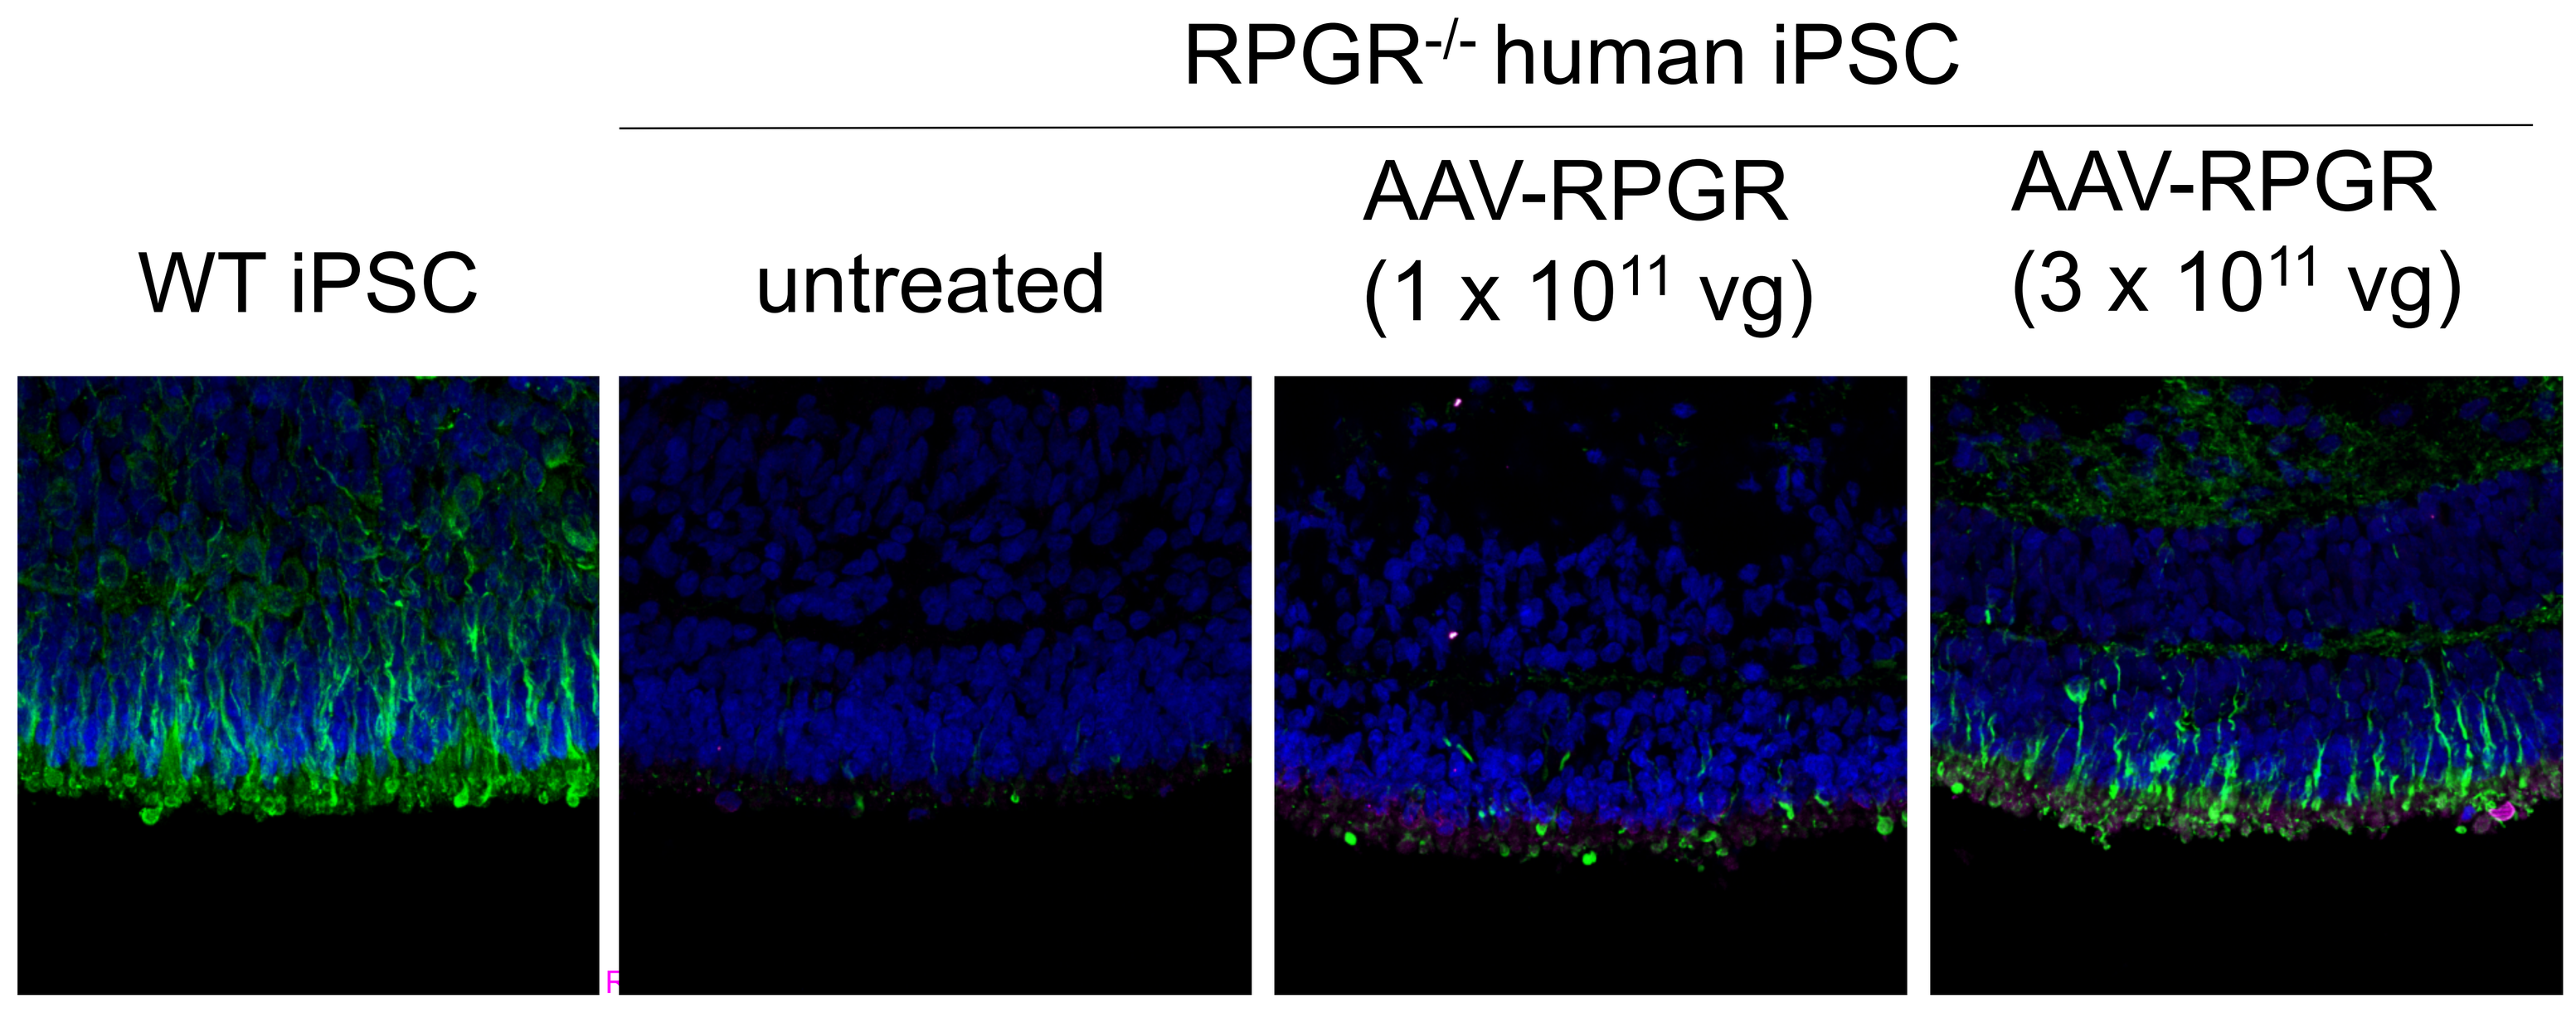
RPGR-/- human iPSC
WT iPSC
untreated
AAV-RPGR (1 x 10^11 vg)
AAV-RPGR (3 x 10^11 vg)

All retinal organoids at week 22 of differentiation using 2D/3D protocol [2]

Toxicology – Retinal function & structure

Adverse effects of subretinal administration of low or high dose (see table below) AAV2/5-hRKp.RPGR, produced using GMP methods, was assessed in wild type C57BL6/J mice and NZW rabbits.

Local effects on retinal structure or function were assessed by electroretinography and histology 7, 28 and 56 days after injection. Vector administration did not affect ERG amplitudes in mice (A, B) or rabbit (not shown), and did not affect outer nuclear layer thickness in either species (rabbit is shown in C and D) at any timepoint.

Long-term (6 months) overexpression of *RPGR*^{ORF15} did not affect the ERG activity (not shown) or structure of the retina in wt mice (E).

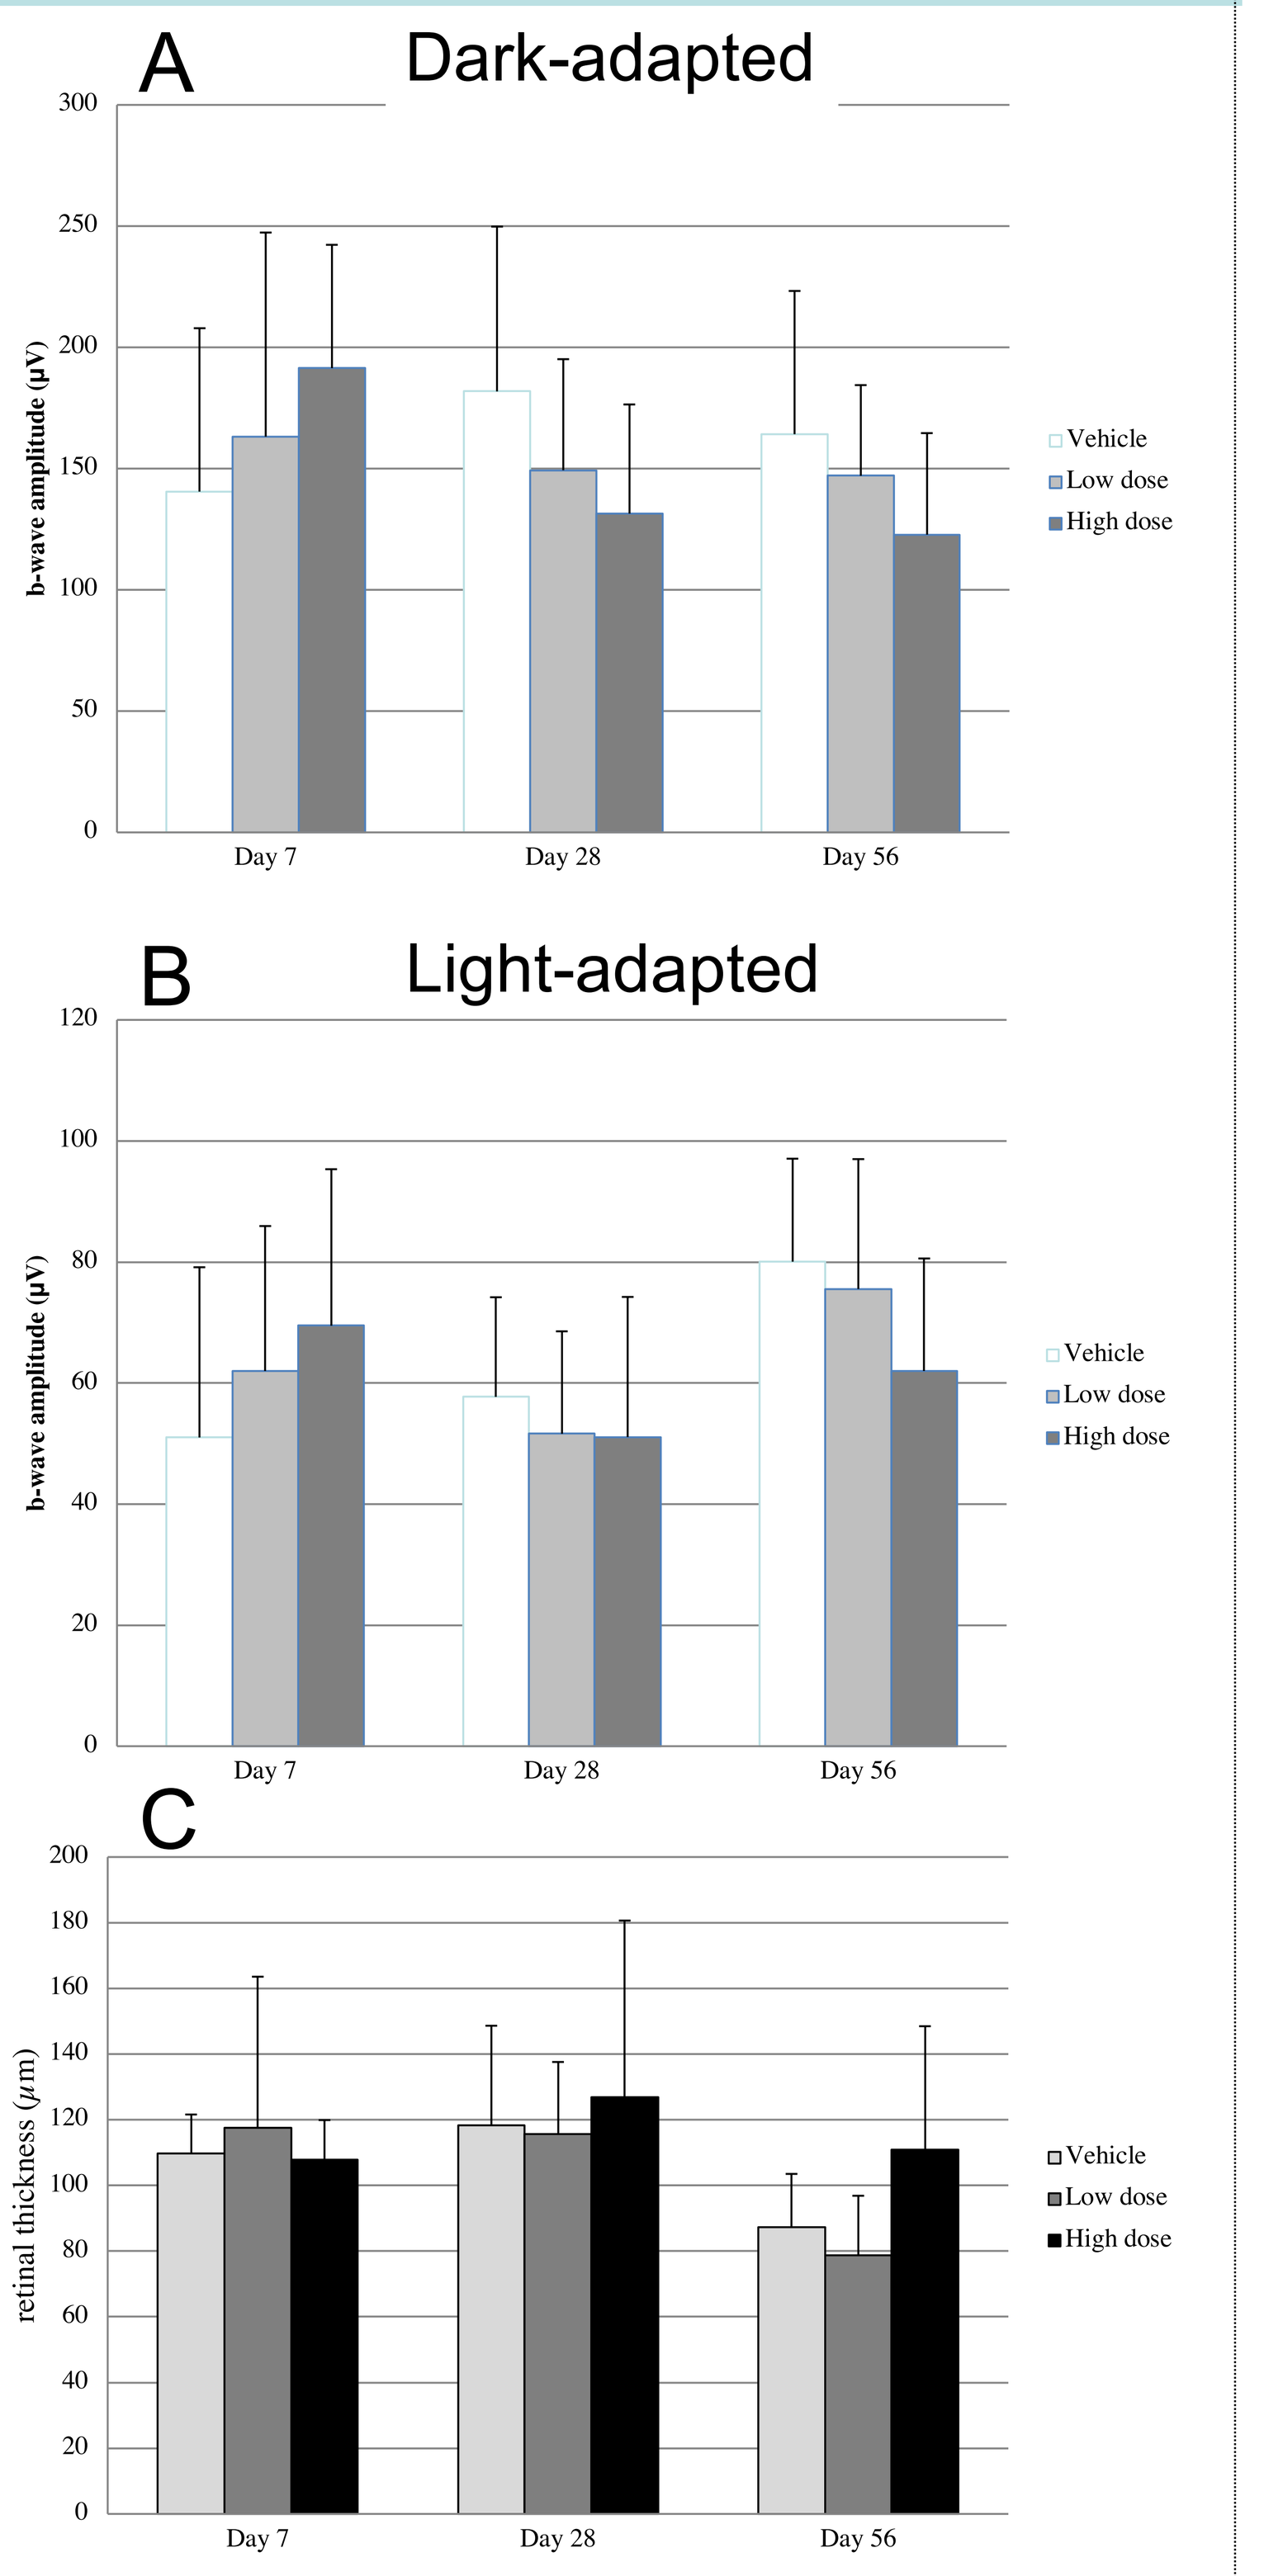
A
Dark-adapted
b-wave amplitude (µV)
Vehicle
Low dose
High dose
Day 7
Day 28
Day 56
B
Light-adapted
C
retinal thickness (µm)

Vector doses used in toxicology studies (vg/eye)

	Low	High
Mouse	1 x 10^9	4 x 10^9
Rabbit	8 x 10^{10}	2.4 x 10^{11}

D

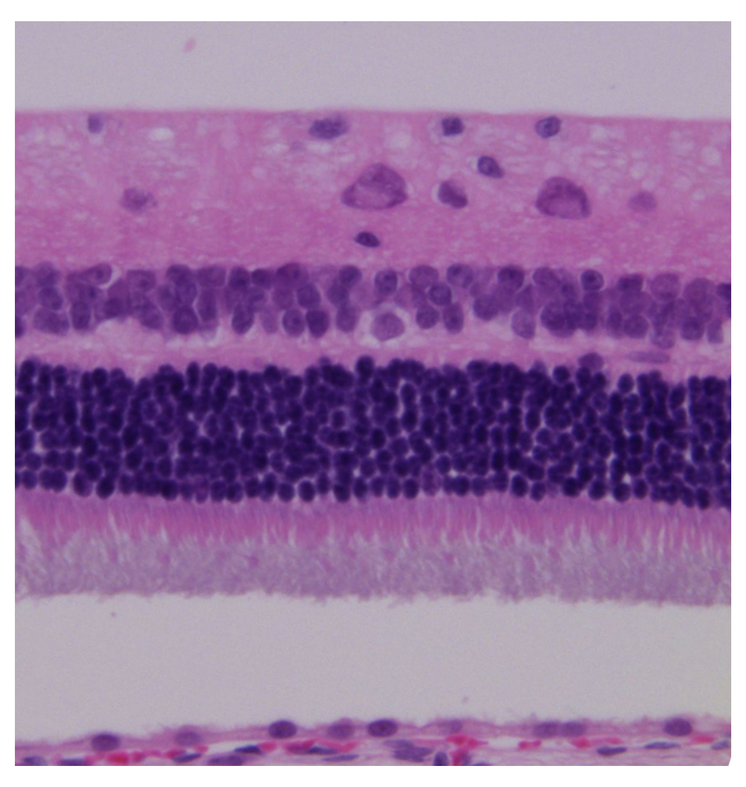

E

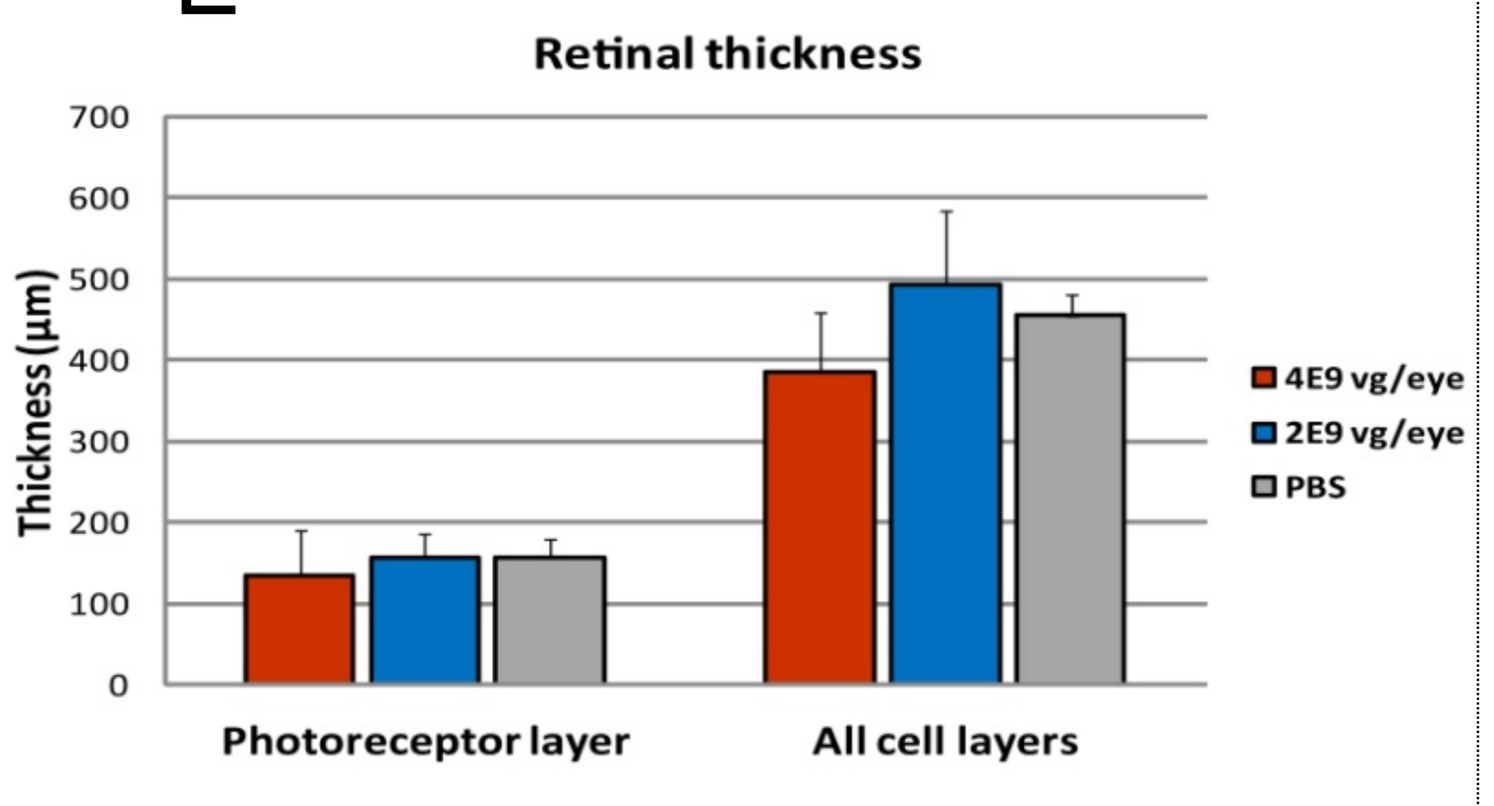
Retinal thickness
Thickness (µm)
4E9 vg/eye
2E9 vg/eye
PBS
Photoreceptor layer
All cell layers

Biodistribution and immune responses

Vector administration did not lead to increased morbidity or mortality in either mice or rabbits.

Moderate neutralising antibody responses (rabbit shown in A) and total antibody titres (mouse shown in B) were present in both species, but did not correlate with pathology.

In the rabbits, vector biodistribution was observed along the tissues of the optic tract and in the liver. Vector copy numbers diminished over time and at day 56 were only detected in the superior colliculus (table).

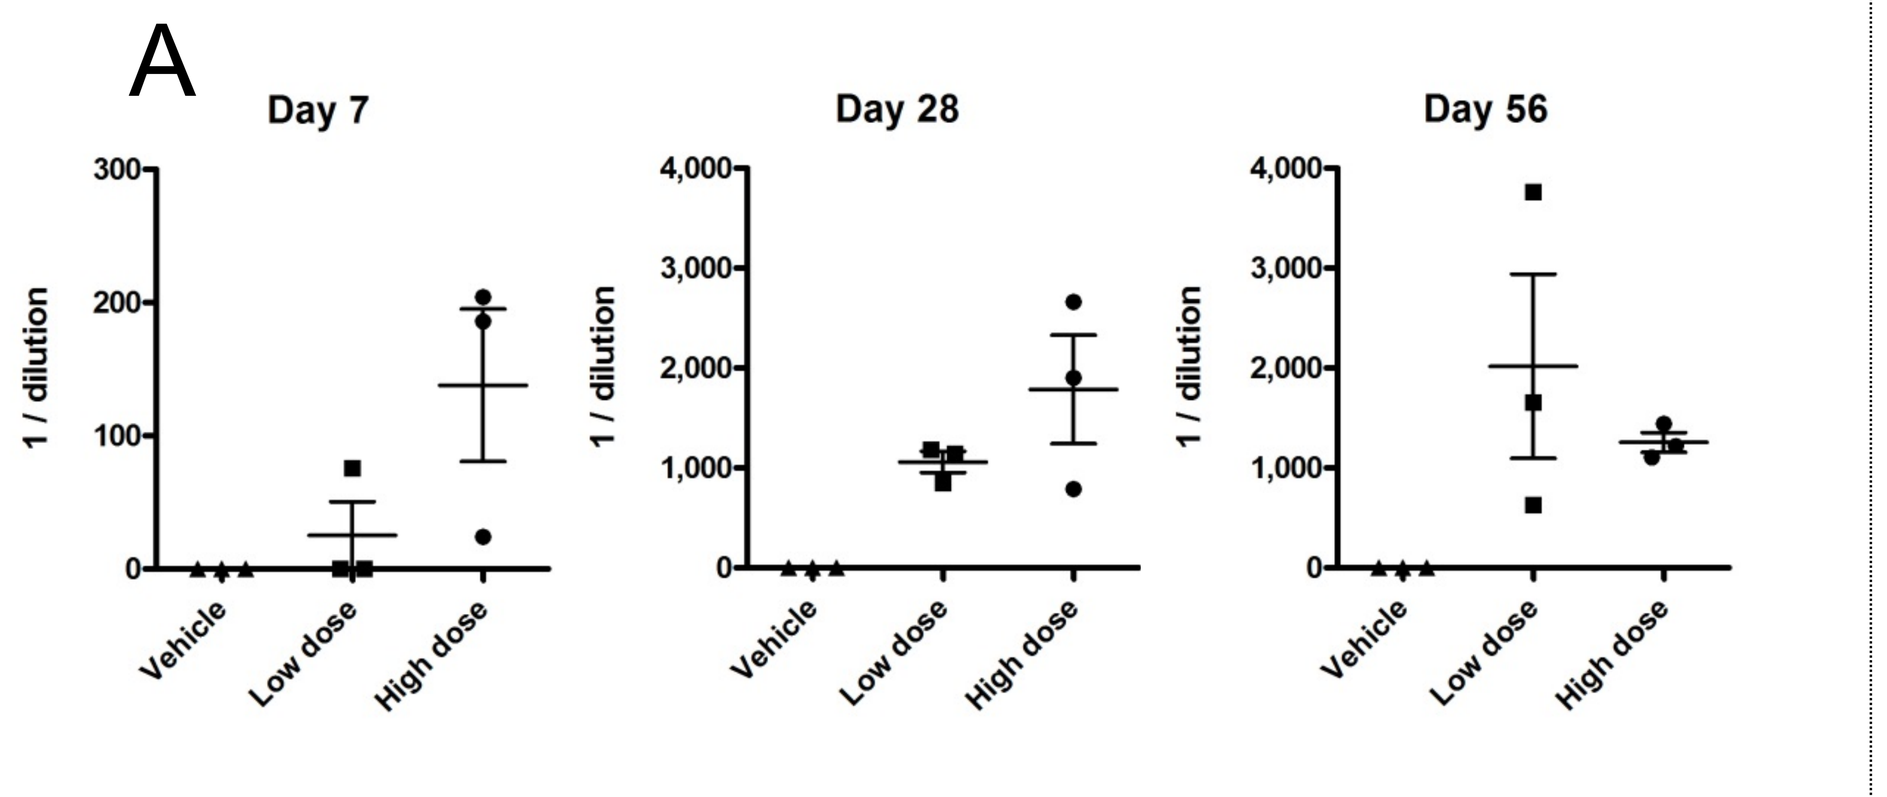
A
Day 7
Day 28
Day 56
1 / dilution
Vehicle
Low dose
High dose

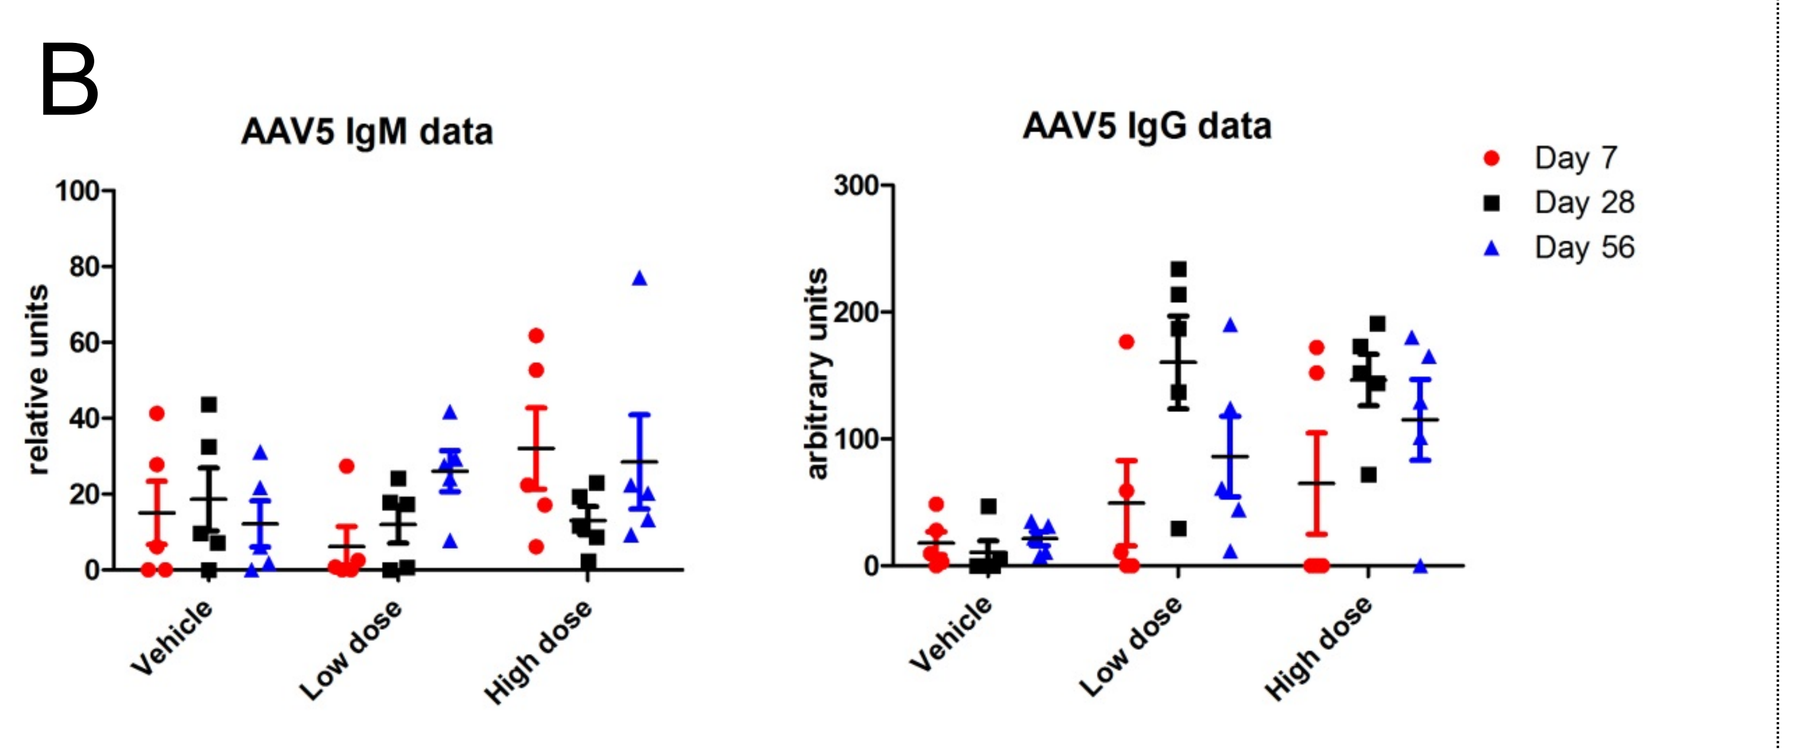
B
AAV5 IgM data
AAV5 IgG data
relative units
arbitrary units
Day 7
Day 28
Day 56
Vehicle
Low dose
High dose

High dose	Rabbit 2 Day 7	Rabbit 8 Day 7	Rabbit 9 Day 7	Rabbit 21 Day 28	Rabbit 25 Day 28	Rabbit 27 Day 28	Rabbit 12 Day 56	Rabbit 14 Day 56	Rabbit 17 Day 56
Retina OS	B	B	B	-	-	-	-	-	-
RPE OS	162	B	134	B	B	B	-	-	-
Vitr. OS	B	B	B	-	-	-	-	-	-
ON OS	158	548	608	B	B	B	-	-	-
Chiasm	440	B	B	B	B	B	-	-	-
Vis. Cort.	B	B	B	-	-	-	-	-	-
Sup. Coll.	874	526	3480	124	B	192	102	198	B
Kidney	B	B	B	-	-	-	-	-	-
Liver	574	2000	3200	141	177	220	B	B	B
Spleen	1340	B	B	B	B	B	-	-	-
Lung	B	B	B	-	-	-	-	-	-
Testis	-	122	B	-	-	B	-	-	-
Ovary	B	-	-	-	-	-	-	-	-
Uterus	B	-	-	-	-	-	-	-	-
Heart	B	B	B	-	-	-	-	-	-
Lymph N.	B	B	B	-	-	-	-	-	-
ON OD	1496	1548	5820	B	B	B	-	-	-

Amount of vector in vg per µg genomic DNA
B: below detection

Conclusions

Subretinal administration of AAV2/5-hRKp.RPGR leads to a preservation of retinal function and structure in *Rpgr*$^{-/-}$ mice. Assessment of adverse effects suggests that subretinal administration of the vector is safe for use in the clinic.

References:
1. Pawlyk *et al.*, Gene Therapy (2016), 23: 196-204.
2. Gonzalez-Cordeiro *et al*, Stem Cell Reports (2017), 12: 820-37.

NHS National Institute for Health Research